Abstract No.66

RXC007, a Potent, Highly Selective ROCK2 Inhibitor, Demonstrates Preclinical Efficacy Across Fibrosis Models and Good Phase 1 Safety Profile, Supporting Development in IPF and ILD

Authors: Nicolas E.S. Guisot, Jane Robertson, Helen Timmis, Peter Bunyard, Helen McKeever and Richard Armer

ROCK and ROCK2 Pathway

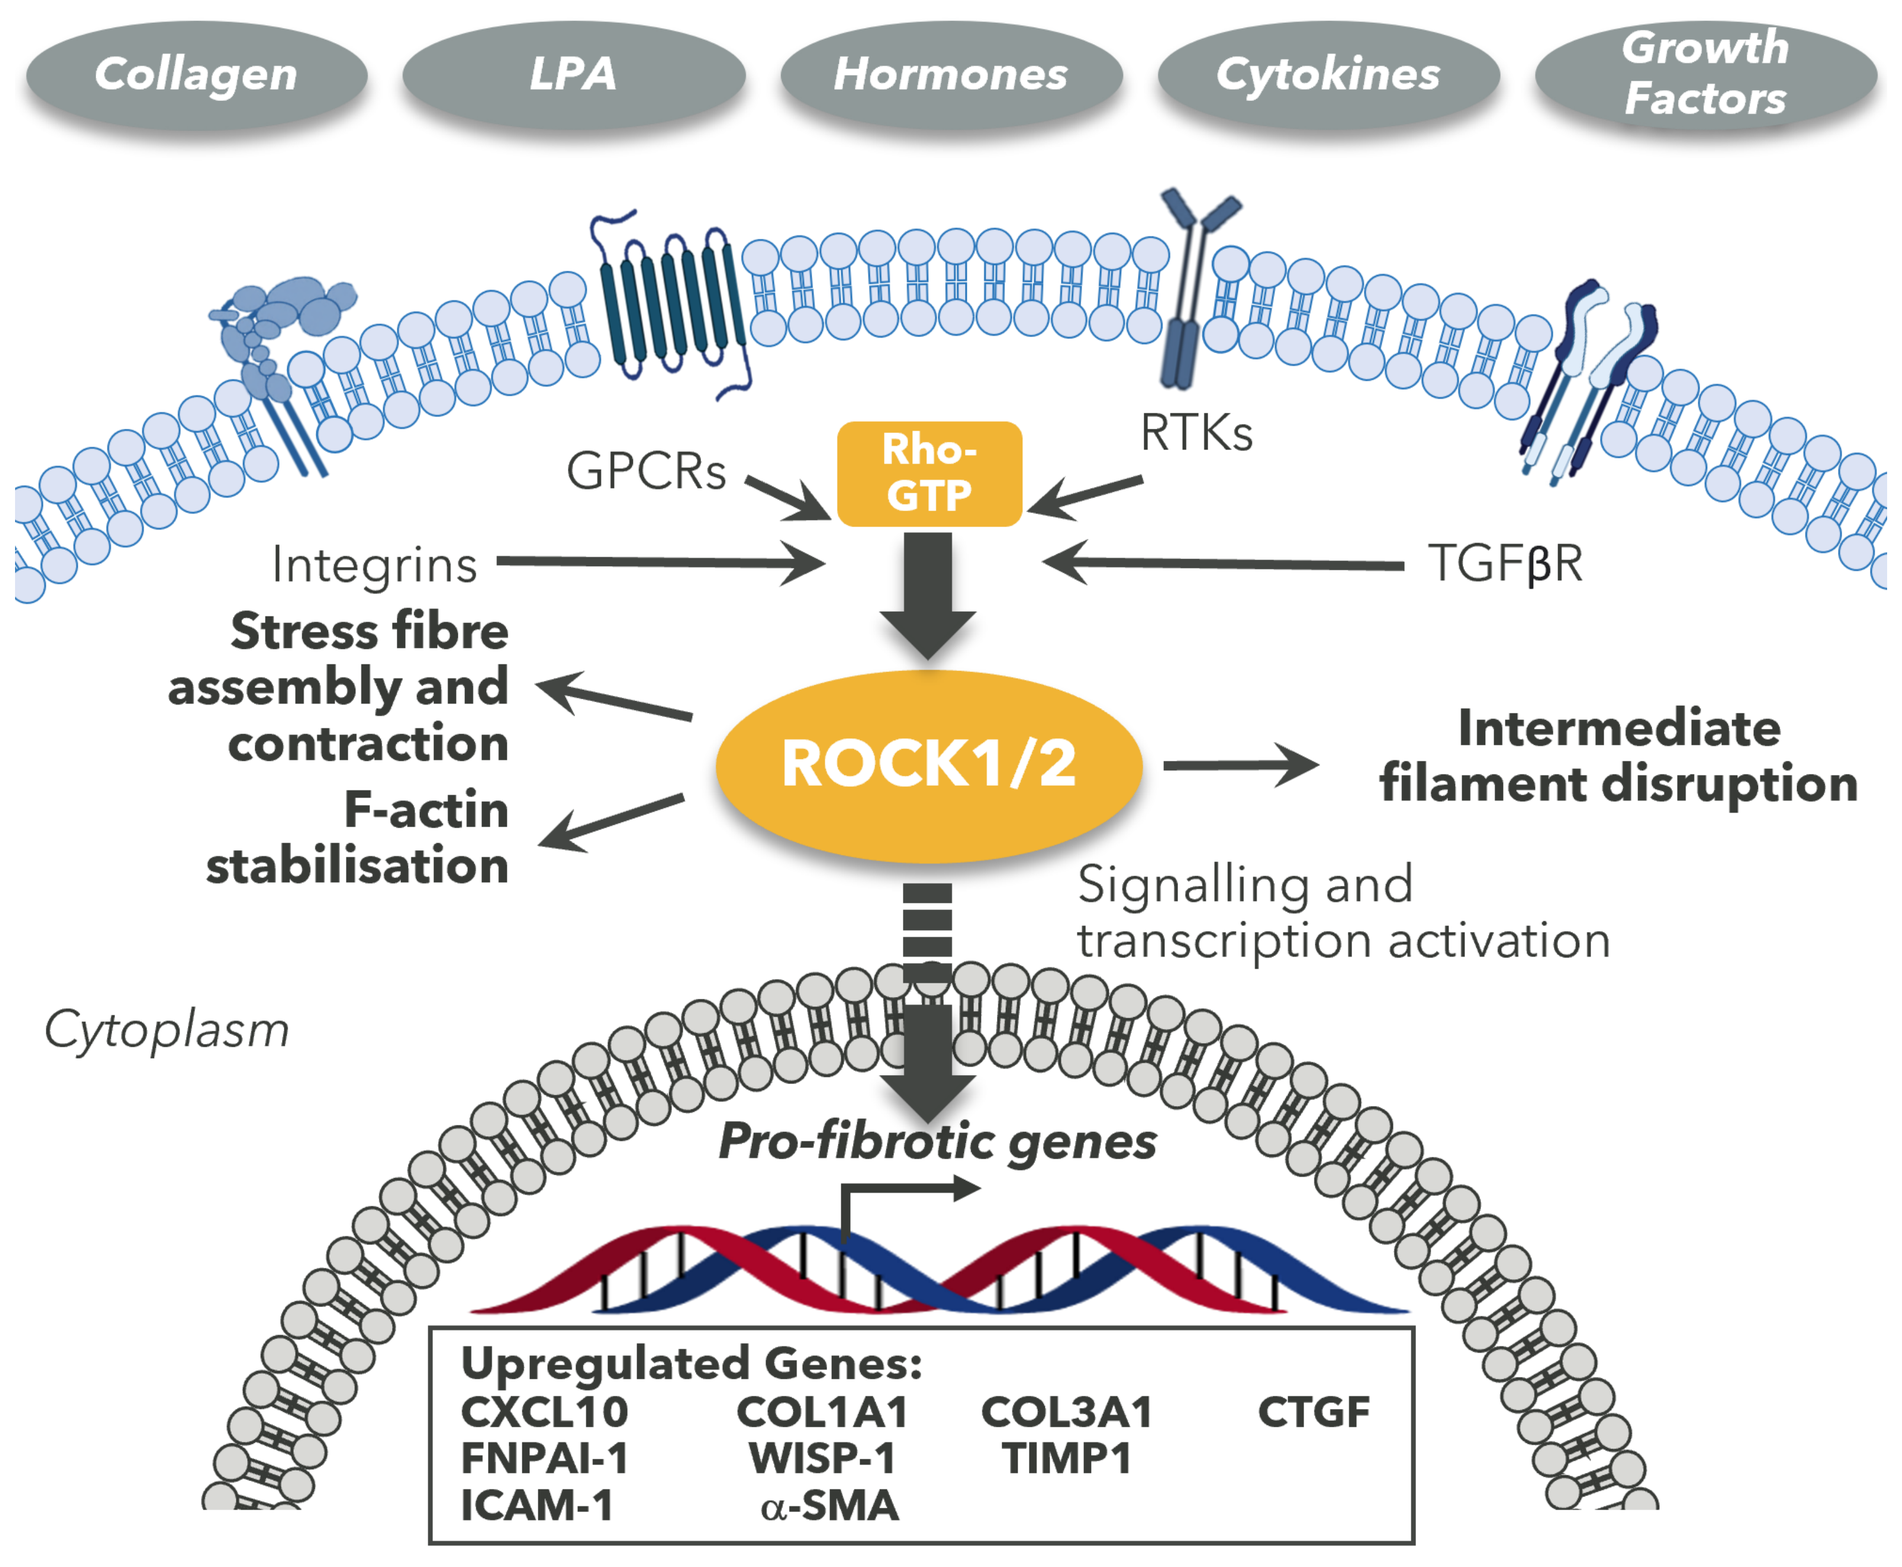
Collagen
LPA
Hormones
Cytokines
Growth Factors
GPCRs
Rho-GTP
RTKs
Integrins
TGFβR
Stress fibre assembly and contraction
F-actin stabilisation
ROCK1/2
Intermediate filament disruption
Signalling and transcription activation
Cytoplasm
Pro-fibrotic genes
Upregulated Genes:
CXCL10 COL1A1 COL3A1 CTGF
FNPAI-1 WISP-1 TIMP1
ICAM-1 α-SMA

- ROCK2 is a kinase that links numerous fibrotic signalling pathways to pro-fibrotic gene expression
- ROCK is involved in diverse cellular processes
- Upregulation of these genes leads to actin cytoskeleton organisation, cell adhesion and motility, proliferation, and extracellular matrix remodelling

Rationale for ROCK2 Inhibition in Interstitial Lung Diseases and Idiopathic Pulmonary Fibrosis

