REDX12271 is a novel, selective DDR1 inhibitor with the potential to treat multiple chronic kidney diseases

Kay Eckersley[1], Inder Bhamra[1], Ana Varela[1], James Ryan[1] , Michal Andrä [1] , Shân Preece[1] , Daniel Wilcock[1] , Peter Bunyard[1] , Nicolas E.S. Guisot[1] , Cliff Jones[1] , Richard Armer[1] , [1]Redx Pharma plc

INTRODUCTION

DDR1 is an Attractive Fibrosis Target

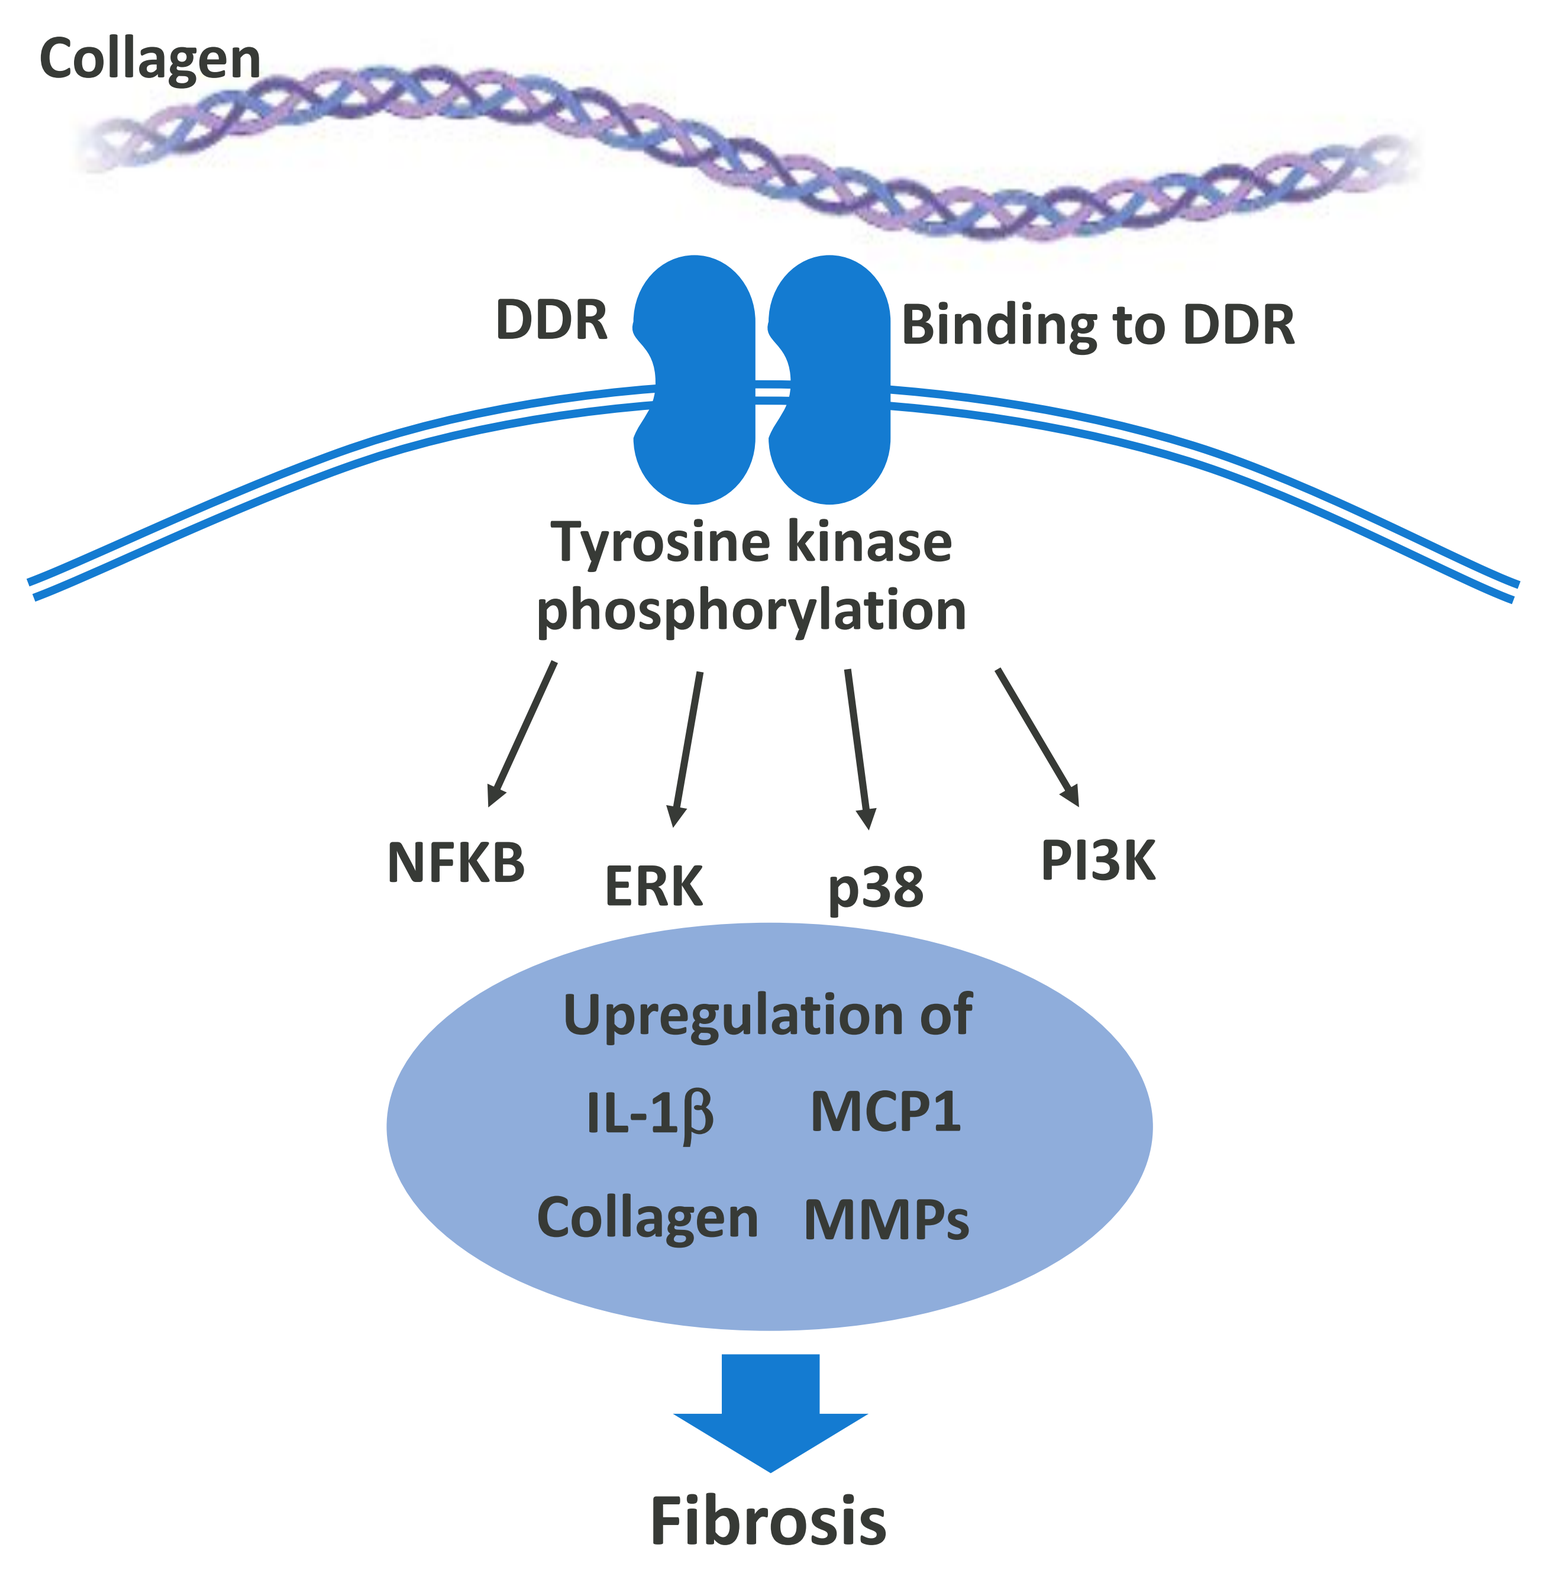
Collagen
DDR
Binding to DDR
Tyrosine kinase phosphorylation
NFKB
ERK
p38
PI3K
Upregulation of
IL-1β MCP1
Collagen MMPs
Fibrosis

