Wnt Pathway Suppression and Fibrosis

Bunyard P, Bhamra I, Eckersley K, Chaplin C, Offer E, Mathieson M, Calcraft PJ, Wright J, MacFaul P, Phillips C, Best S, Jones C, Armer R.
Redx Pharma Plc, Alderley Park, Alderley Edge, United Kingdom.

BACKGROUND

- Wnt signalling is known to be important for tissue remodelling in several pathologies including cancer, auto-immunity and fibrosis[1,2,3].
- Porcupine (PORCN) is a membrane-bound O-acyltransferase required for and dedicated to palmitoylation of Wnt ligands, an essential step in the processing of Wnt ligands for secretion[4].
- Inhibition of Wnt signalling is likely to impact on several mechanisms that underpin tissue remodelling in fibrotic diseases such as suppression of inflammation, reduction of apoptosis, prevention of epithelial mesenchymal transition and inhibition of fibroblast activation[5,6,7,8].

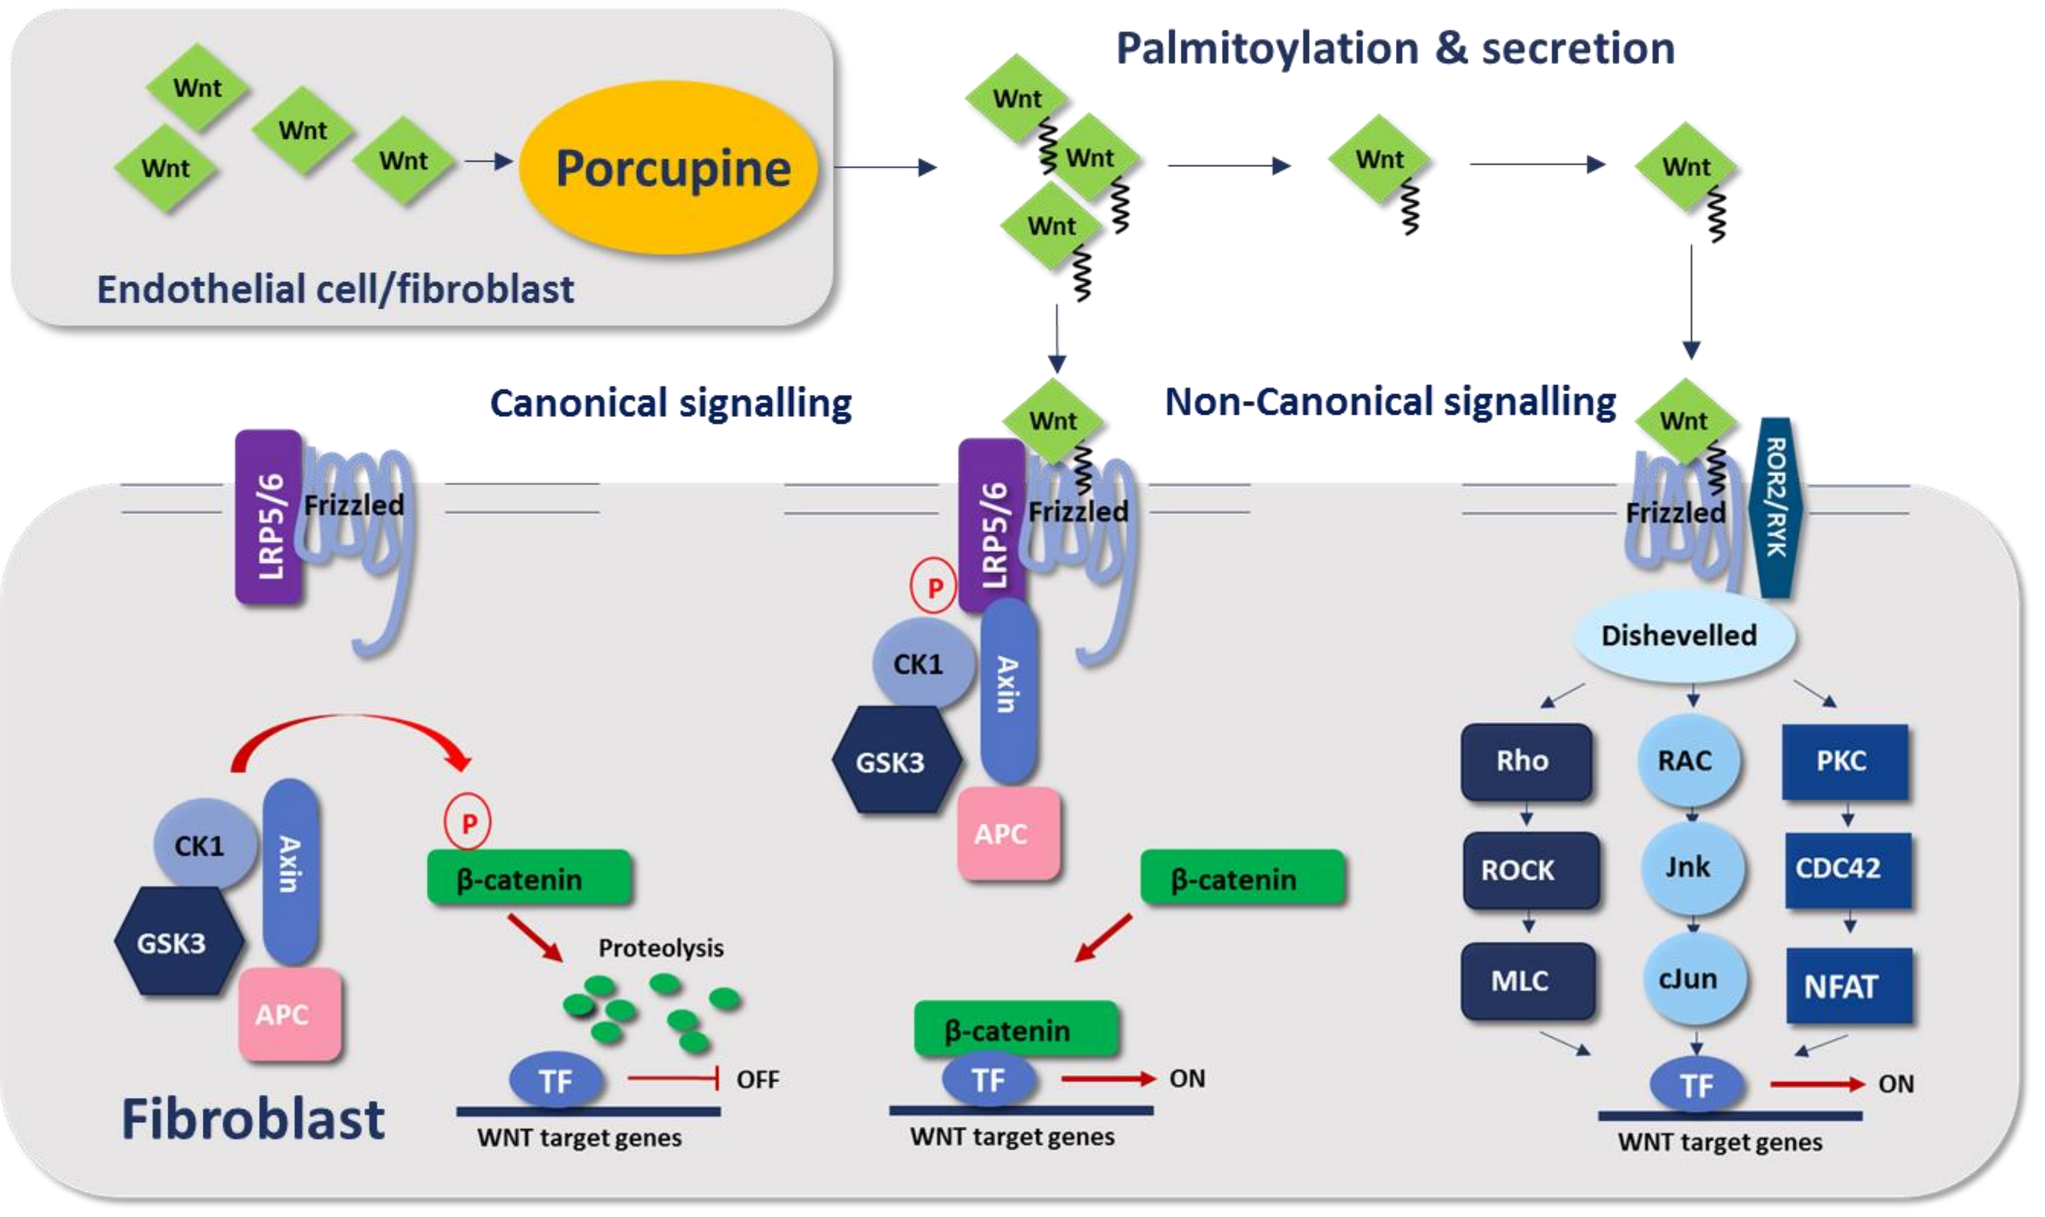
Wnt
Porcupine
Endothelial cell/fibroblast
Palmitoylation & secretion
Canonical signalling
Non-Canonical signalling
LRP5/6
Frizzled
ROR2/RYK
CK1
GSK3
Axin
APC
P
β-catenin
Proteolysis
TF
OFF
ON
WNT target genes
Fibroblast
Dishevelled
Rho
ROCK
MLC
RAC
Jnk
cJun
PKC
CDC42
NFAT