ROCK activity in mouse & human lung tissue

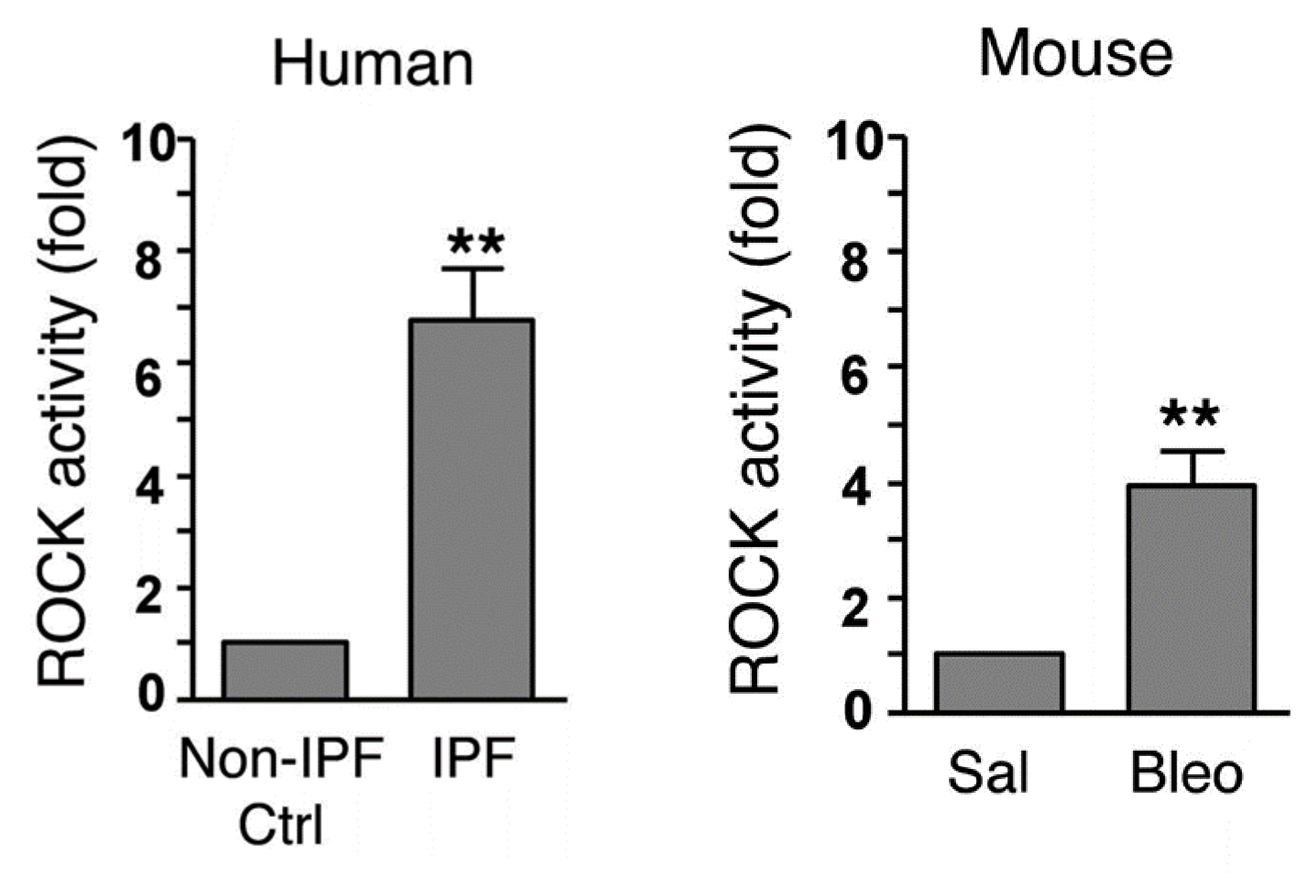
Human
Mouse
ROCK activity (fold)
Non-IPF Ctrl
IPF
Sal
Bleo
**

Fibroblasts & myofibroblasts isolated from lungs of patients with IPF (n = 8), non-IPF control subjects (n = 6)
Fibroblasts & myofibroblasts isolated from lungs of bleomycin-treated mice (n = 8), and saline-treated mice (n = 8)
ROCK activity was measured by exogenous phosphorylation of a substrate peptide

Adapted from Zhou2013[1]

- ROCK activity significantly increased in fibroblasts isolated from human IPF tissue and bleomycin induced murine lung fibrosis

IHC analysis of ROCK2 expression in resected IPF lung tissue

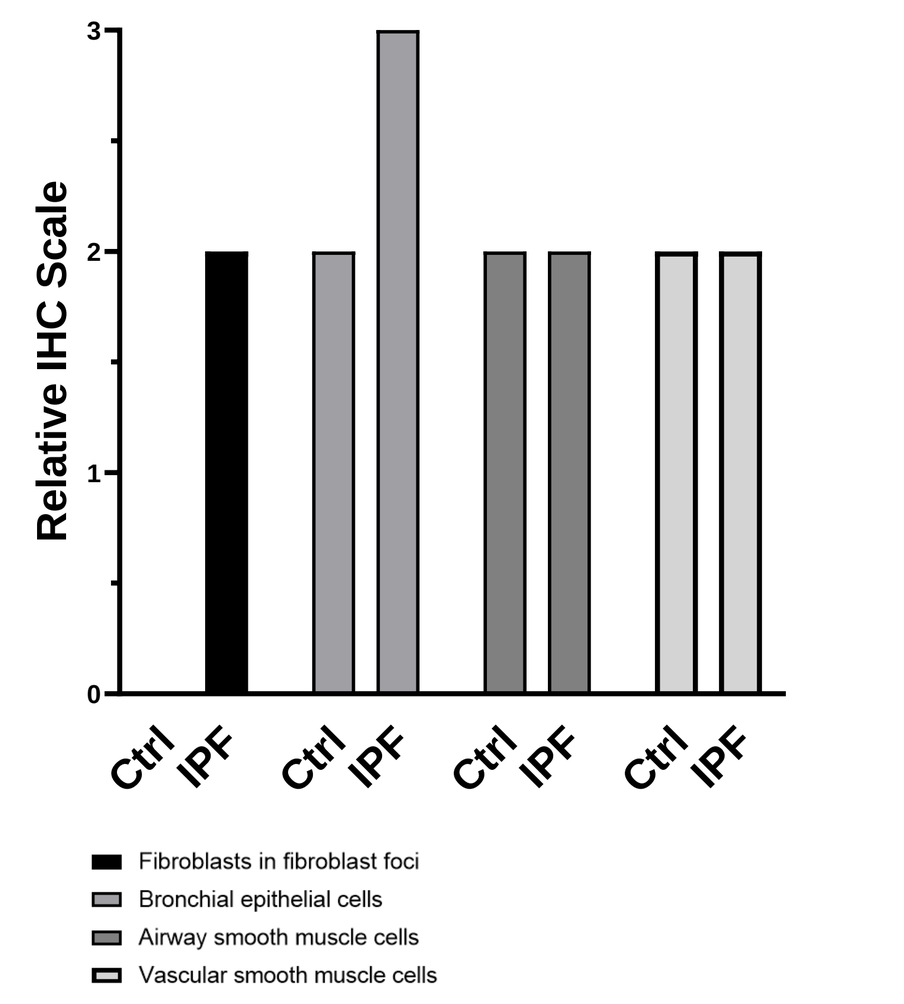
Relative IHC Scale
Ctrl IPF Ctrl IPF Ctrl IPF Ctrl IPF
Fibroblasts in fibroblast foci
Bronchial epithelial cells
Airway smooth muscle cells
Vascular smooth muscle cells

Intensity of immunostaining evaluated on a scale of 0 to 3
N=6 per group; NF: not found

Adapted from Shimizu2014[2]

- IPF fibrotic foci fibroblasts & bronchial epithelial cells show significant increases in ROCK2 expression
- ROCK2 expression is not increased in airway or vascular smooth muscle cells

Single cell IPF cell atlas data

Banovich/Kropski database

Disease	Subjects
cHP	2
Control	10
IPF	12
NSIP	3
sarcoidosis	2
Unclassifiable ILD	1