- Discoidin domain receptors (DDRs) are collagen-activated receptor tyrosine kinases which have been shown to be highly expressed in many fibrotic diseases.
- DDRs are an emerging new target with the potential to treat multiple fibrotic conditions.
- REDX12271 is a novel, selective and potent orally available DDR1 inhibitor
- Selective inhibition of DDR1 with REDX12271 reduces inflammation and fibrosis in a mouse unilateral ureteral obstruction (UUO) model in both prophylactic and therapeutic intervention.
- REDX12271 lead indication: Chronic kidney disease (CKD) - a range of conditions with high unmet need
- Potential for disease modifying efficacy across multiple fibrotic conditions

Rationale for Targeting DDR1 in CKD

Increased expression of DDR1 in injured and diseased kidney

DDR1 expression is upregulated in acute kidney injury (AKI)

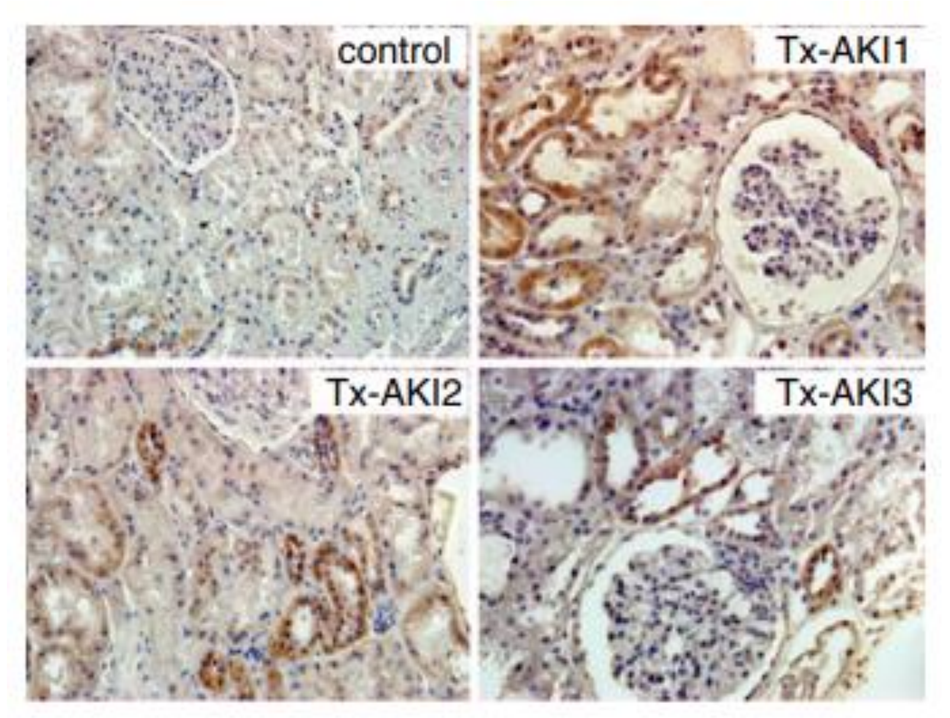
control
Tx-AKI1
Tx-AKI2
Tx-AKI3

Adapted from Chiusa, 2019

- Paraffin kidney sections from control and biopsy specimens from three different patients with transplant AKI
- Upregulated DDR1 expression is evident in the tubules of injured kidneys

DDR1 immunostaining in human kidney biopsies

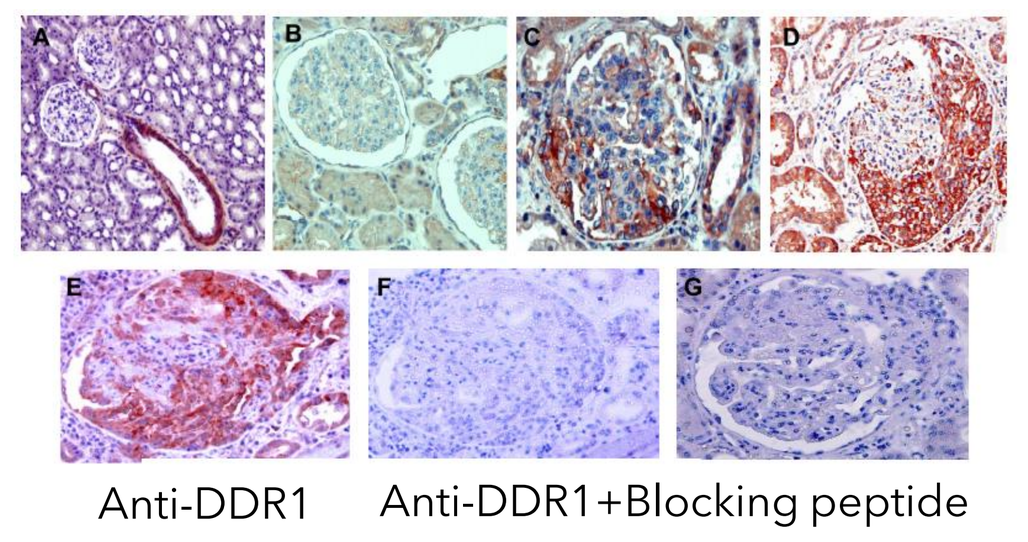
Anti-DDR1
Anti-DDR1+Blocking peptide

Adapted from Keroch, 2012

- In control kidneys (A), DDR1 was predominantly expressed in vessels
- In glomerular disease (B-D), DDR1 was strongly expressed in podocytes and cellular crescents