Figure 1. *Both the canonical and non-canonical Wnt signalling pathways play a role in fibroblast activation.*

RESULTS

RXC006 displays potent *in vitro* Wnt pathway inhibition

RXC006 is a potent inhibitor of canonical Wnt signalling

- Analysis of RXC006 suppression of Wnt signalling was determined using a ß-catenin reporter assay and ß-catenin immunofluorescence.
- L-Wnt3a cells, overexpressing Wnt3a, were dosed with RXC006, then the conditioned media (CM) was collected from these cells and transferred to the ß-catenin reporter cell line to determine pathway activation.
- CM from RXC006 treated cells suppresses ß-catenin activation.
- RXC006 suppresses β-catenin stabilization directly in the Wnt-3a expressing cell line.

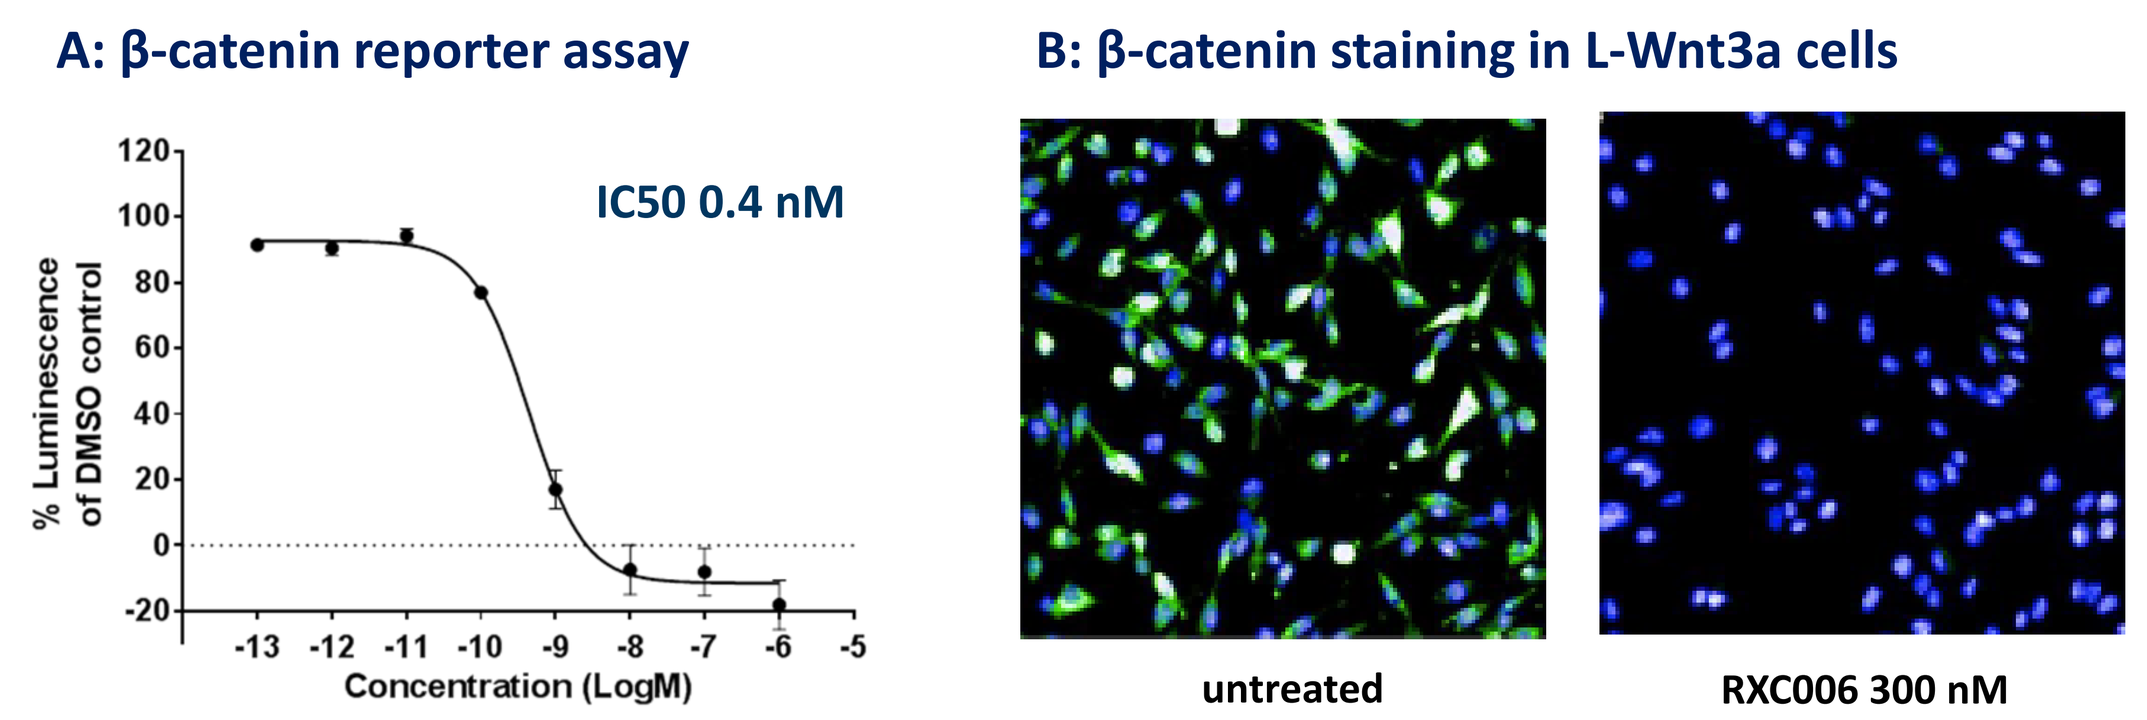
A: β-catenin reporter assay
IC50 0.4 nM
% Luminescence of DMSO control
Concentration (LogM)
B: β-catenin staining in L-Wnt3a cells
untreated
RXC006 300 nM

Figure 2. *(A) ß-catenin reporter cell line was treated with CM from L-Wnt3a cells cultured ±RXC006 for 24 h. (B) Immunofluorescence shows that RXC006 supresses ß-catenin stabilisation in L-Wnt3a cells (representative images shown).*

RXC006 reduces Wnt3a-induced α-SMA expression in human lung fibroblasts

- Wnt3a, in CM from L-Wnt3a cells, induces differentiation of primary human lung fibroblasts (HLFs) to myofibroblasts, indicated by the expression α-SMA.
- CM from cells treated with RXC006, inhibits the expression of α-SMA in HLFs.