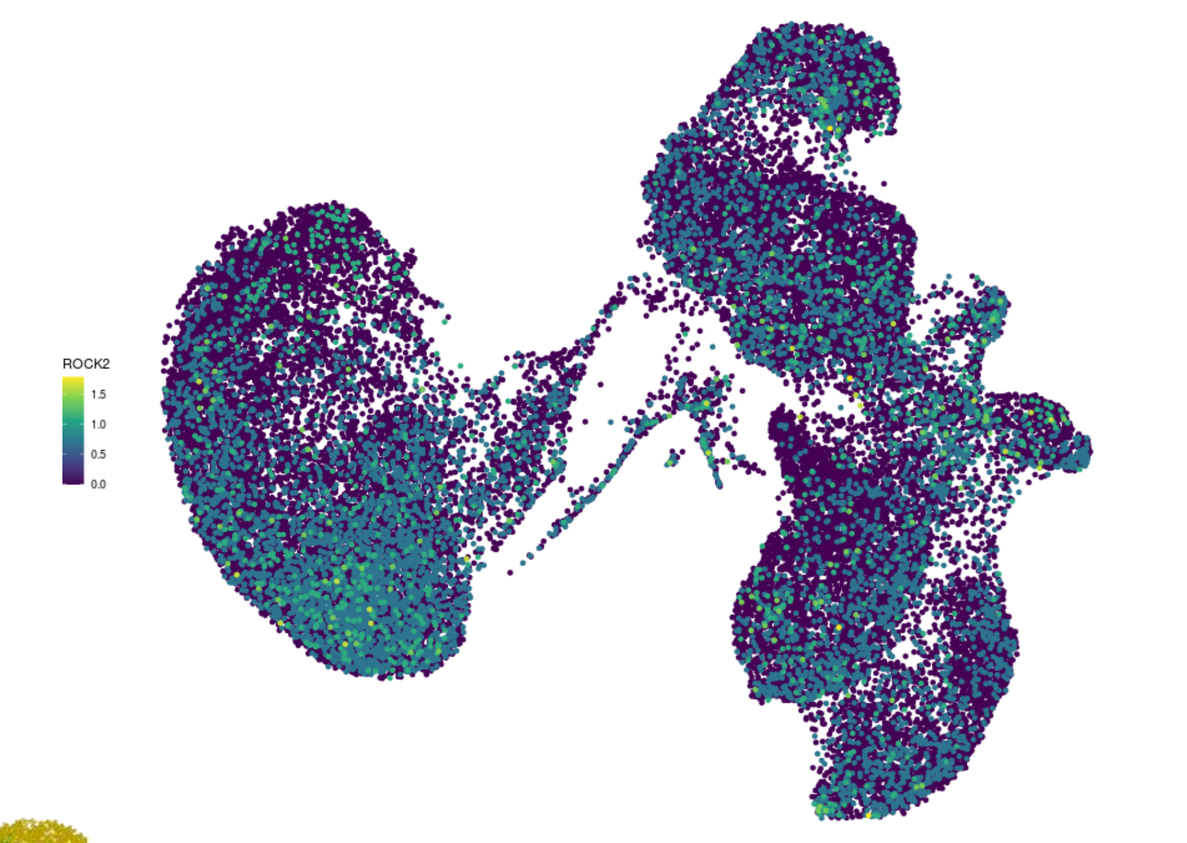
ROCK2

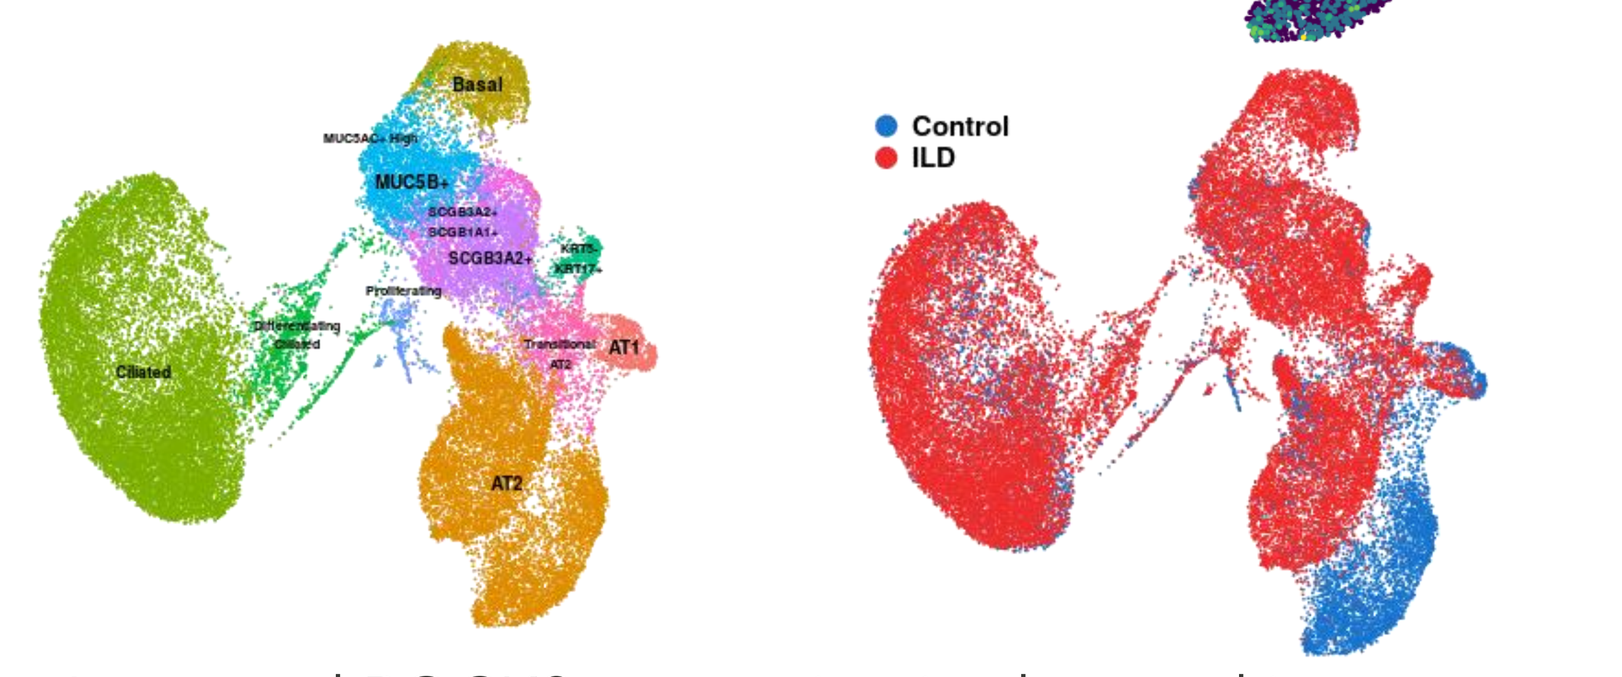
Basal
Ciliated
AT2
AT1
Control
ILD

- Increased ROCK2 expression is observed in ciliated bronchial epithelial cells

cHP: Chronic hypersensitivity pneumonitis; ILD: Interstitial lung disease; IPF: Idiopathic pulmonary fibrosis; NSIP: Nonspecific interstitial pneumonia; ROCK: Rho associated protein kinase

Preclinical Murine Validation of ROCK2 Inhibition

ROCK2 reduction is sufficient to protect from bleomycin-induced pulmonary fibrosis in mouse models[3]

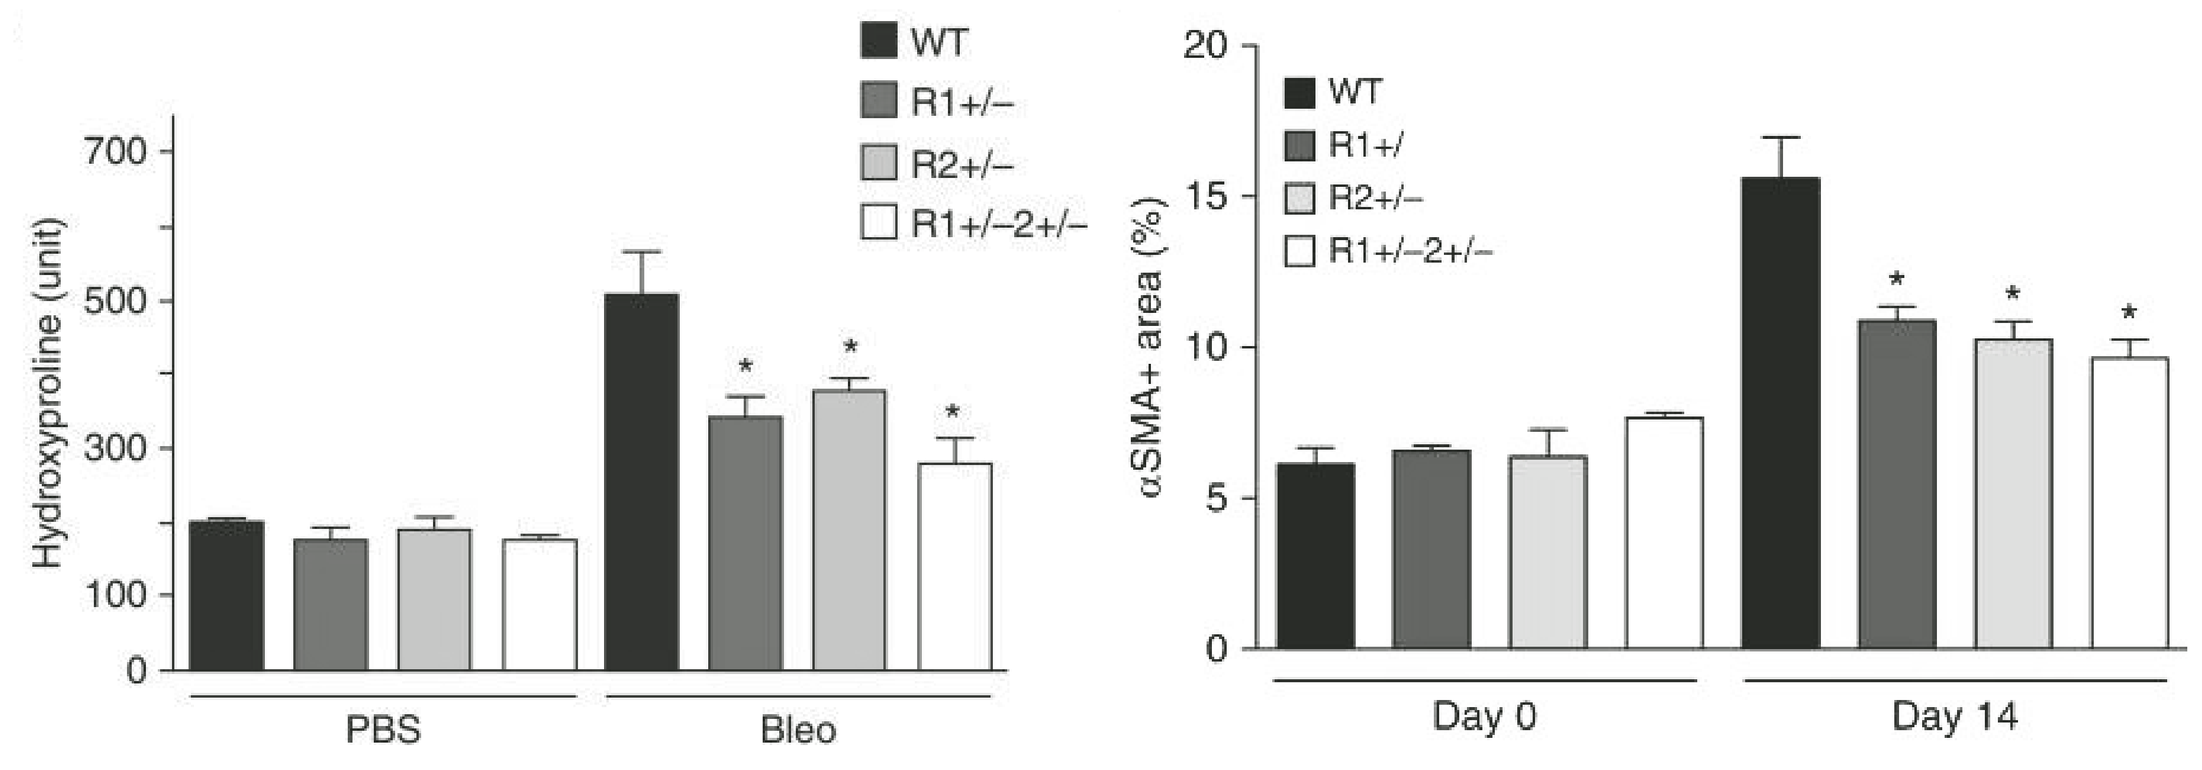
Hydroxyproline (unit)
αSMA+ area (%)
WT
R1+/–
R2+/–
R1+/–2+/–
PBS
Bleo
Day 0
Day 14

- Haploinsufficient ROCK1, ROCK2 and ROCK1/ROCK2 mice show reduced hydroxyproline expression in bleomycin induced lung fibrosis at day 14
- Similar reduction in α-SMA expression in fibroblasts is observed in the 3 haploinsufficient phenotypes

RXC007 Reduces Fibrosis and Collagen Deposition in a Therapeutic Murine Bleomycin-induced Lung Fibrosis