DDR1 expression is upregulated in human CKD patients

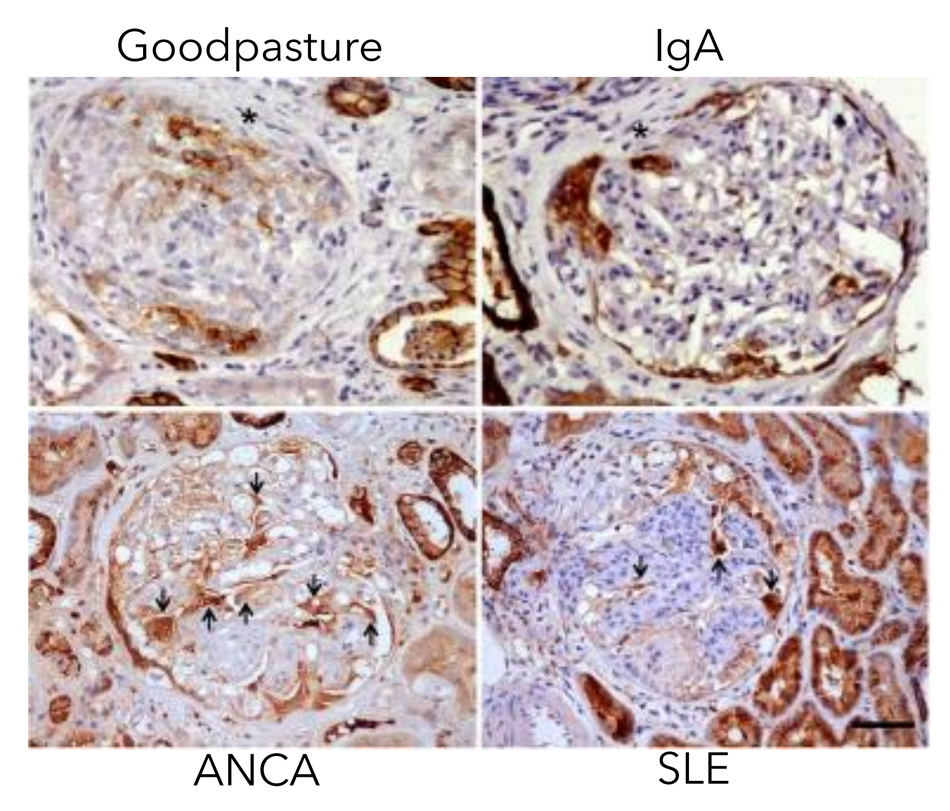
Goodpasture
IgA
ANCA
SLE

Adapted from Moll, 2018

- DDR1 expression is increased in 4 different types of human crescentic glomerulonephritis
- Injured podocytes (arrows) strongly express DDR1

DDR1 knockout mice are protected from inflammation and fibrosis

Loss of DDR1 reduces renal inflammation and fibrosis

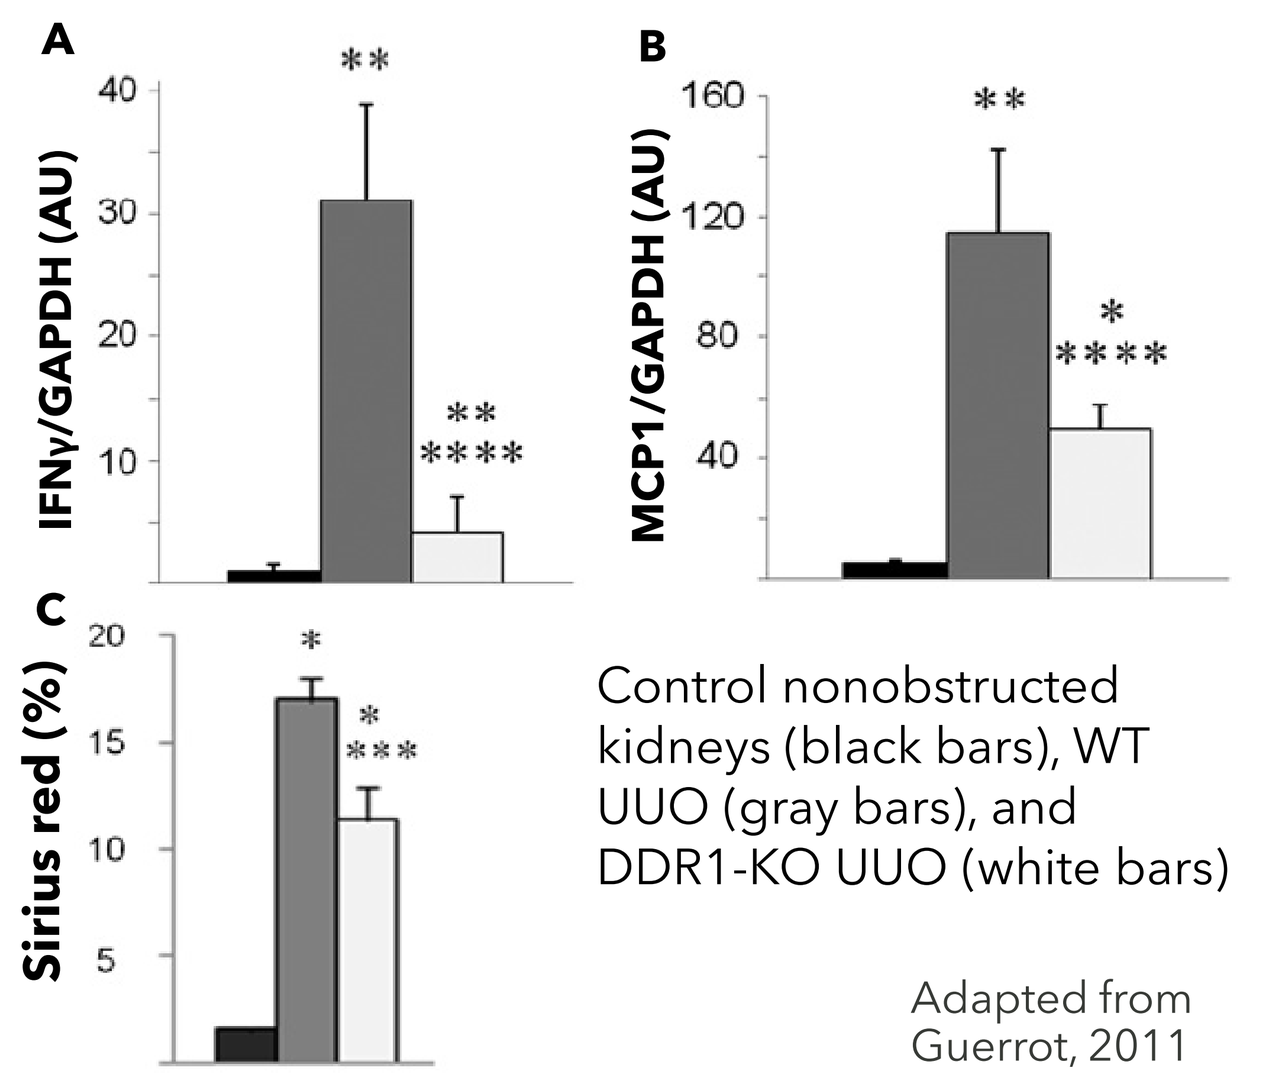
A
IFNγ/GAPDH (AU)
B
MCP1/GAPDH (AU)
C
Sirius red (%)

Control nonobstructed kidneys (black bars), WT UUO (gray bars), and DDR1-KO UUO (white bars)

Adapted from Guerrot, 2011

- Reduction in Sirius red staining (A) and expression of proinflammatory cytokines IFNγ (B) and MCP1/CCL2 (C)