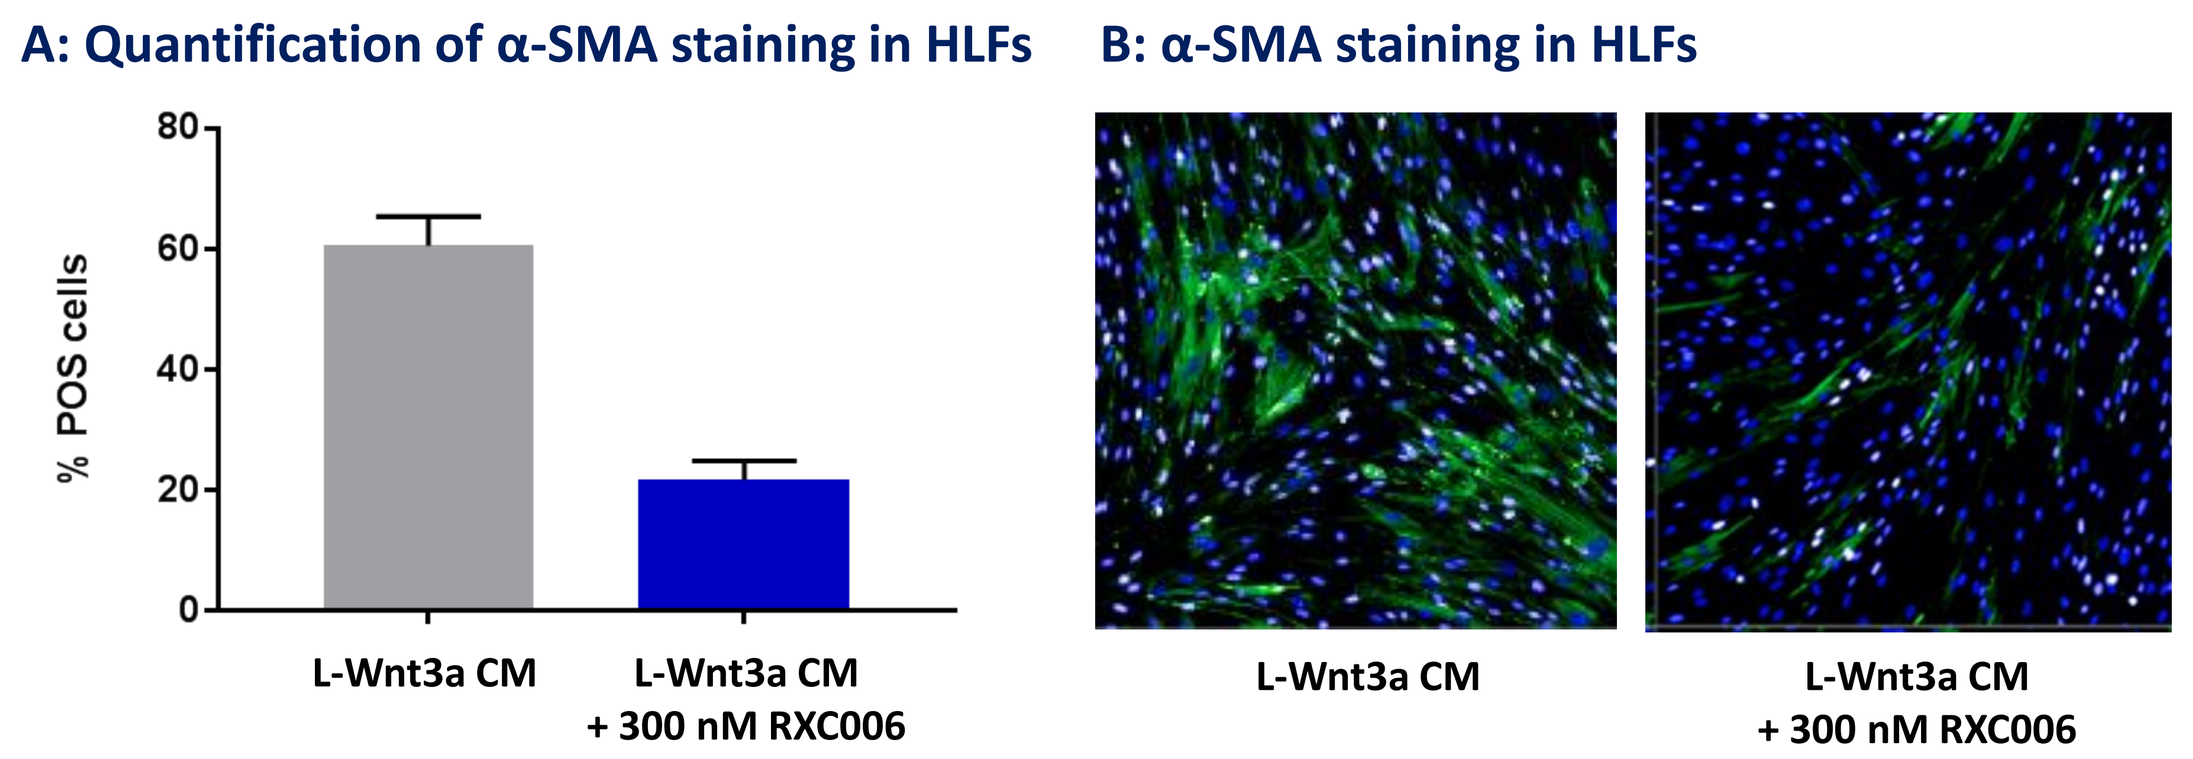
A: Quantification of α-SMA staining in HLFs
% POS cells
L-Wnt3a CM
L-Wnt3a CM + 300 nM RXC006
B: α-SMA staining in HLFs
L-Wnt3a CM
L-Wnt3a CM + 300 nM RXC006

Figure 3. *CM from L-Wnt3a cells cultured ±RXC006 for 48 h was diluted 1:2 and transferred onto HLFs. Expression of α-SMA was determined by immunofluorescence. (A) shows mean±SEM n=3 and (B) shows representative images of α-SMA expression.*

RXC006 is a potent inhibitor of non-canonical Wnt5a release in human lung fibroblasts

- TGFß induces the expression and release of Wnt5a from HLFs.
- RXC006 potently inhibits the release of Wnt5a from HLFs leading to an intracellular accumulation of Wnt5a.

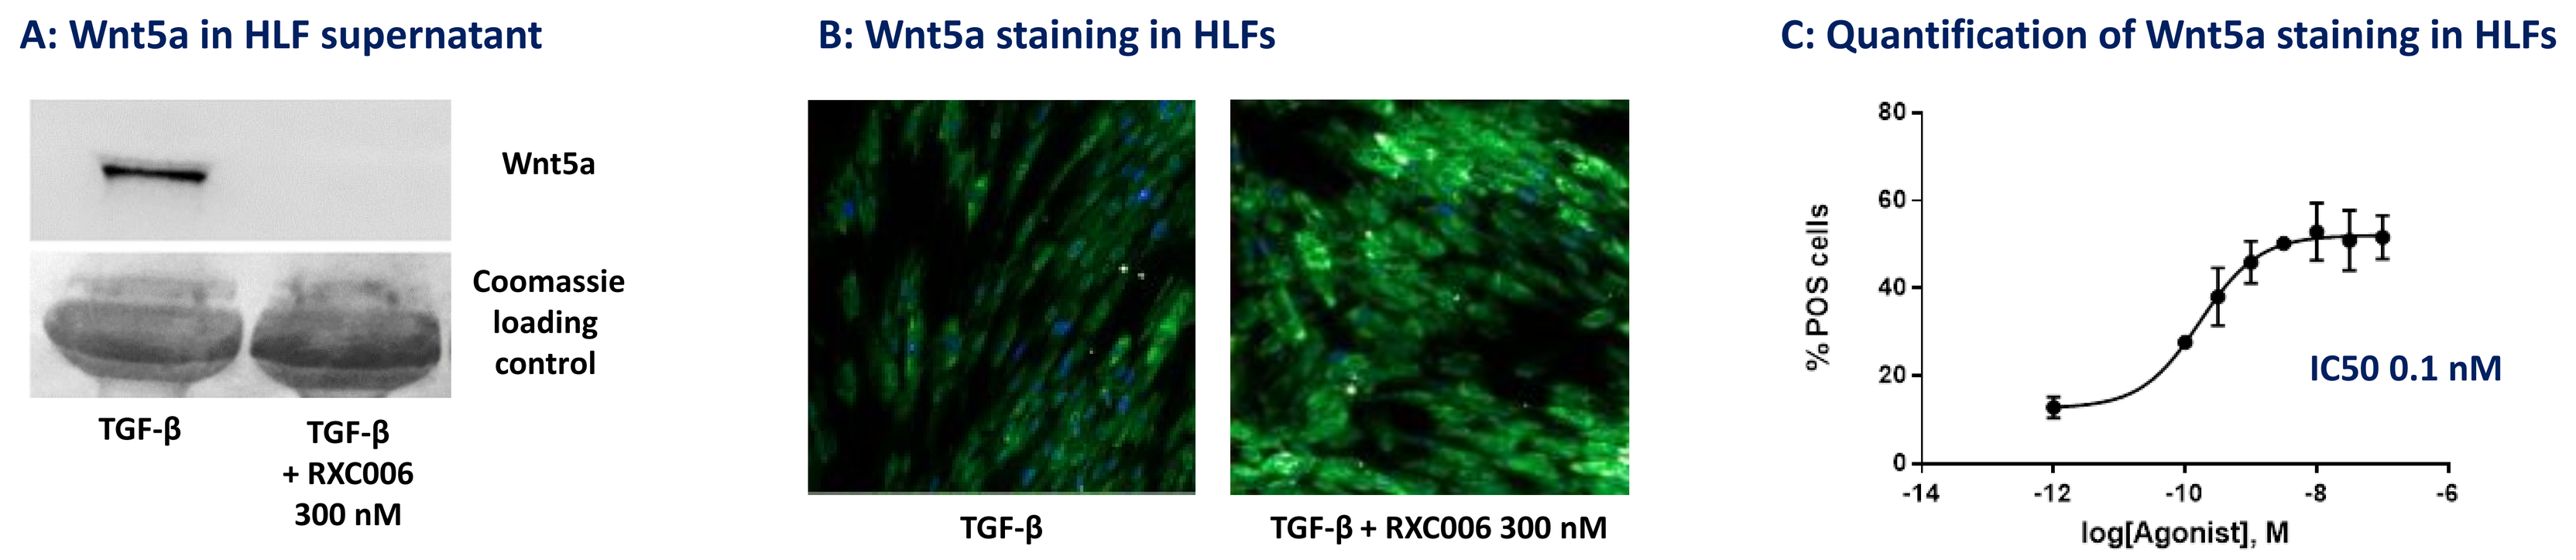
A: Wnt5a in HLF supernatant
Wnt5a
Coomassie loading control
TGF-β
TGF-β + RXC006 300 nM
B: Wnt5a staining in HLFs
TGF-β
TGF-β + RXC006 300 nM
C: Quantification of Wnt5a staining in HLFs
% POS cells
IC50 0.1 nM
log[Agonist], M

Figure 4. *HLFs were incubated with TGFß at 10 ng/mL ±RXC006. (A) Supernatant was concentrated and analysed by western blot for the expression of Wnt5a. Cells were also analysed for the expression of Wnt5a by immunofluorescence (B) shows representative images and (C) shows quantification of Wnt5a expression.*

RESULTS

RXC006 displays favourable properties in *in vitro* and *in vivo* ADME assays

- *In vitro* metabolic stability, permeability and free fraction are supportive of oral administration. PK data across preclinical species show good bioavailability and exposure (data not shown).