Ashcroft score / Mason's trichrome / Picrosirius red

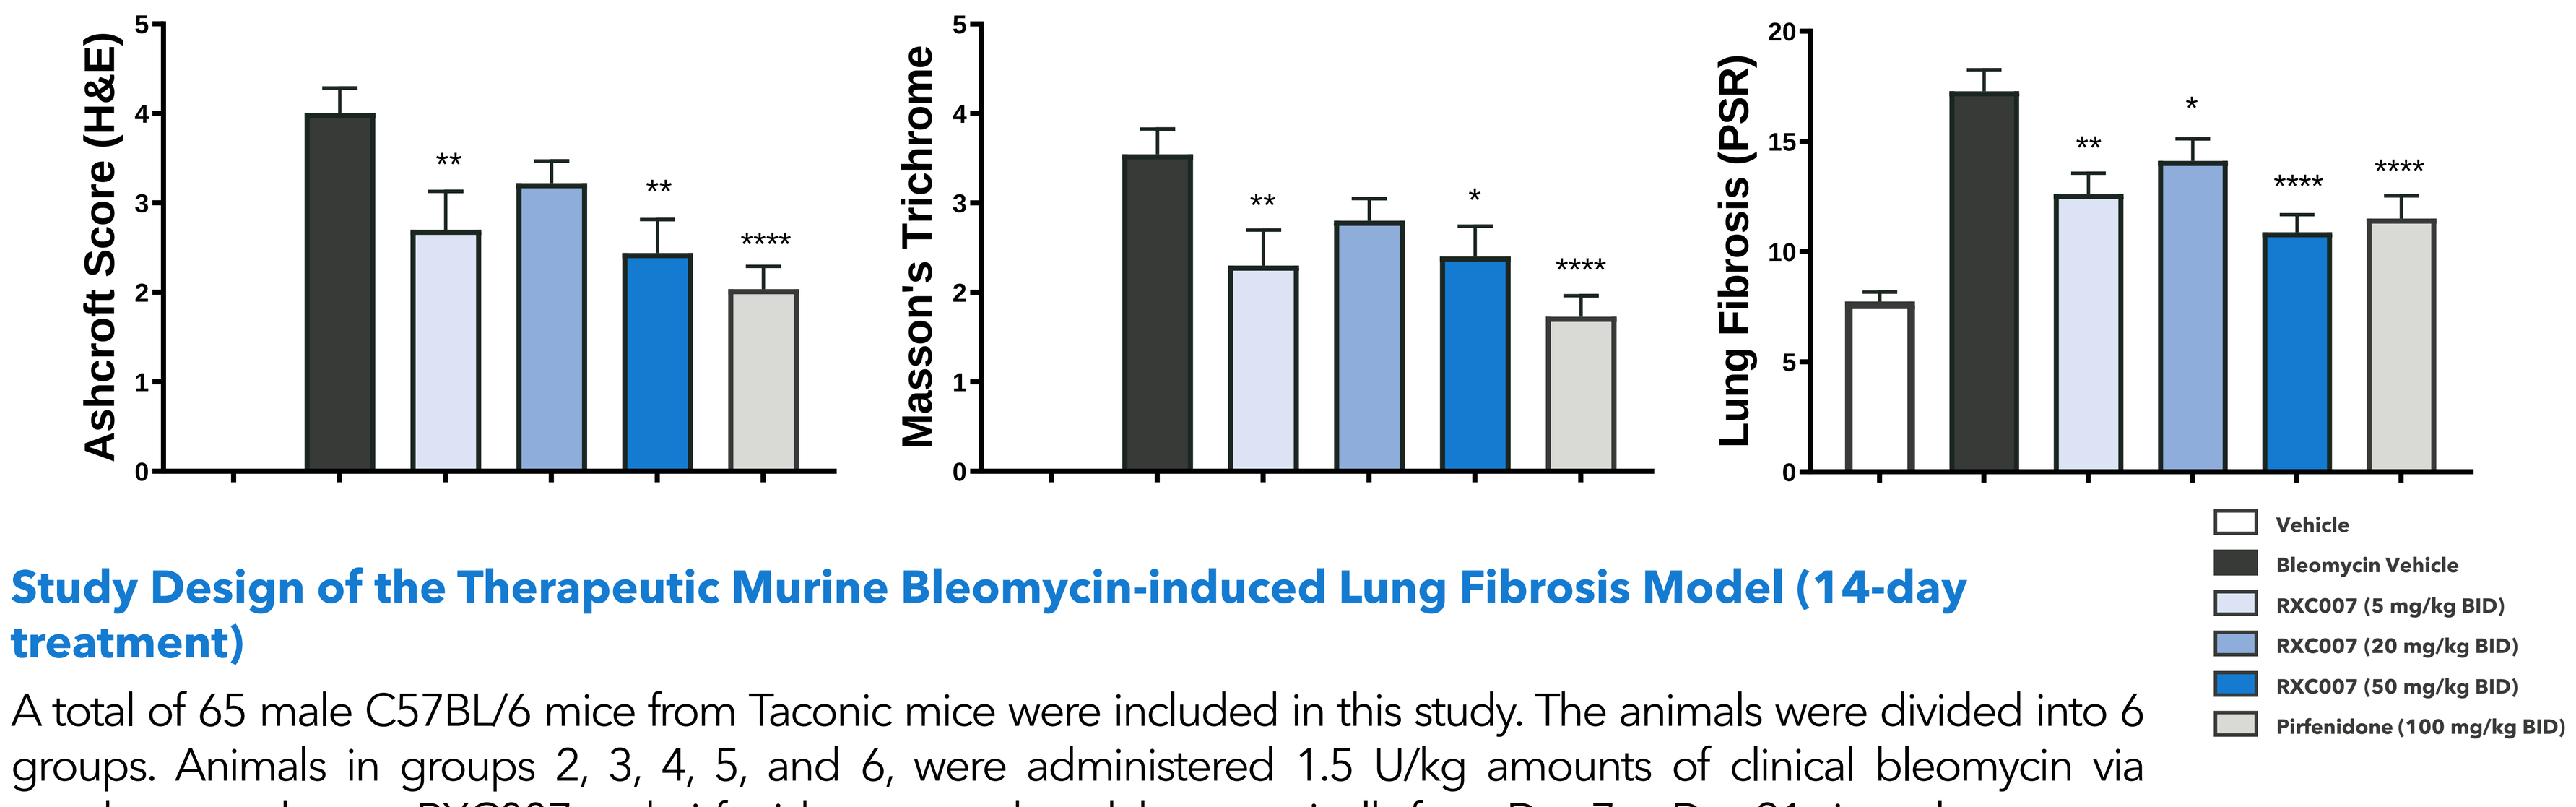
Ashcroft Score (H&E)
Masson's Trichrome
Lung Fibrosis (PSR)
Vehicle
Bleomycin Vehicle
RXC007 (5 mg/kg BID)
RXC007 (20 mg/kg BID)
RXC007 (50 mg/kg BID)
Pirfenidone (100 mg/kg BID)

Study Design of the Therapeutic Murine Bleomycin-induced Lung Fibrosis Model (14-day treatment)

A total of 65 male C57BL/6 mice from Taconic mice were included in this study. The animals were divided into 6 groups. Animals in groups 2, 3, 4, 5, and 6, were administered 1.5 U/kg amounts of clinical bleomycin via oropharyngeal route. RXC007 and pirfenidone were dosed therapeutically from Day 7 to Day 21 via oral gavage.

Suppression of the expression of genes strongly associated with fibrosis in IPF

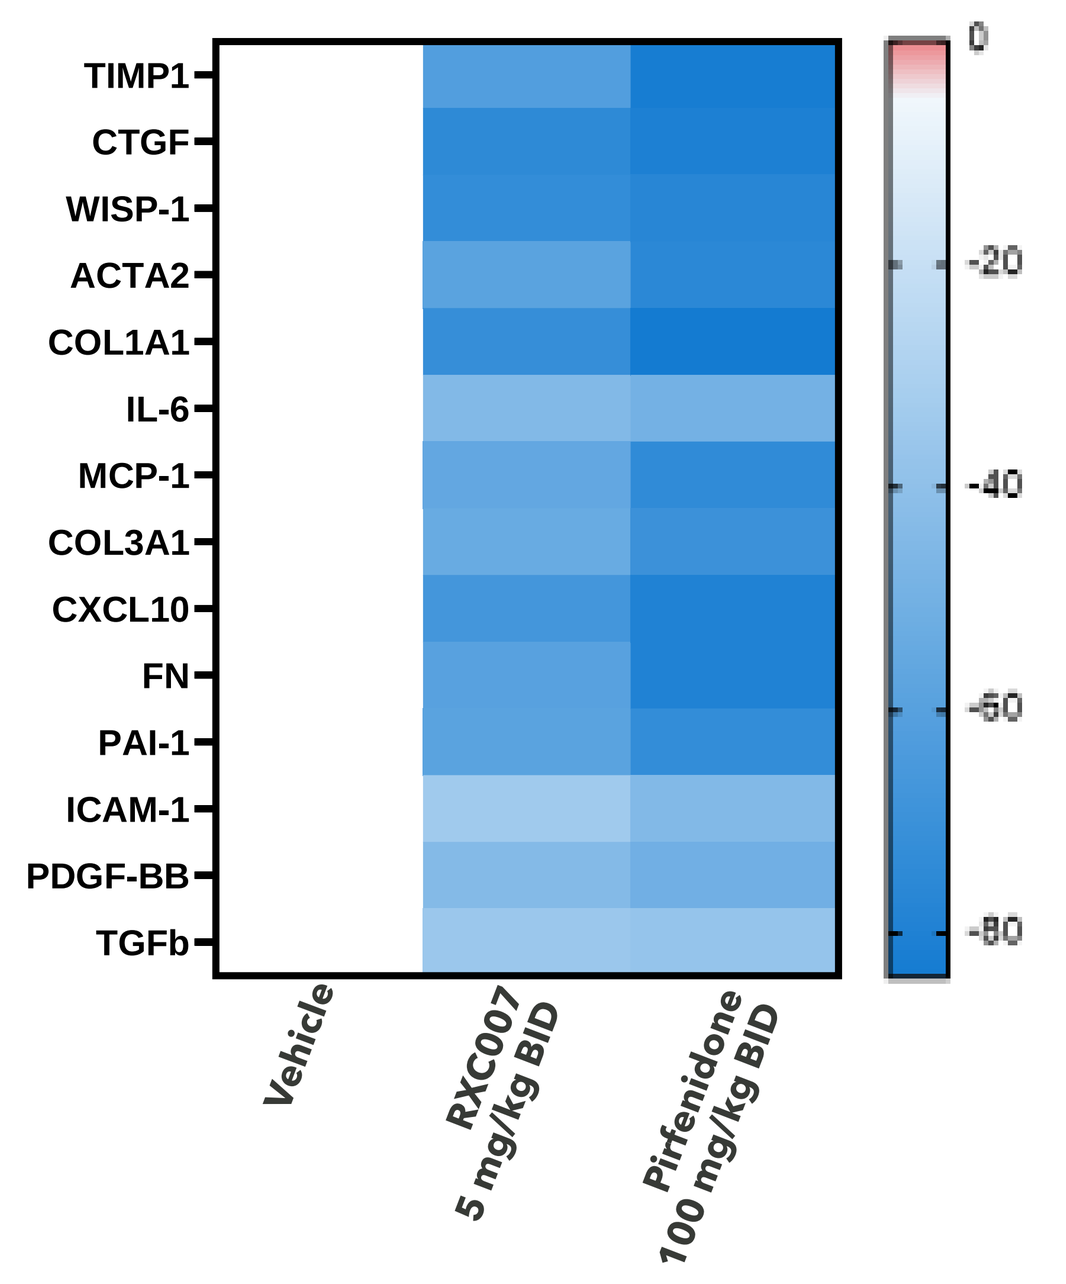
TIMP1
CTGF
WISP-1
ACTA2
COL1A1
IL-6
MCP-1
COL3A1
CXCL10
FN
PAI-1
ICAM-1
PDGF-BB
TGFb
Vehicle
RXC007 5 mg/kg BID
Pirfenidone 100 mg/kg BID

Pleiotropic Effects of RXC007 in a Murine Sclerodermatous chronic Graft versus Host Model

Significant reduction of collagen content and collagen deposition in the skin following administration of RXC007