Loss of DDR1 reduces functional severity of crescentic glomerulonephritis

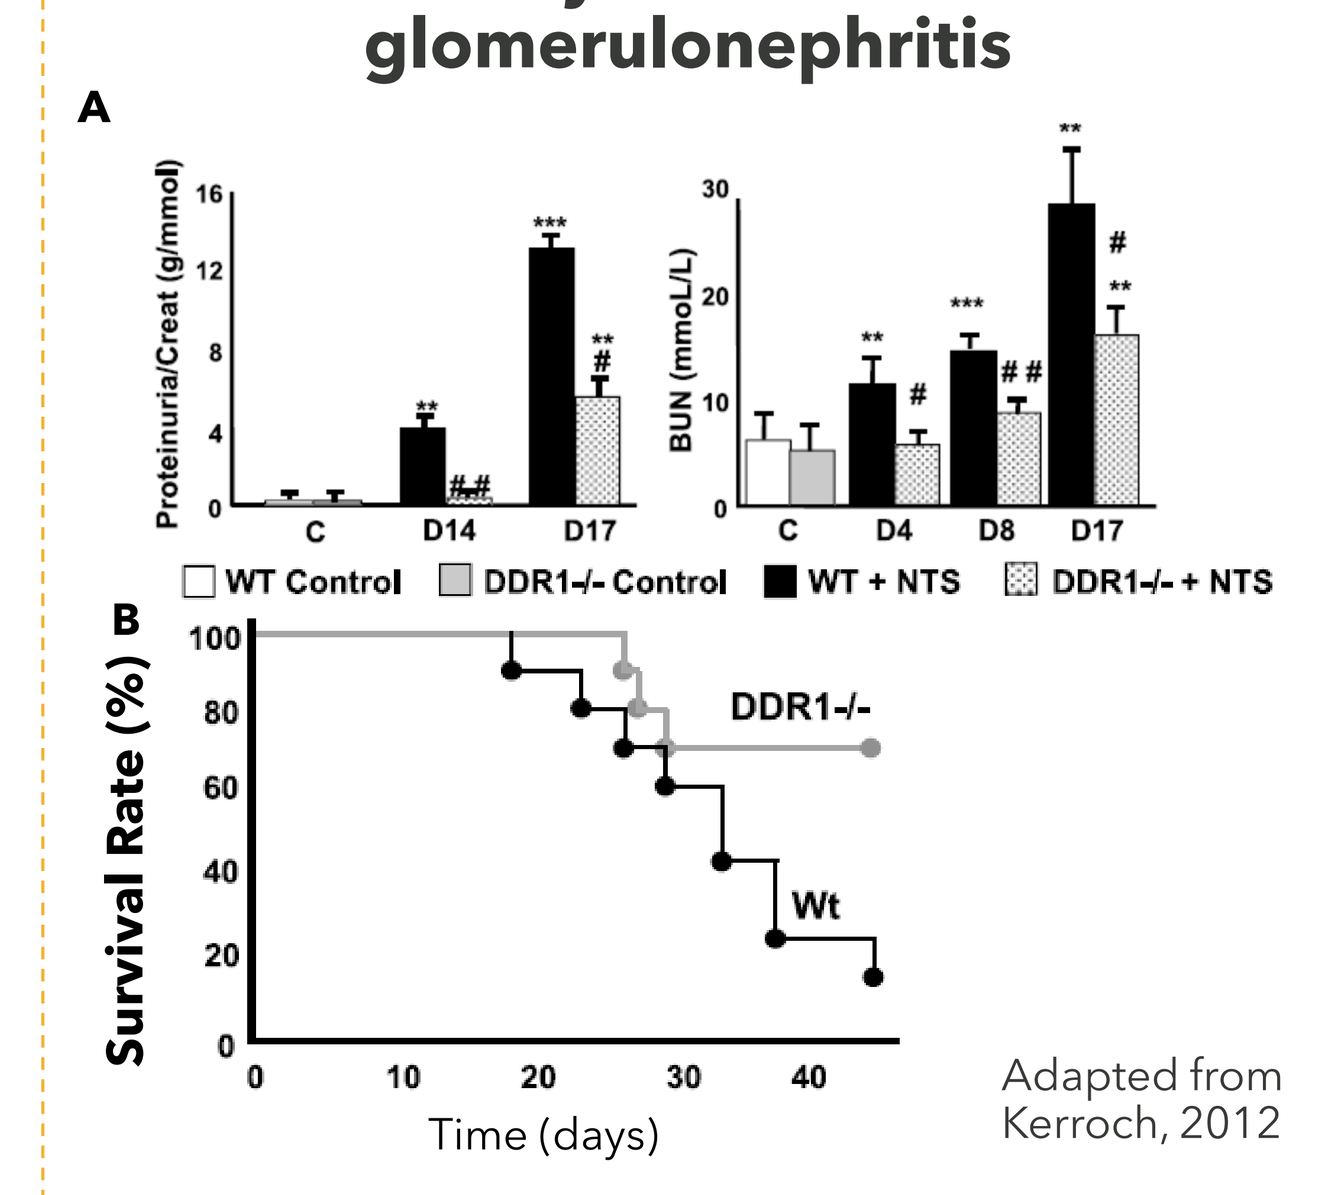
A
Proteinuria/Creat (g/mmol)
BUN (mmol/L)
WT Control
DDR1-/- Control
WT + NTS
DDR1-/- + NTS
B
Survival Rate (%)
DDR1-/-
Wt
Time (days)

Adapted from Kerroch, 2012

- DDR1-/- mice present improved renal function (A) and survival (B)