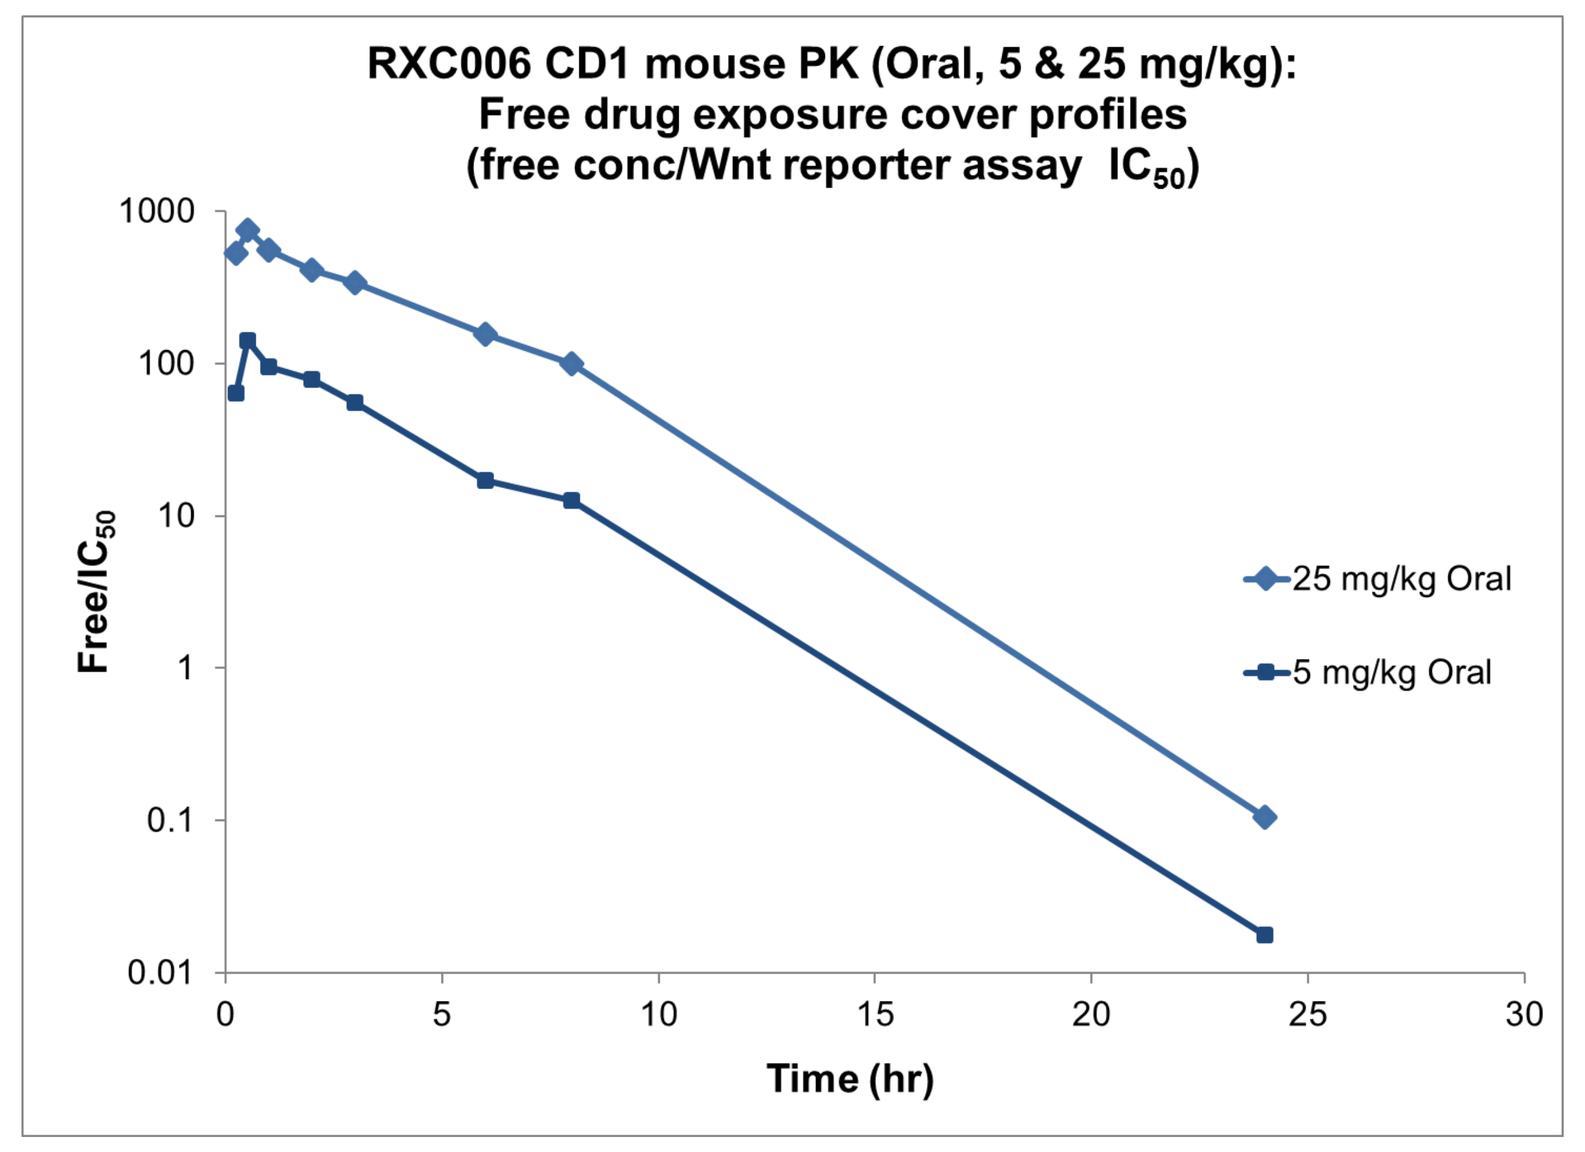
RXC006 CD1 mouse PK (Oral, 5 & 25 mg/kg): Free drug exposure cover profiles (free conc/Wnt reporter assay IC50)
Free/IC50
Time (hr)
25 mg/kg Oral
5 mg/kg Oral

Figure 5. *PK profile of RXC006 in CD-1 mice at 5 and 25 mg/kg oral dose.*

	RXC006
Renzo cellular reporter assay IC_{50} (nM)	0.4
HPAF2 cell proliferation pGI_{50} (nM)	0.8
% free PPB (hu/m)	16 / 3
logD	2.9
Thermodynamic solubility pH 7.4 (mg/L) FaSSIF (pH 6.5) (mg/L) FeSSIF (pH 5.0) (mg/L)	2.2 7 153
CACO2 P_{app} ($x10^{-6}$cm/sec) (AB) / ER (BA/AB) MDCK2-MDR1 P_{app} ($x10^{-6}$cm/sec) (AB) / ER (BA/AB)	15 / 1.7 27/1.5
Hu Clint (mics/heps)	44 / 4
Mouse Clint (mics/heps)	24 / 23
hERG (µM) (IC_{50})	>33

Table 1. *Table of ADME properties for RXC006.*

RXC006 demonstrates efficacy when dosed therapeutically in the UUO model

A: Protocol:

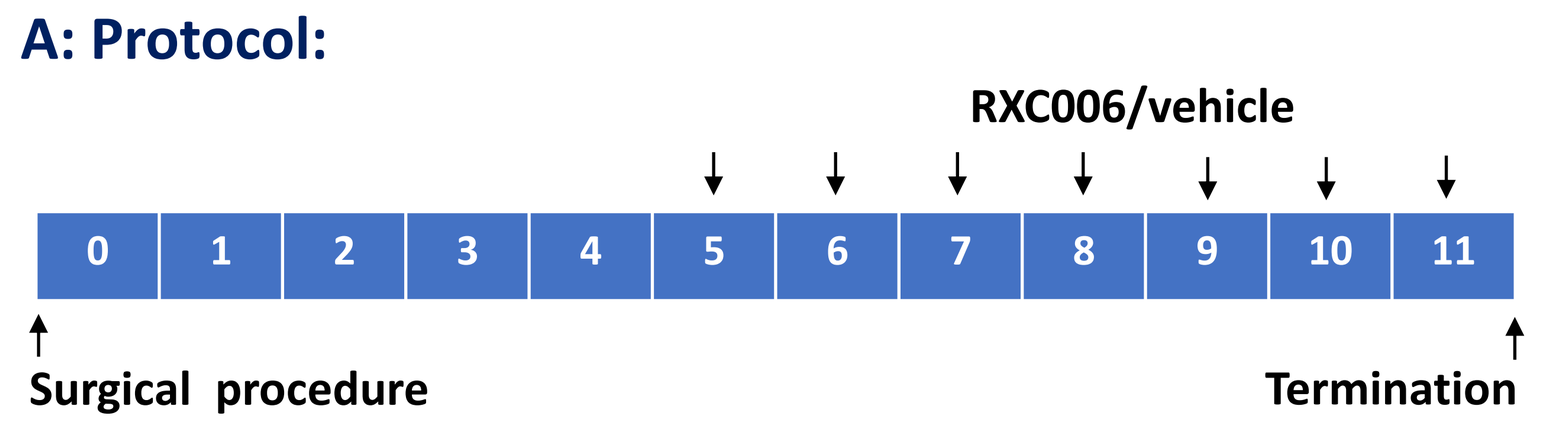
RXC006/vehicle
0 1 2 3 4 5 6 7 8 9 10 11
Surgical procedure
Termination

- RXC006 dosed therapeutically in the UUO model of kidney fibrosis demonstrated significant anti-fibrotic effects.
- Pathway engagement was demonstrated via the suppression of Axin-2 and there was a significant reduction in collagen deposition and pro-fibrotic mediators such as anti-CTGF.

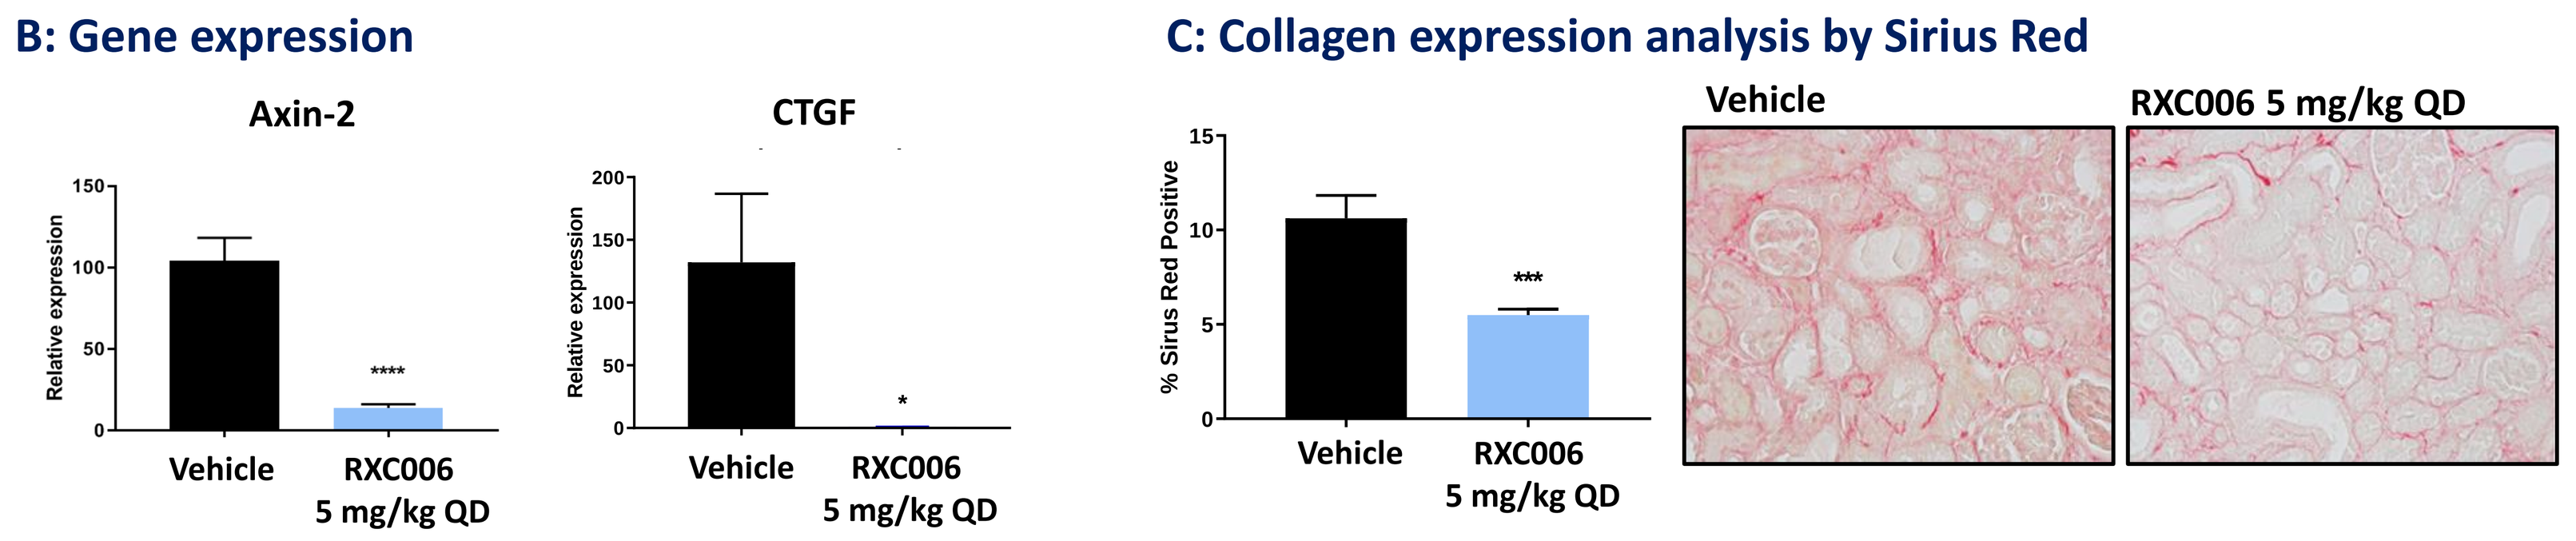
B: Gene expression
Axin-2
CTGF
Relative expression
Vehicle
RXC006 5 mg/kg QD
C: Collagen expression analysis by Sirius Red
% Sirus Red Positive
Vehicle
RXC006 5 mg/kg QD

Figure 6. *C57BL/6 male mice (n=10 per cohort) aged 8-12 weeks were subjected to the a unilateral ureteral obstruction (UUO) procedure, where a ureter from one kidney was tied to induce injury, inflammation and fibrosis in the kidney. After 6 days RXC006 5 mg/kg QD or vehicle was administered by oral gavage in 0.5% CMC + 0.1% tween for a further 6 days (A). (B) shows whole kidney gene expression and (C) collagen expression assessed by Sirius Red histology analysis.*

RXC006 demonstrates therapeutic efficacy in the CCl_4 liver fibrosis model

- Redx porcupine inhibitors RXC006 and RXC004 significantly reduced the increased liver weight caused by repeated administration of CCl_4 suggesting Wnt suppression was able to ameliorate deleterious tissue remodelling caused by hyperplasia and fibrosis.
- Fibrosis suppression was confirmed by the reduction of hydroxyproline content in the liver.