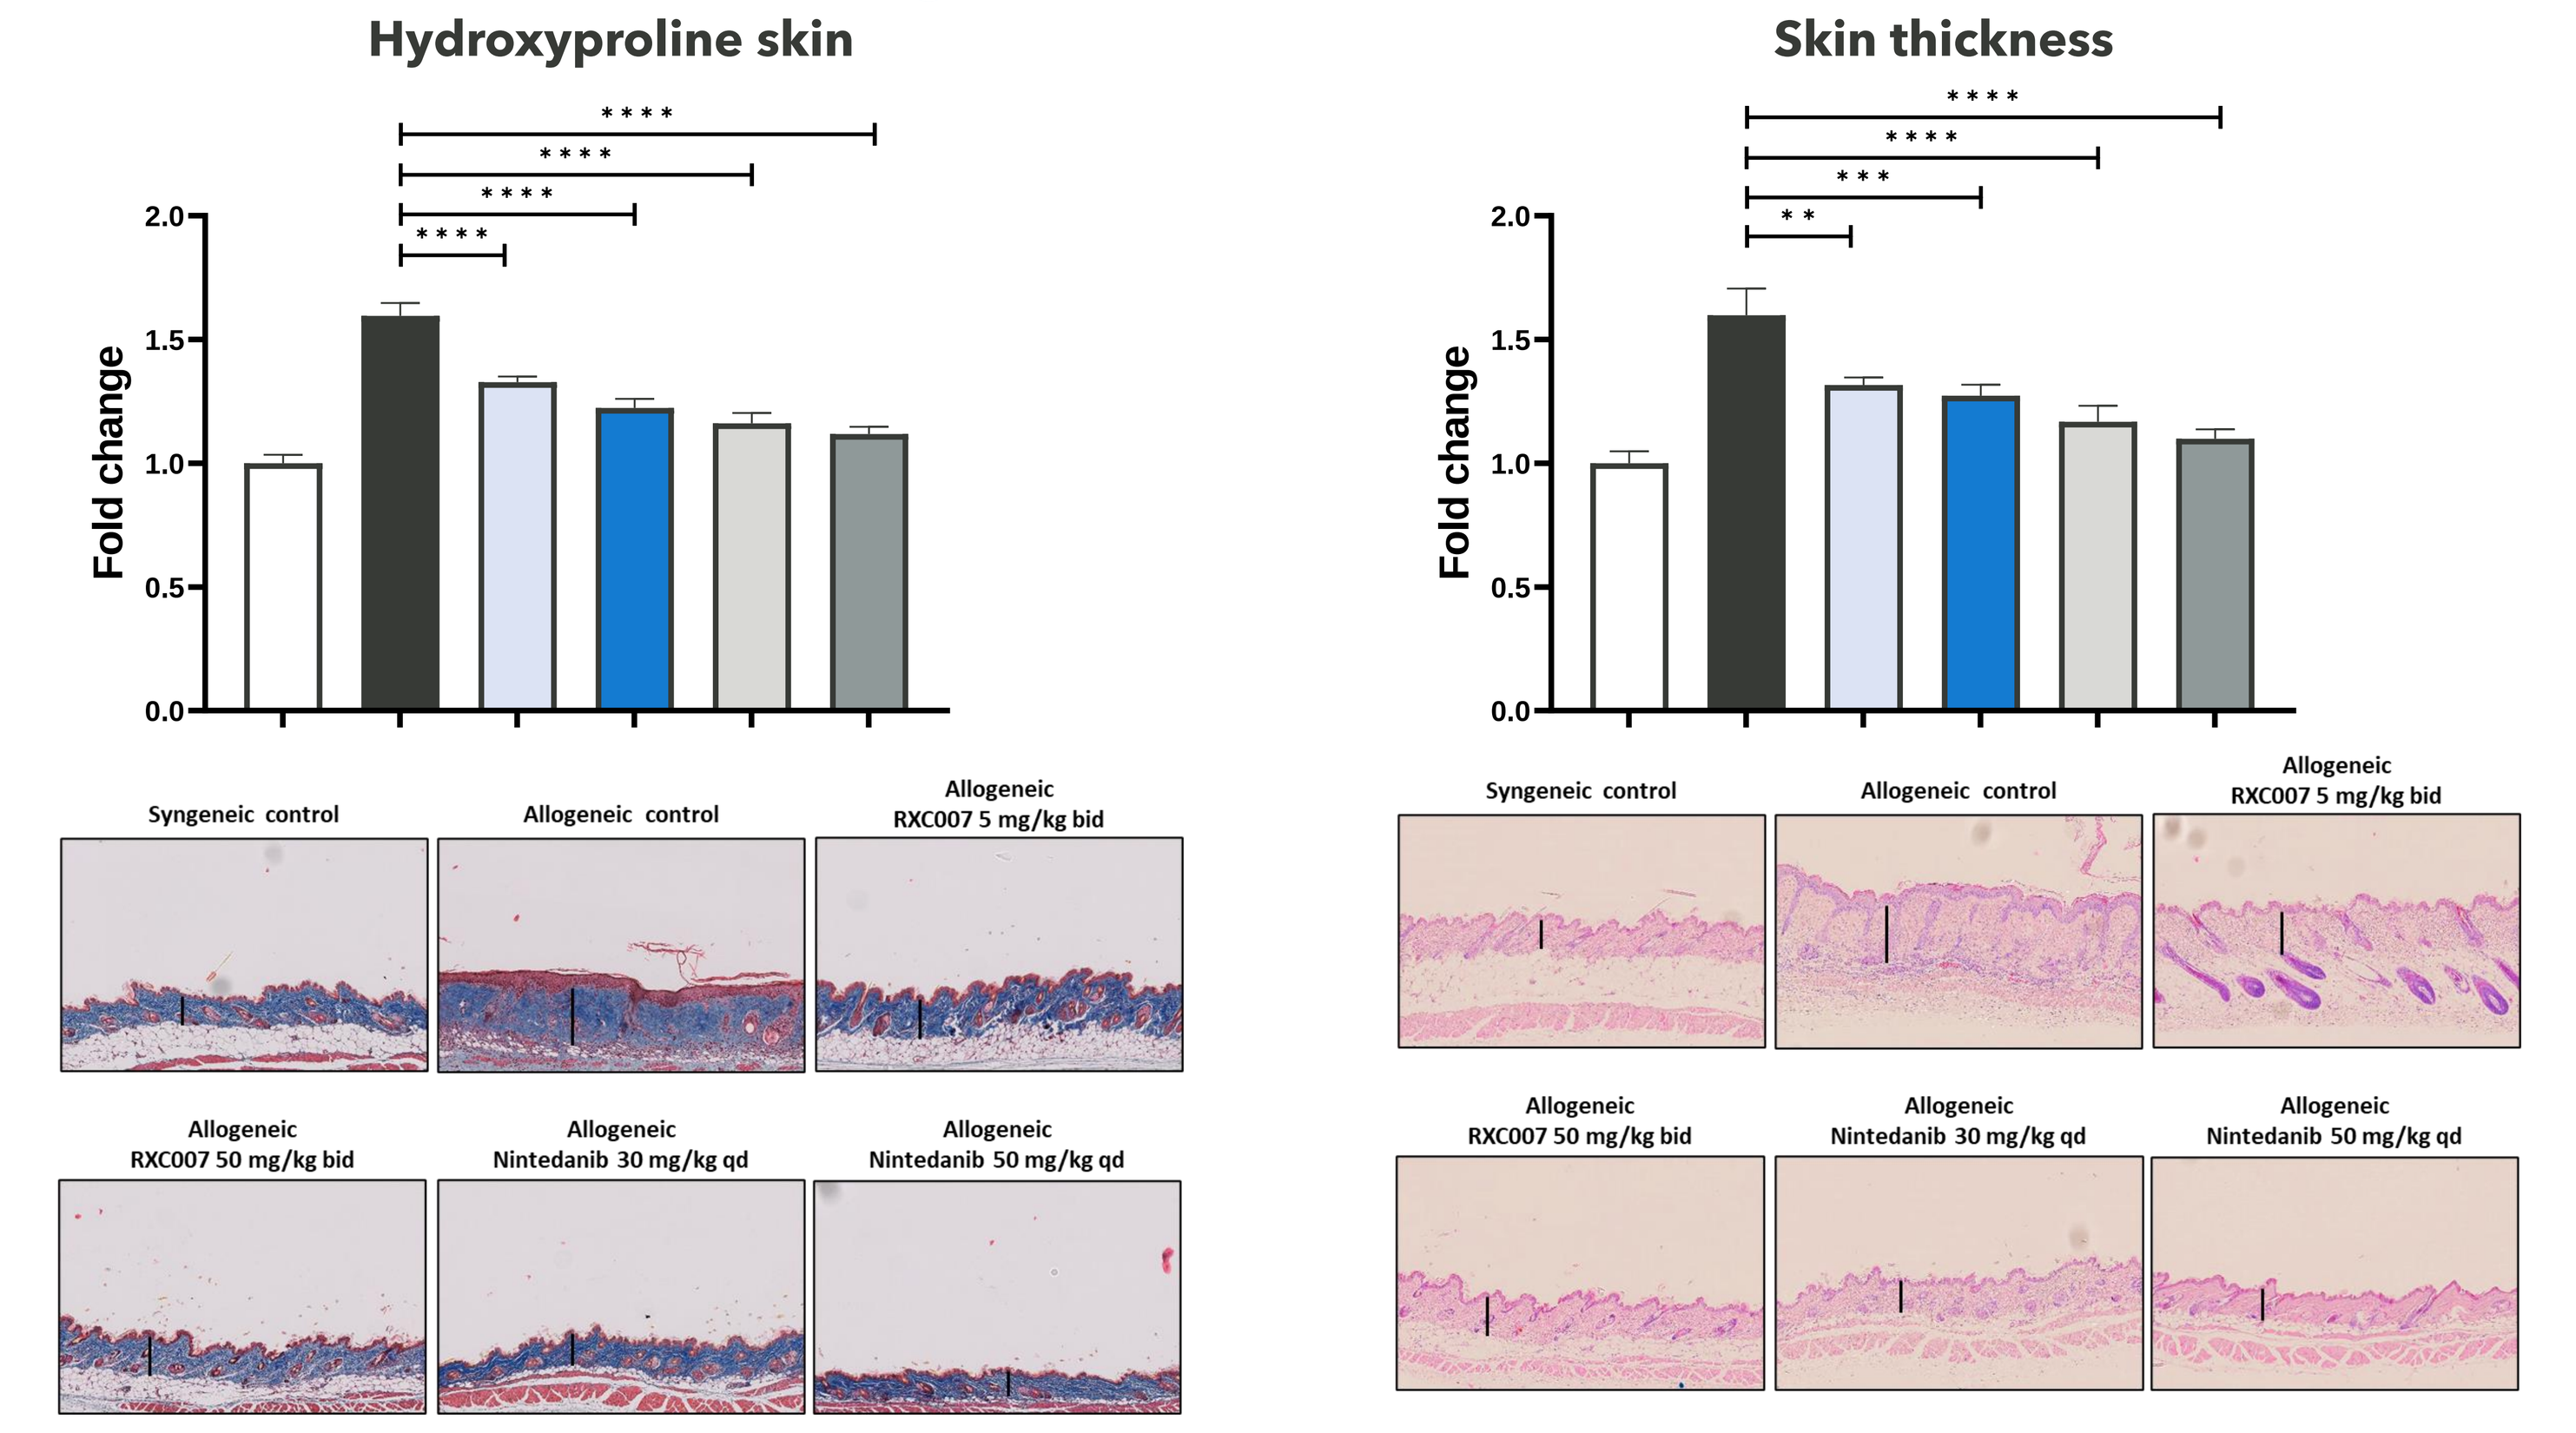
Hydroxyproline skin
Skin thickness
Fold change
Syngeneic control
Allogeneic control
Allogeneic RXC007 5 mg/kg bid
Allogeneic RXC007 50 mg/kg bid
Allogeneic Nintedanib 30 mg/kg qd
Allogeneic Nintedanib 50 mg/kg qd