Loss of DDR1 delays renal fibrosis in hereditary type IV collagen disease

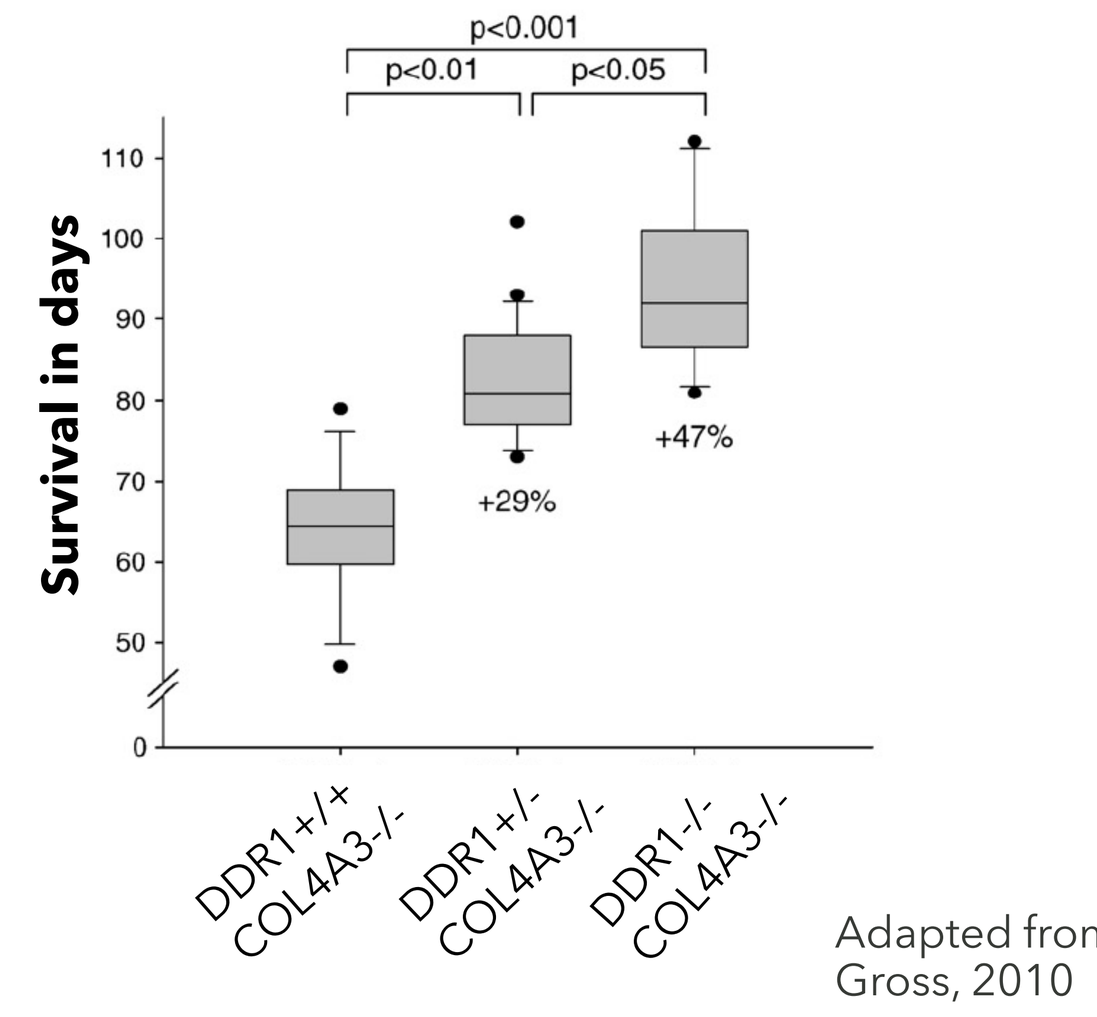
p<0.001
p<0.01
p<0.05
Survival in days
+29%
+47%
DDR1+/+ COL4A3-/-
DDR1+/- COL4A3-/-
DDR1-/- COL4A3-/-

Adapted from Gross, 2010

- Nephoprotective effect of loss DDR1 on renal failure and lifespan

RESULTS

REDX12271 is a Potent and Selective DDR1 Inhibitor

REDX12271 prevents *in vitro* activation of DDR1

Biochemical assay

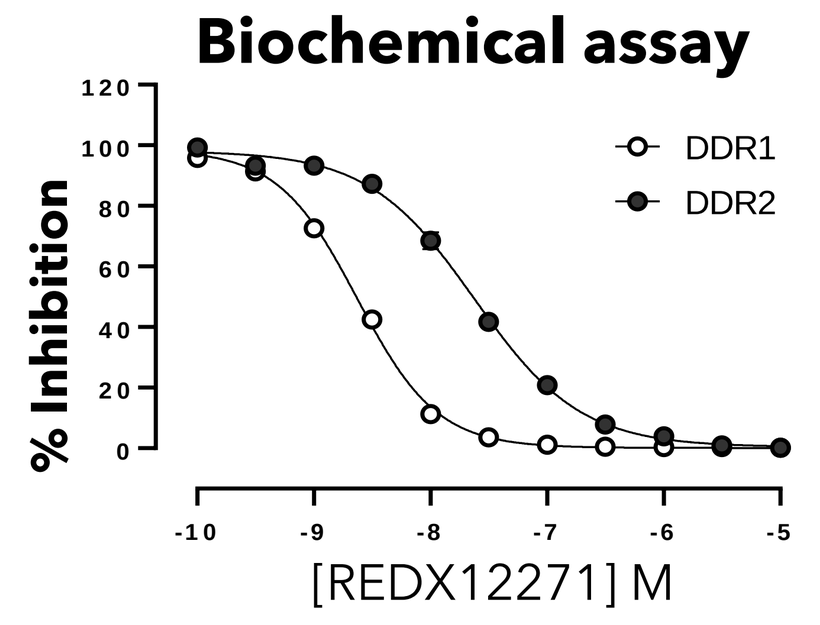
% Inhibition
DDR1
DDR2
[REDX12271] M

REDX12271 potently inhibits DDR1 in a binding assay with pIC_{50} 8.7 +/- 0.01

Cellular target engagement

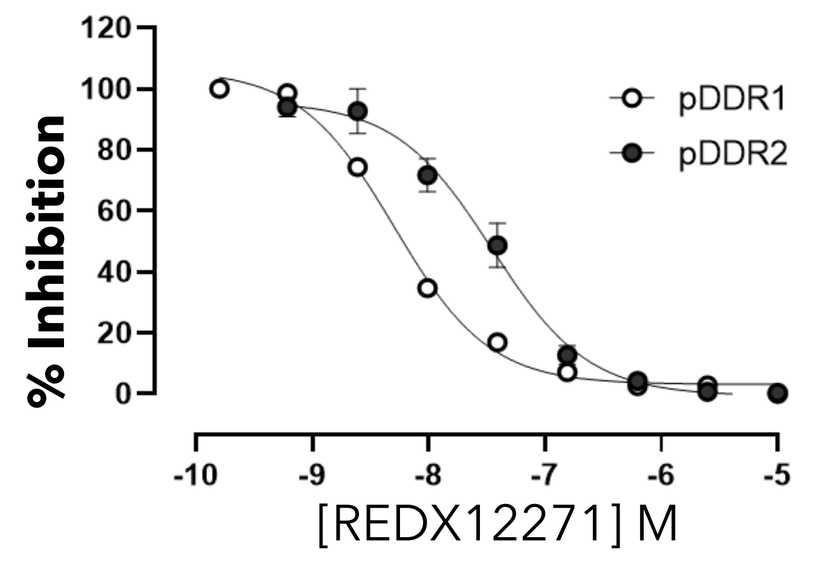
% Inhibition
pDDR1
pDDR2
[REDX12271] M

REDX12271 potently inhibits p-DDR1 in HEK-DDR1 and HEK-DDR2 over-expressing cells with pIC_{50} 8.2 +/- 0.1