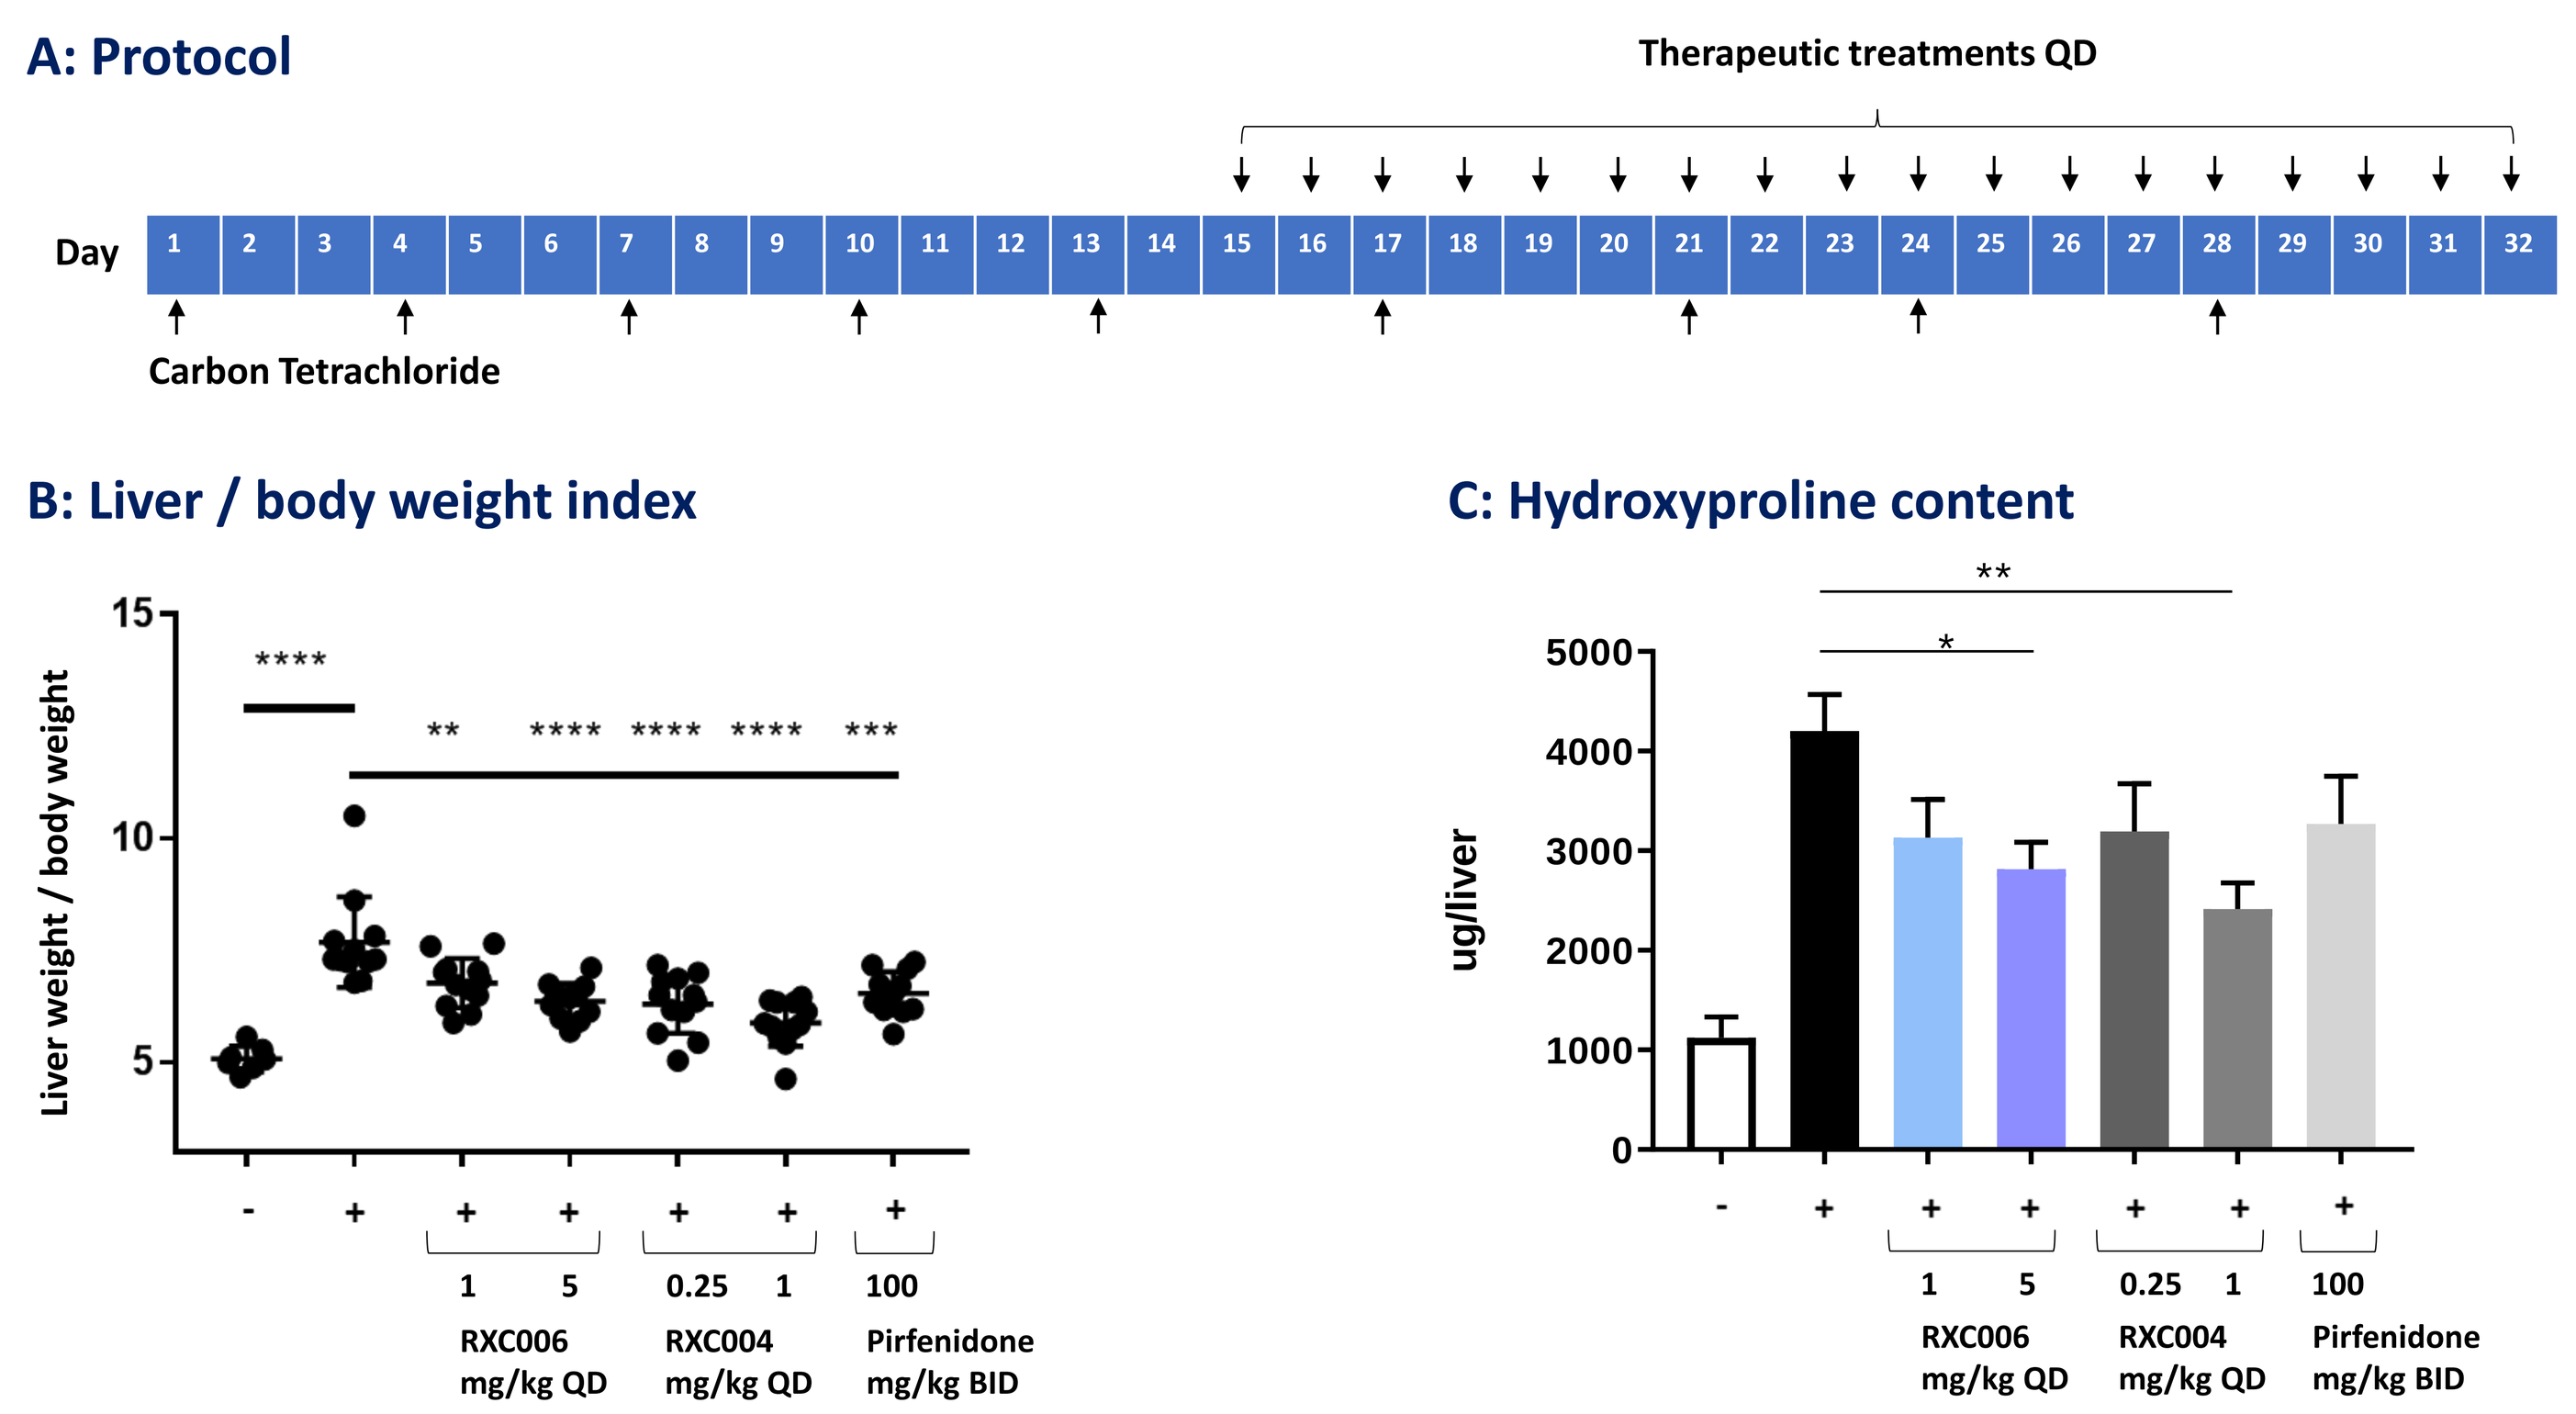
A: Protocol
Therapeutic treatments QD
Day
Carbon Tetrachloride
B: Liver / body weight index
Liver weight / body weight
RXC006 mg/kg QD
RXC004 mg/kg QD
Pirfenidone mg/kg BID
C: Hydroxyproline content
ug/liver