Similar effect on the cutaneous cGVHD score between nintedanib and RXC007

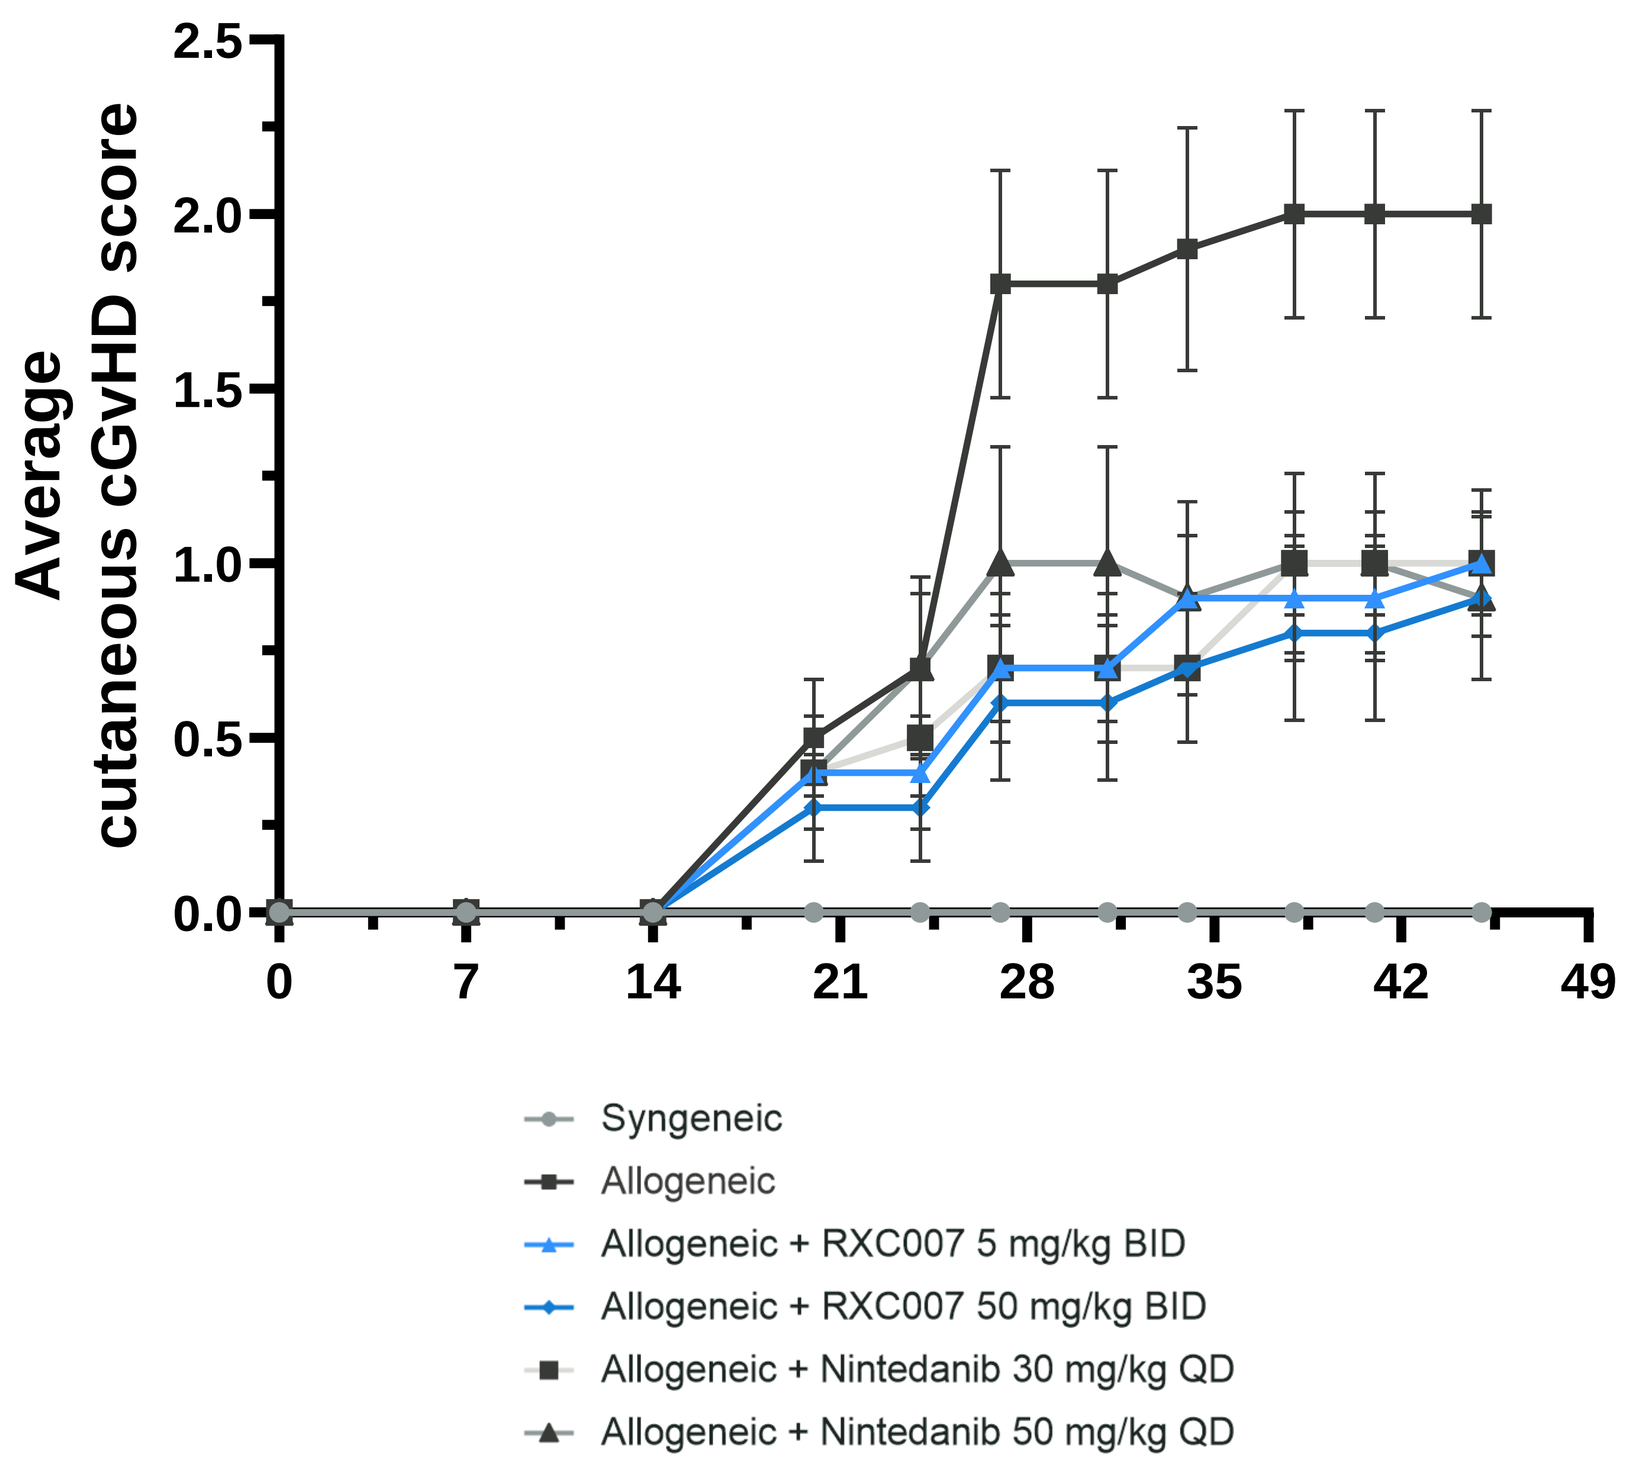
Average cutaneous cGvHD score
Syngeneic
Allogeneic
Allogeneic + RXC007 5 mg/kg BID
Allogeneic + RXC007 50 mg/kg BID
Allogeneic + Nintedanib 30 mg/kg QD
Allogeneic + Nintedanib 50 mg/kg QD

Significant reduction of fibrosis score and collagen content in the lungs around the bronchioles following administration of RXC007