- REDX12271 is a potent inhibitor of DDR1 with a ten-fold selectivity over DDR2

REDX12271 has a favourable PK and selectivity profile

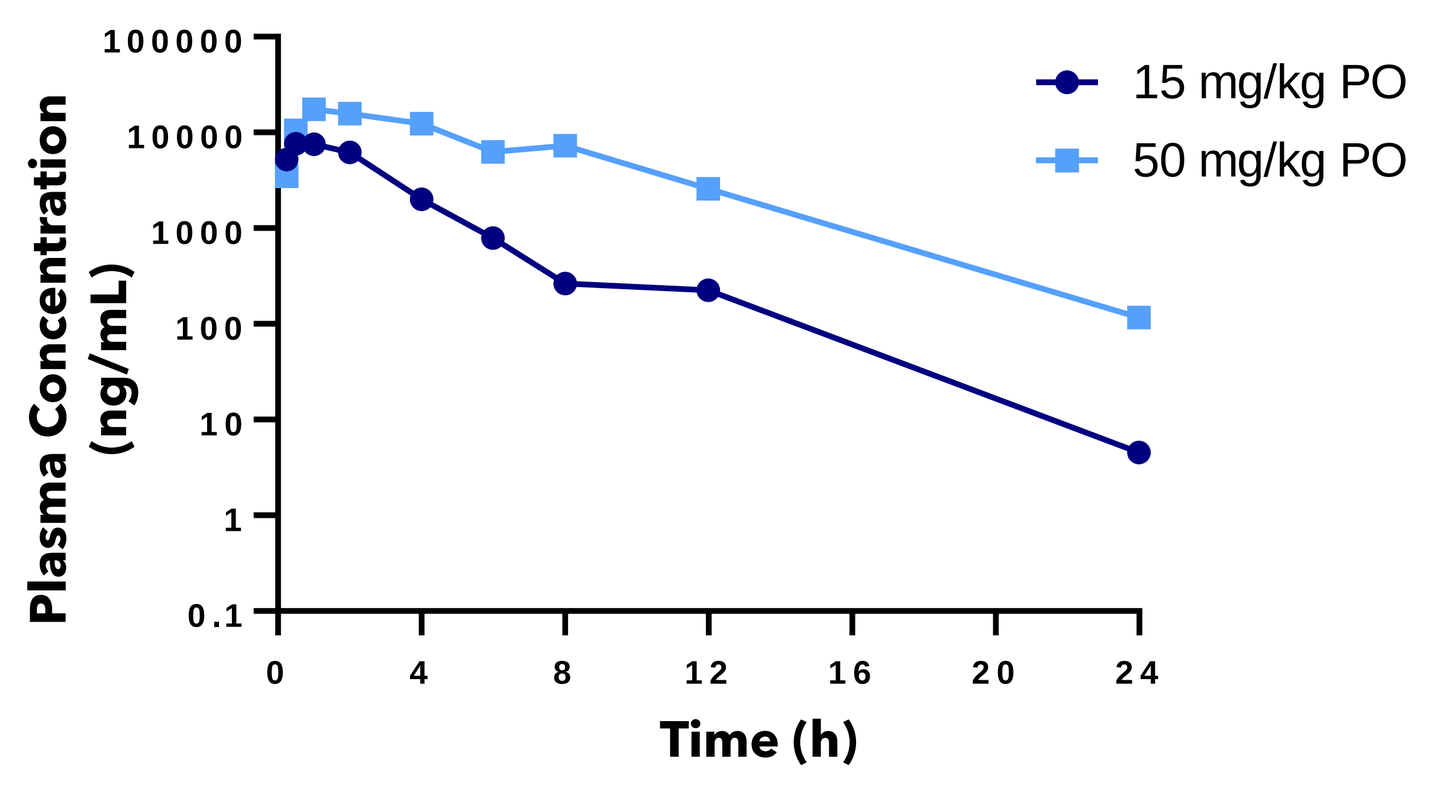
Plasma Concentration (ng/mL)
Time (h)
15 mg/kg PO
50 mg/kg PO

- Mean plasma concentration vs time profile after 15 mg/kg and 50 mg/kg PO in C57BL mouse
- Kinase selectivity profile:
 10 kinases >90% Inhibition at 1000 nM
- Follow-up IC50s reveal 90 fold selectivity against off-target kinases

DDR1 Inhibitors Modulate Inflammatory and Fibrotic Pathways

DDR1 regulates proinflammatory MCP-1 via phosphorylation of BCR

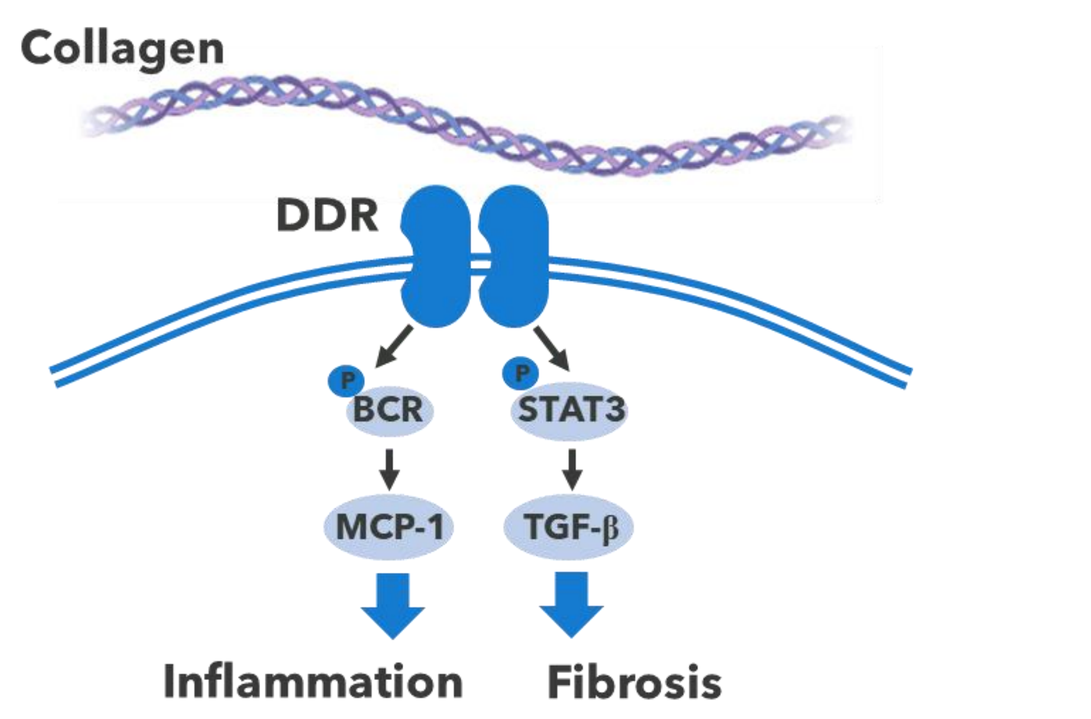
Collagen
DDR
BCR
STAT3
MCP-1
TGF-β
Inflammation
Fibrosis