Figure 7 *C57BL/6 male mice aged 10-12 weeks were administered carbon tetrachloride (CCl_4) in mineral oil i.p. 2x per week for 3 weeks, sham mice received 0.9% saline in mineral oil. Compounds were administered BID/QD from day 15 (A) by oral gavage in 0.5% CMC + 0.1% tween. Animals were sacrificed on day 32. Liver to body weight ratio (B) and liver hydroxyproline content (C) were assessed. Data are plotted mean±SEM for n=10 animals.*

RXC006 suppresses fibrosis therapeutically in the lung bleomycin model

- Porcupine inhibitors RXC006 and RXC004 dosed therapeutically demonstrated significant anti-fibrotic effects in the murine bleomycin lung fibrosis model.
- Compounds caused significant reductions in lung weight, collagen deposition and reduction in pro-fibrotic genes - as exemplified by CTGF. Axin-2 and TGFβ gene expression was also reduced (data not shown).
- The anti-fibrotic effect was seen in a therapeutic context when compound was dosed from day 7 when the initial injury and inflammatory phases had subsided.

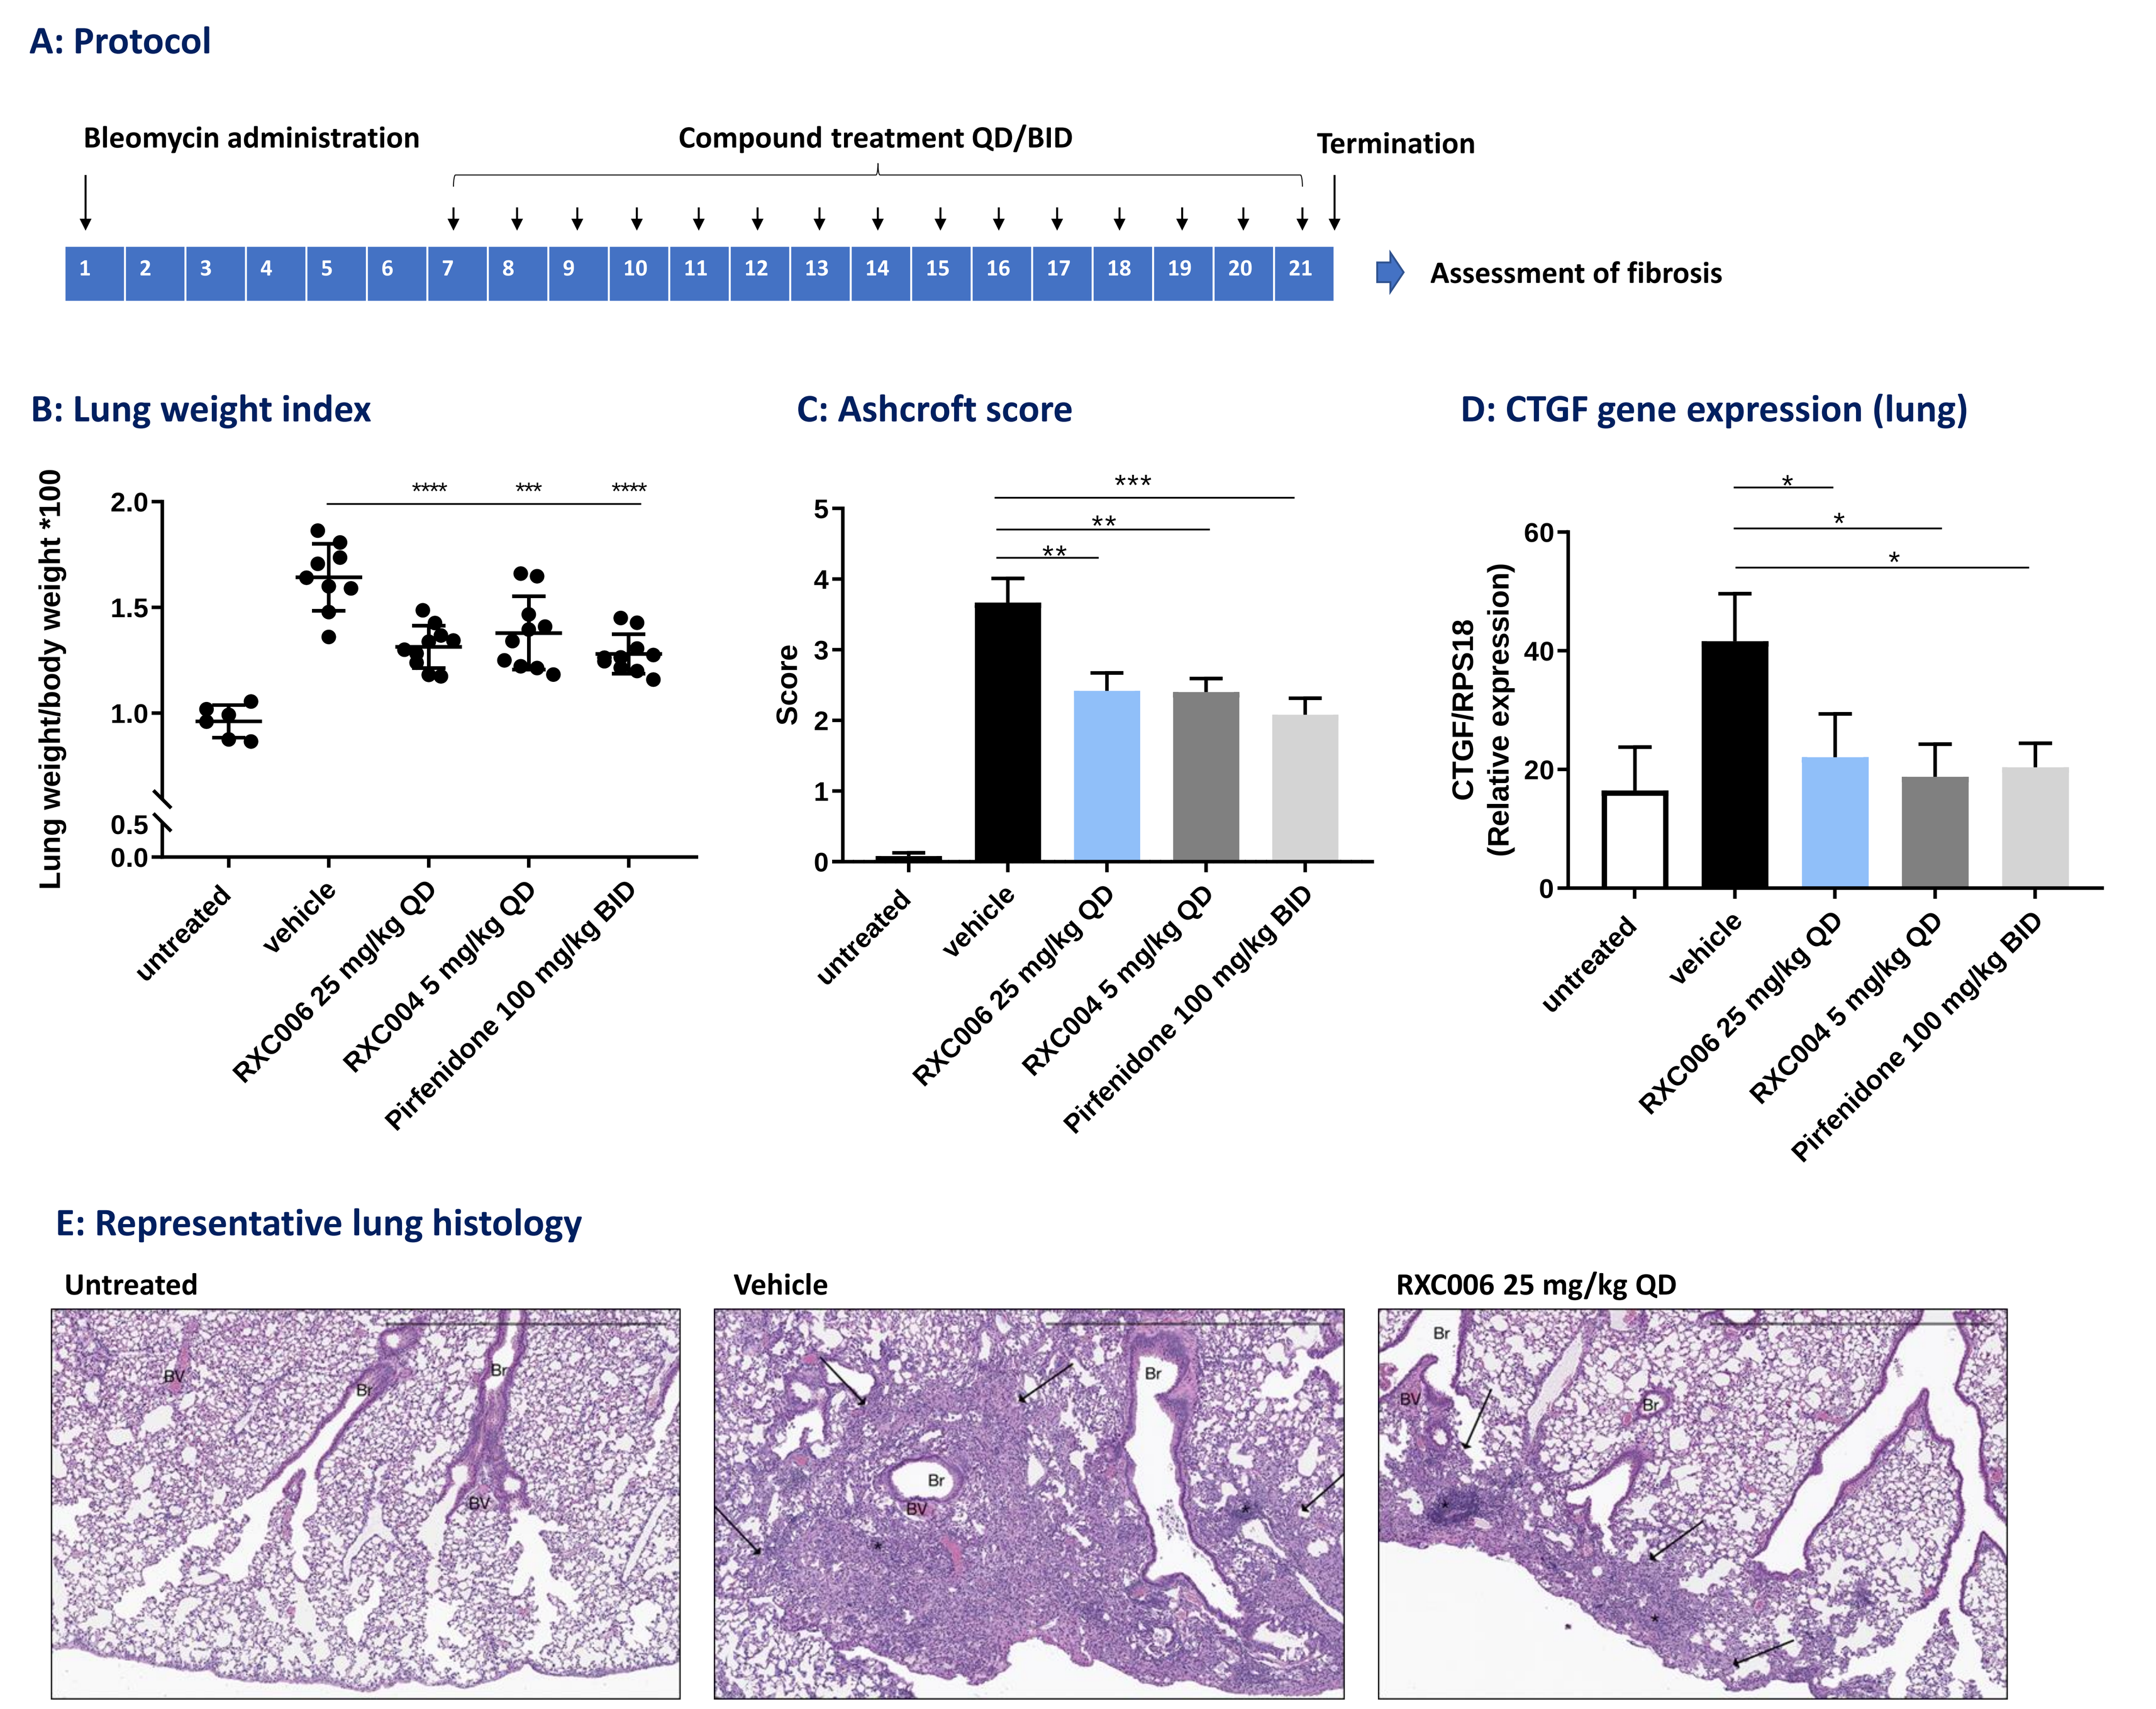
A: Protocol
Bleomycin administration
Compound treatment QD/BID
Termination
Assessment of fibrosis
B: Lung weight index
Lung weight/body weight *100
C: Ashcroft score
Score
D: CTGF gene expression (lung)
CTGF/RPS18 (Relative expression)
untreated
vehicle
RXC006 25 mg/kg QD
RXC004 5 mg/kg QD
Pirfenidone 100 mg/kg BID
E: Representative lung histology
Untreated
Vehicle
RXC006 25 mg/kg QD