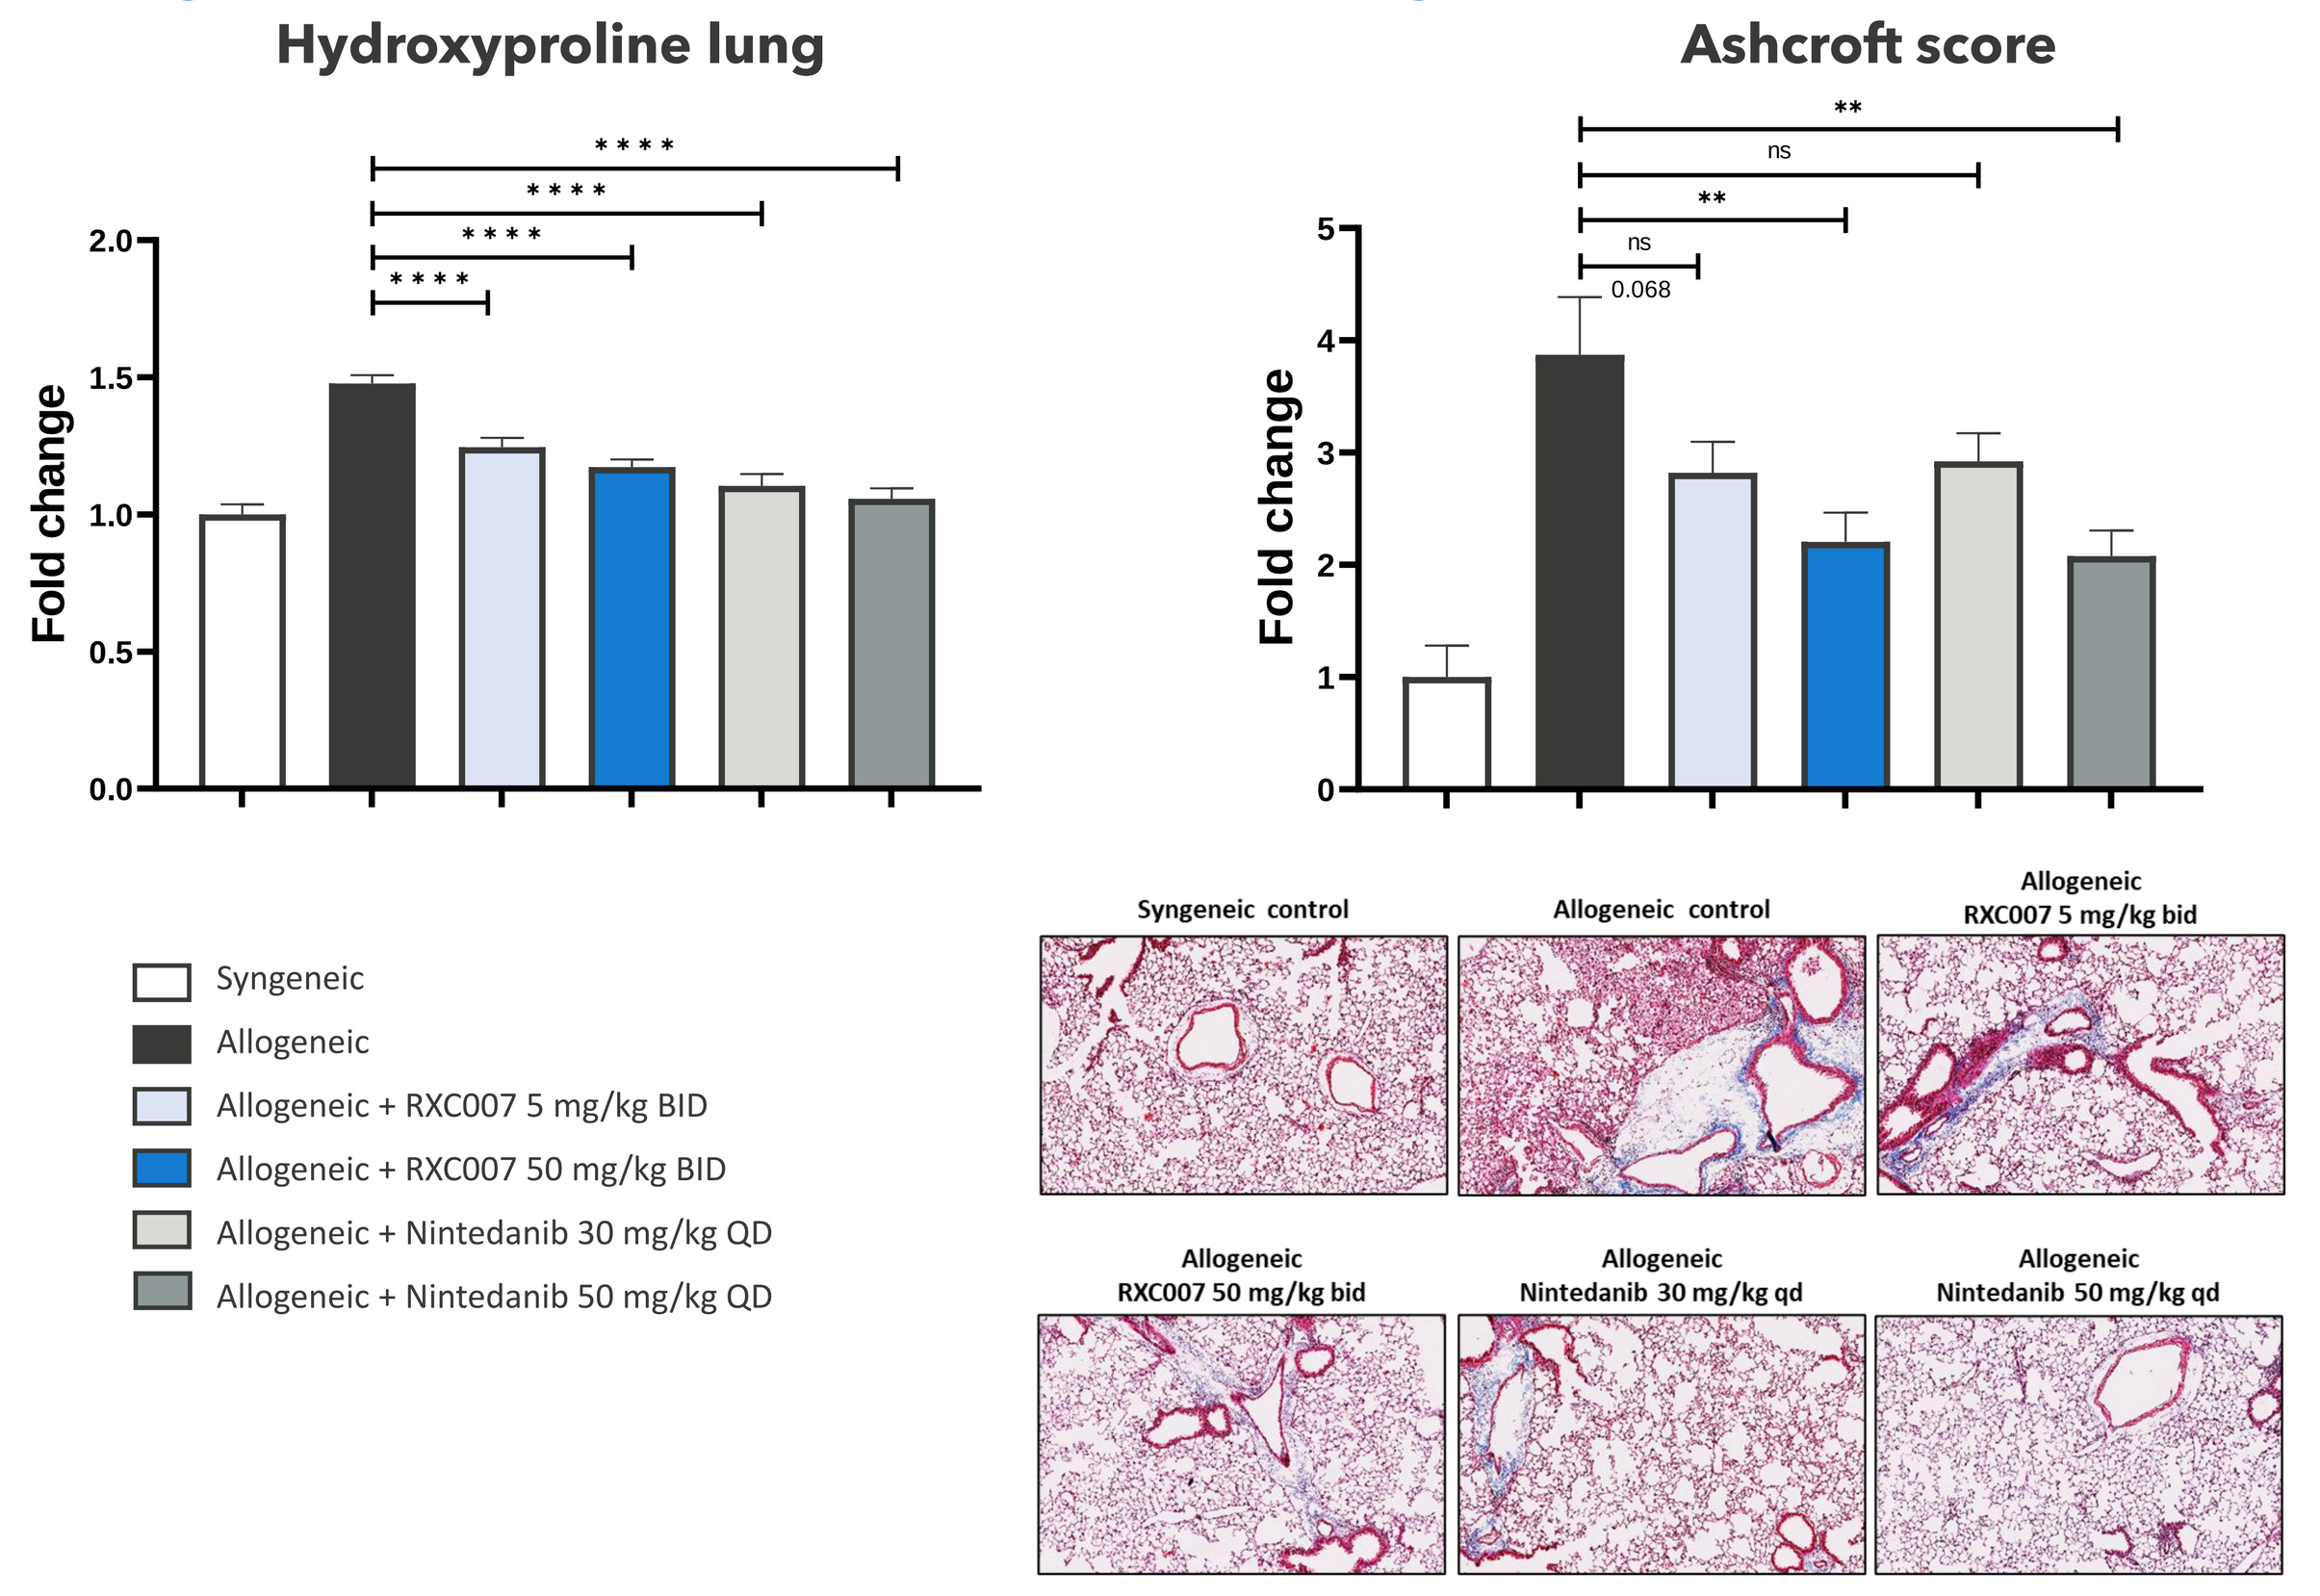
Hydroxyproline lung
Ashcroft score
Fold change
Syngeneic
Allogeneic
Allogeneic + RXC007 5 mg/kg BID
Allogeneic + RXC007 50 mg/kg BID
Allogeneic + Nintedanib 30 mg/kg QD
Allogeneic + Nintedanib 50 mg/kg QD
Syngeneic control
Allogeneic control
Allogeneic RXC007 5 mg/kg bid
Allogeneic RXC007 50 mg/kg bid
Allogeneic Nintedanib 30 mg/kg qd
Allogeneic Nintedanib 50 mg/kg qd

Study Design of the B10.D2→Balb/c (H-2(d)) minor histocompatibility antigen-mismatched model (28-day treatment)

Recipient mice (BALB/c (H-2d)) of an age of 8 weeks received total body irradiation with 700 cGy. Six hours after irradiation, all BALB/c (H-2d) recipients received bone marrow from either BALB/c (H-2d) in a syngeneic or B10.D2 (H-2d) in an allogeneic transplantation manner. For transplantation, 5 x 10^6 splenocytes and 2 x 10^6 bone marrow cells from donor mice were resuspended in 0.2 mL of PBS and injected via tail veins. To reflect the clinical situation with treatment only upon clinical signs of cGvHD, administration of RXC007 and nintedanib started 21 days after bone marrow transplantation and thus several days after the first clinically detectable manifestations of cGvHD in allogeneically transplanted mice. RXC007 and nintedanib administered by oral gavage BID for 28 days.

RXC007/0001 – A Randomised, Double-blind, Placebo-controlled, Single-ascending Dose and Multiple Dose, First-in-human, Phase 1

Study Design

Double-blind, placebo-controlled, to assess the safety and tolerability of RXC007 in healthy subject cohorts following:

- Single dose: Part 1 (SAD) – Dose escalation cohorts
- Multiple doses (14 days): Part 2 (MD) – Dose expansion cohort

Good Safety Profile

- No SAEs reported in SAD and multidose cohorts
- Safe and well tolerated with few treatment emergent adverse events reported
- All AEs transient, mild and reversible with no dose changes required
- No evidence of hypotension, validating rationale for selective ROCK2 inhibition

Drug-like Pharmacokinetic Profile

- Pharmacokinetics as predicted from preclinical data
- Mean half-life 9–11 hours, potentially suitable for once-daily dosing
- Essentially linear exposure in SAD from 2 mg QD to 70 mg BID
- No significant differences between 50 mg fed and fasted cohorts
- Achieved biologically relevant exposures from 20 mg BID and above based on preclinical models