DDR activation induces p-BCR

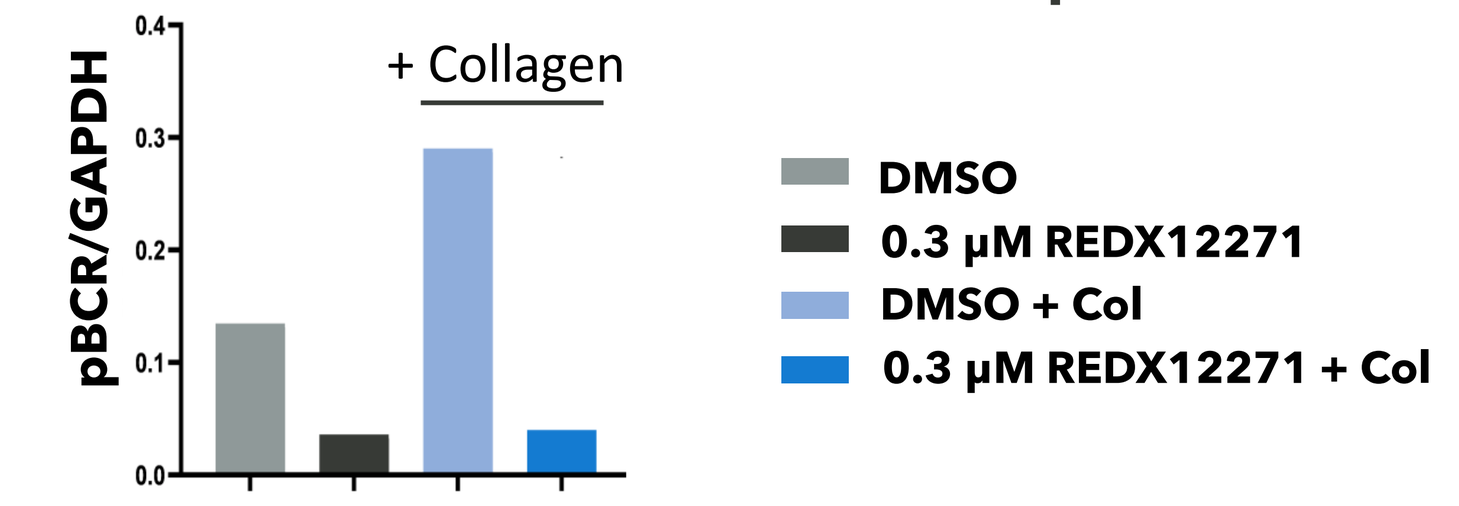
pBCR/GAPDH
+ Collagen
DMSO
0.3 μM REDX12271
DMSO + Col
0.3 μM REDX12271 + Col

- Inhibition of DDR1 in HEK-DDR1 cells results in decreased phosphorylation of native BCR as determined by immunoblotting

Pathway analysis reveals role of DDR1 in tissue remodelling

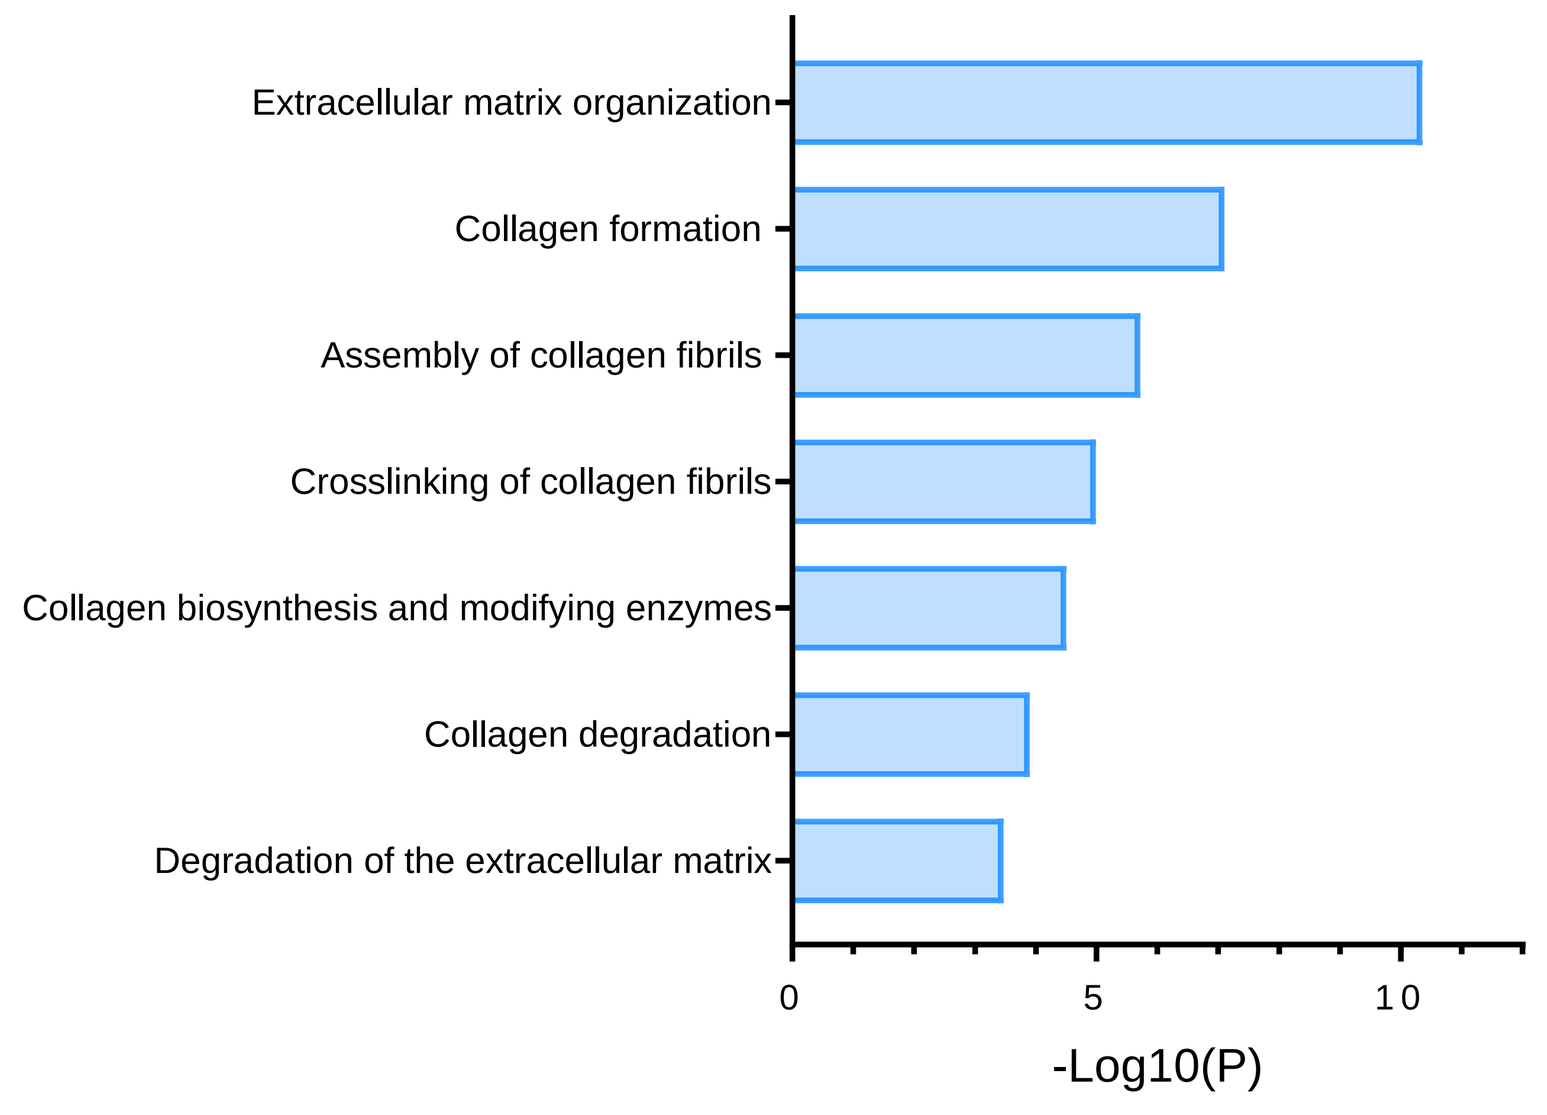
Extracellular matrix organization
Collagen formation
Assembly of collagen fibrils
Crosslinking of collagen fibrils
Collagen biosynthesis and modifying enzymes
Collagen degradation
Degradation of the extracellular matrix
-Log10(P)