Figure 8 *C57BL/6 male mice between the ages of 6 to 8 weeks were given 1.5 U/mL of bleomycin on day 1 via oropharyngeal administration. Compounds were administered by oral gavage in 0.5% CMC + 0.1% tween either QD/BID from day 7 until day 21 (A). Lung weight index (B), Ashcroft score and analysis of CTGF gene expression in the lung (C) are shown plotted mean±SEM for n=10 animals. In (E) histology sections representative of group mean Ashcroft scores are shown. Scale Bar indicates 1.2 mm. Small regions of dense collagenous connective tissue (fibrosis; black arrows) and lymphocyte infiltrates/aggregates (*) are present. A bronchiole (Br) and blood vessels (BV) are present.*

SUMMARY

- Redx porcupine inhibitor RXC006 is a potent suppressor of both canonical and non-canonical signalling pathways.
- RXC006 can potently suppress the release of Wnt3a from L-Wnt3a cells and Wnt5a from HLFs.
- Porcupine inhibition of Wnt3a release into the supernatant reduces the ability of the supernatant to activate HLFs - as shown by reduced α-SMA expression.
- RXC006 potently suppresses collagen deposition in murine models of kidney, liver and lung fibrosis over a range of different concentrations.
- RXC006 has been nominated as a candidate for development in Idiopathic Pulmonary Fibrosis and IND enabling studies have been initiated with first time in human studies expected to begin in 2020.

References: **1.** Nusse R, Varmus H. EMBO J. 2012 Jun 13;31(12):2670-84. **2.** Castellone M, Laukkanen M. Front Biosci (Schol Ed). 2017 Jan 1;9:31-45. **3.** Miao CG, Yang Y. Cell Signal. 2013 Oct;25(10):2069-78. **4.** Herr P, Basler K. Dev Biol. 2012 Jan 15;361(2):392-402. **5.** Moon J, Zhou H. Proc Natl Acad Sci U S A. 2017 Feb 14;114(7):1649-1654. **6.** Chen CW, Beyer C. Ann Rheum Dis. 2016 Apr;76(4):773-778. **7.** Thompson, BA. Cancer Res 2015;75(15 Suppl):Abstract nr 5071. **8.** Madan B 2016 May;89(5):1062-74. doi: 10.1016/j.kint.

Redx Pharma