RXC007_0001 Single Ascending Dose cohorts / RXC007_0001 Multi-dose cohort

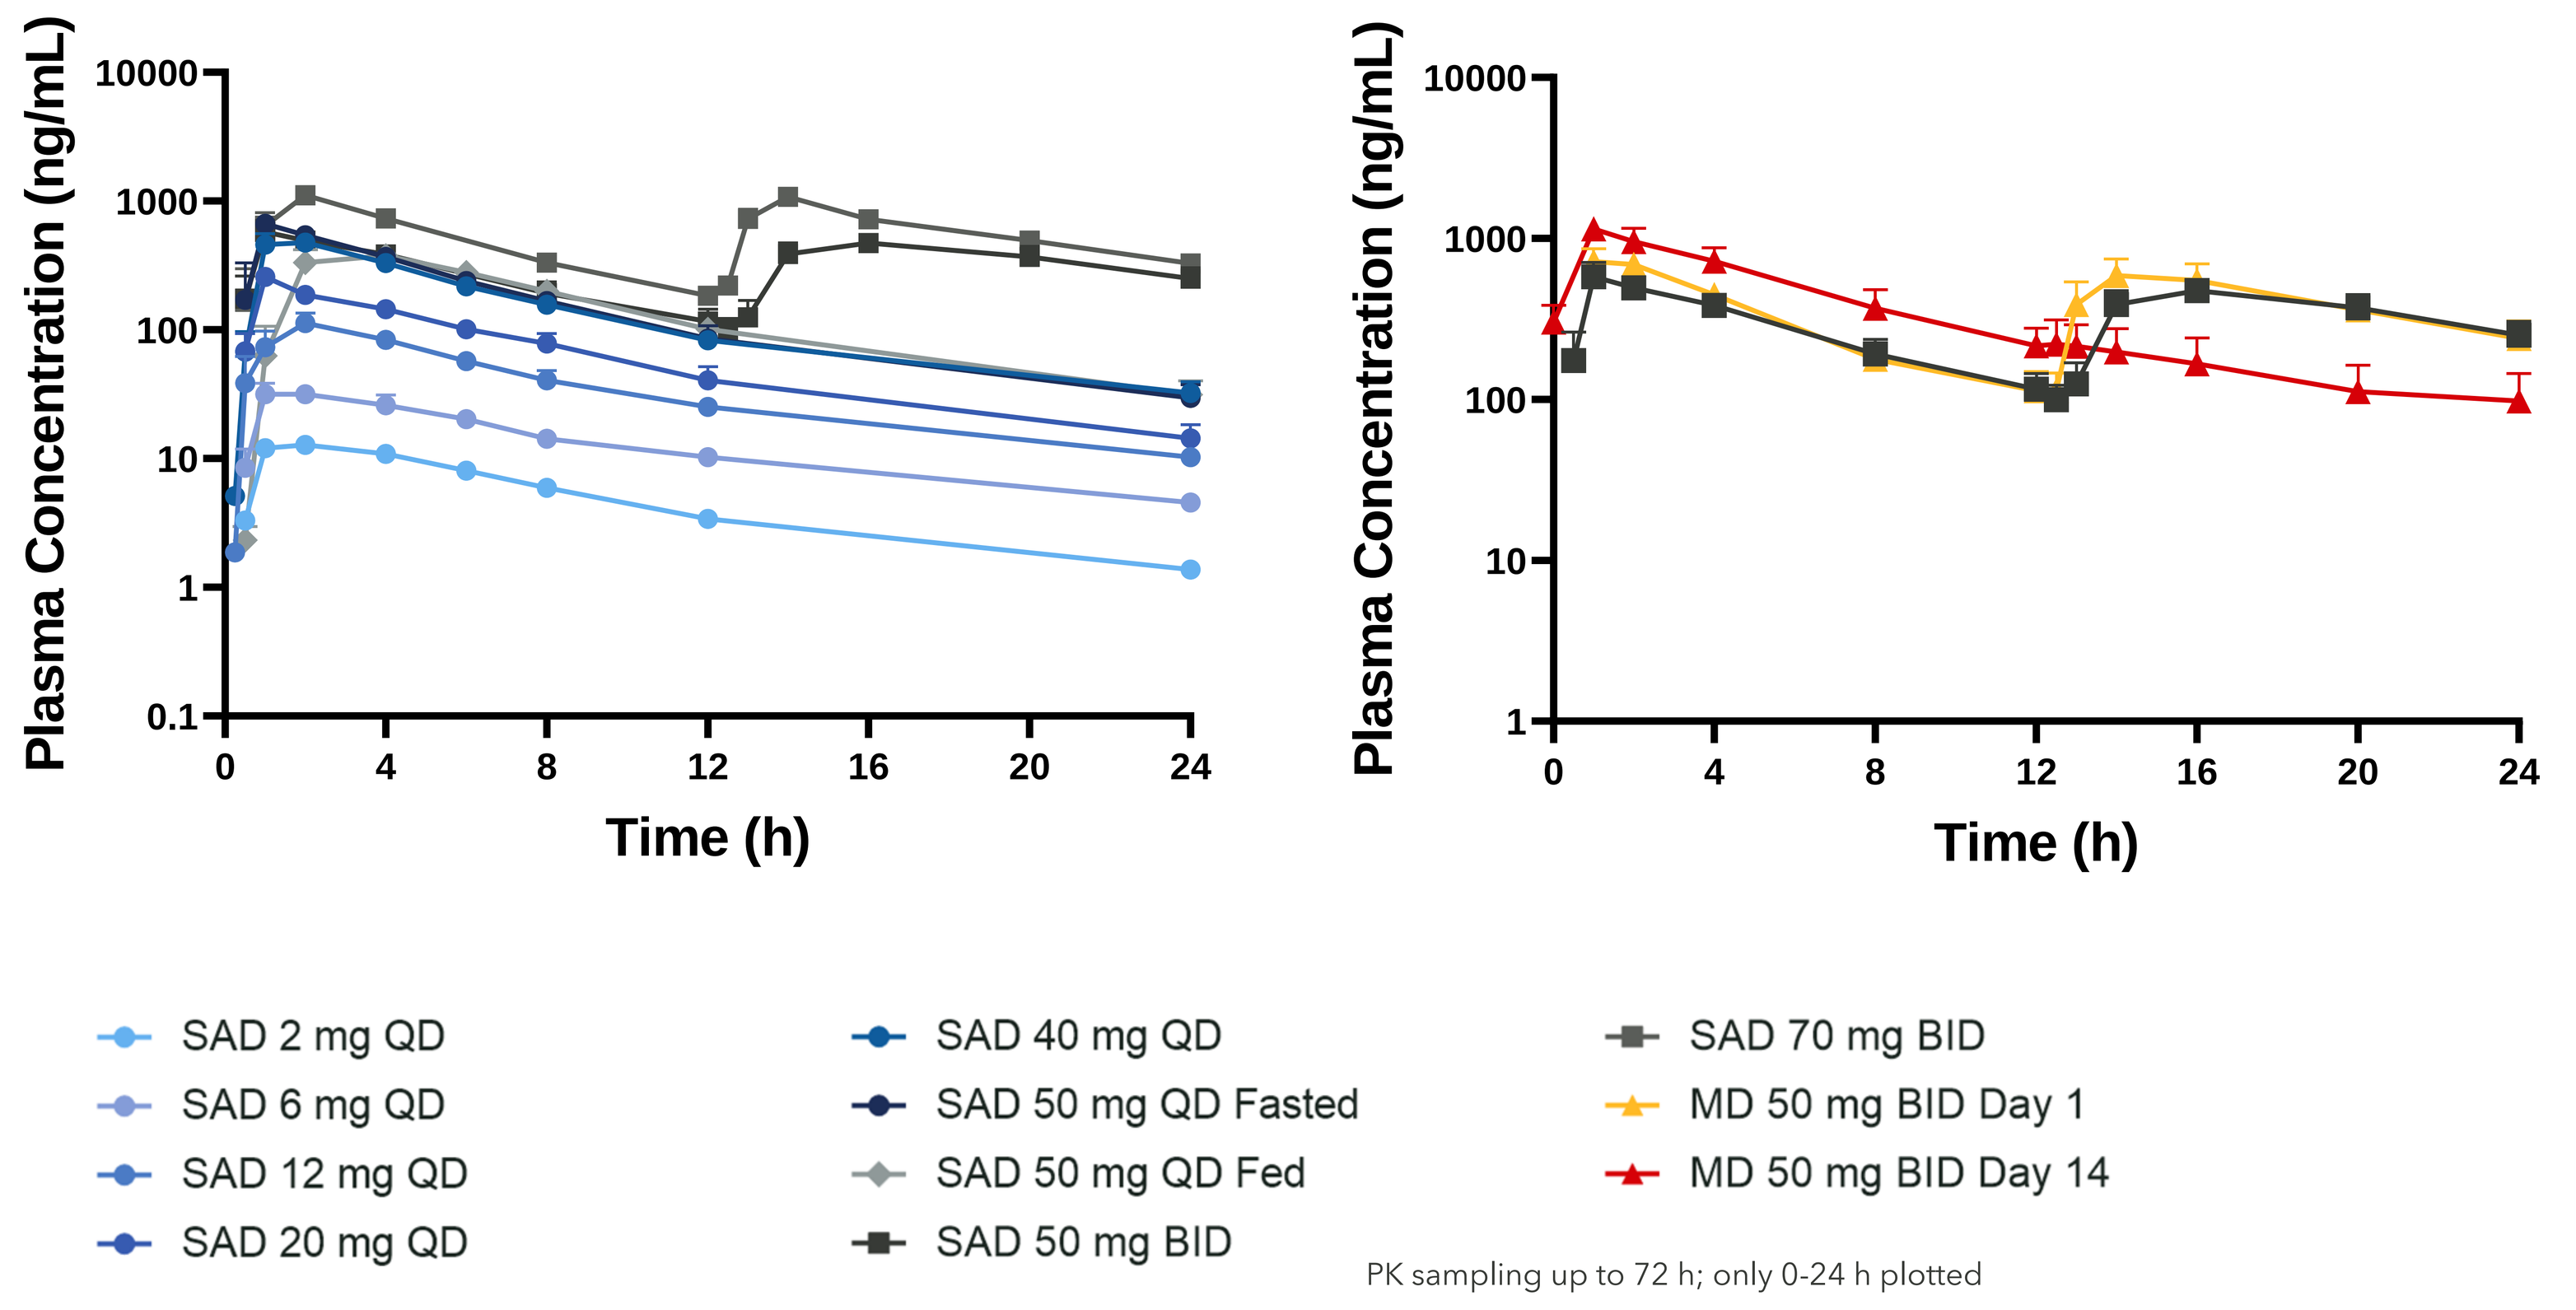
Plasma Concentration (ng/mL)
Time (h)
SAD 2 mg QD
SAD 6 mg QD
SAD 12 mg QD
SAD 20 mg QD
SAD 40 mg QD
SAD 50 mg QD Fasted
SAD 50 mg QD Fed
SAD 50 mg BID
SAD 70 mg BID
MD 50 mg BID Day 1
MD 50 mg BID Day 14

PK sampling up to 72 h; only 0-24 h plotted

Summary

- RXC007 is a highly potent, selective and orally-active ROCK2 inhibitor
- ROCK2 is a validated, compelling target at a key junction in cell signalling pathways central to fibrosis
- Robust preclinical efficacy data across disease models supports clinical development plan in lung fibrosis – IPF and CF-ILD
- Phase 1 data in SAD and multi dose cohorts confirms drug like profile for safety and PK
 - No significant adverse events observed in healthy volunteers and predictable PK profile
- Phase 2a in IPF expected to commence H2 2022, is a dose-ranging study to inform Phase 2b dose
 - Initial 12-week Phase 2a study for early efficacy readouts, safety and tolerability in IPF patients +/- SoC, in addition to target and disease biomarker engagement
- Phase 2b planned for RXC007 plus SoC over 12 months with lung function (FVC) as primary endpoint

References

1. **Zhou et al**; *J Clin Invest. 2013 Mar;123(3):1096-108.*
2. **Shimizu et al**; Int J Immunopathol Pharmacol. 2014 Jan-Mar;27(1):37-44.
3. **Knipe et al**; *Am J Respir Cell Mol Biol. 2018 Apr; 58(4): 471-48.*

Redx Pharma, Block 33S, Mereside, Alderley Park, Cheshire, SK10 4TG, UK e: n.guisot@redxpharma.com, www.redxpharma.com

ICLAF 2022