- Overview of extracellular matrix (ECM) organization reactome pathways enriched for regulated genes compared to UUO Vehicle at significance level p<0.05
- Significant modulation of pathways associated with tissue remodelling observed with DDR1 inhibition in a mouse model of UUO

REDX12271 Reduces Inflammation and Fibrosis in a Prophylactic Murine Unilateral Ureteral Obstruction Model

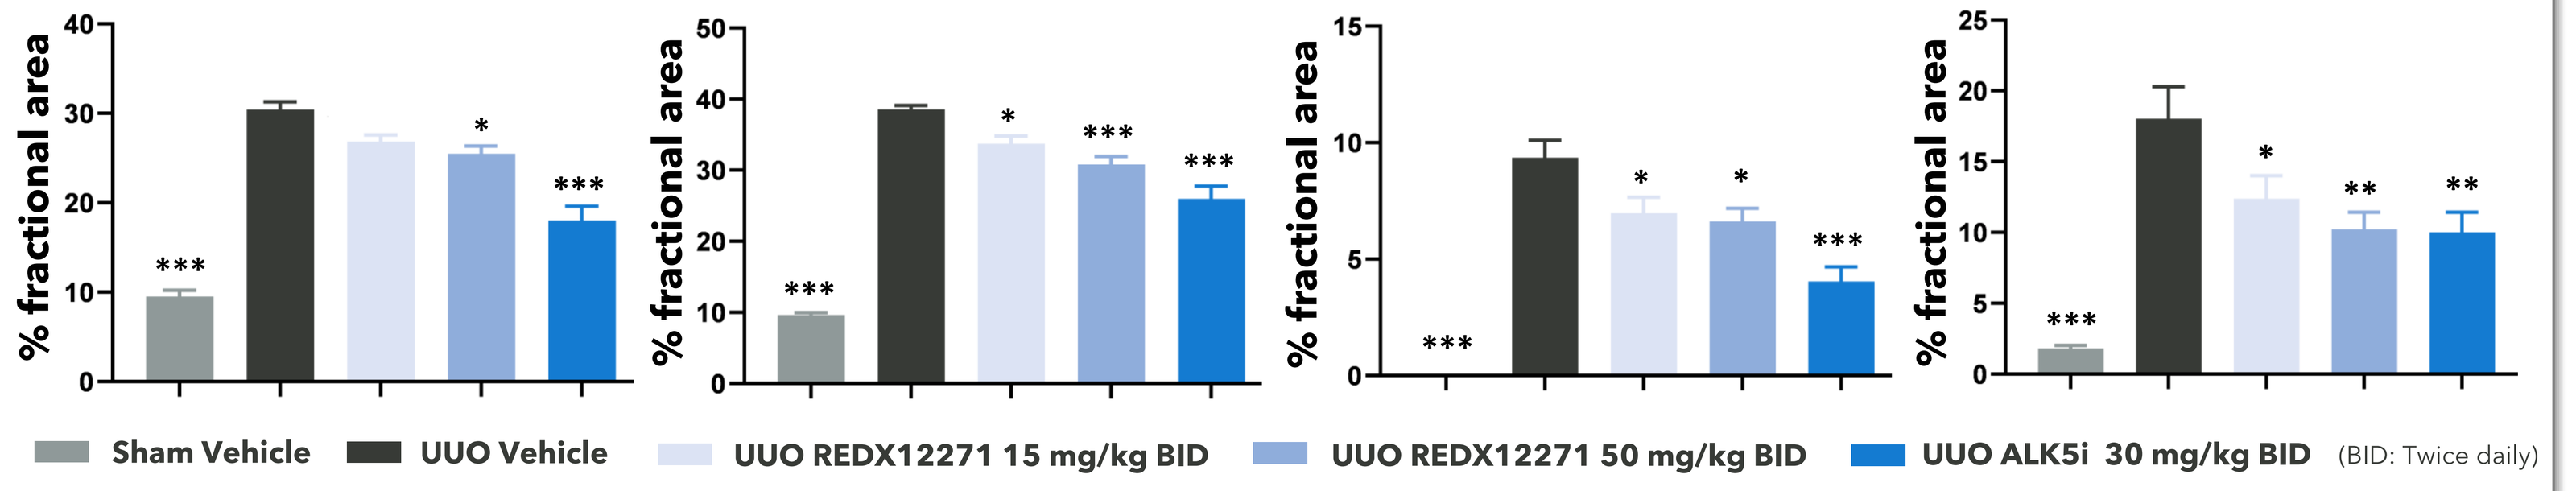
Collagen 1
α-Smooth muscle actin
KIM-1
F4/80
% fractional area
Sham Vehicle
UUO Vehicle
UUO REDX12271 15 mg/kg BID
UUO REDX12271 50 mg/kg BID
UUO ALK5i 30 mg/kg BID
(BID: Twice daily)

Representative images
Kidney histology Col1A1

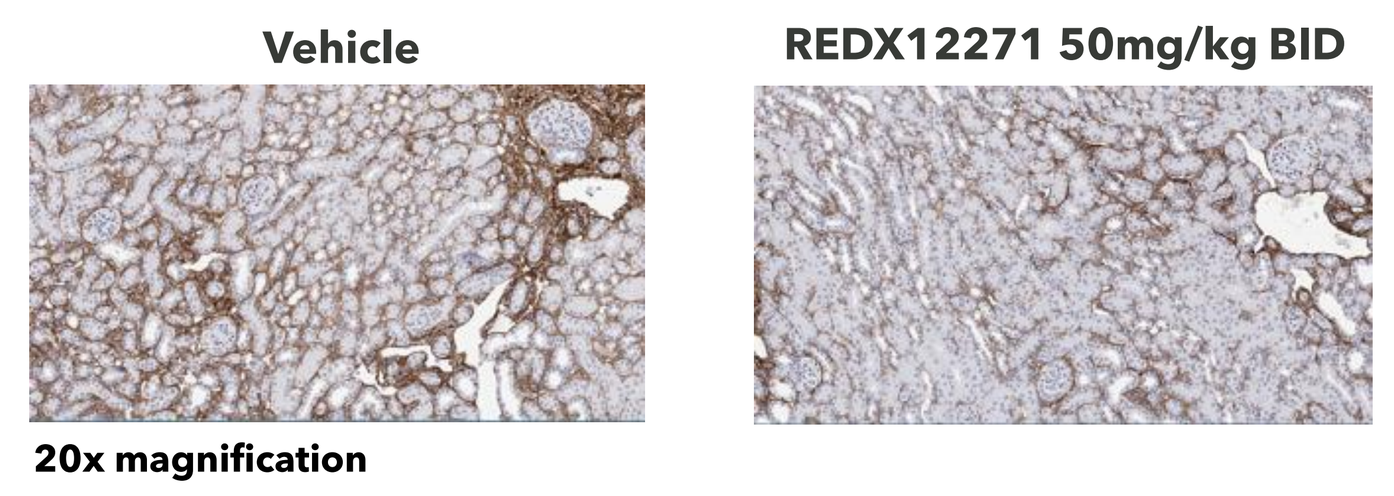
Vehicle
REDX12271 50mg/kg BID

20x magnification

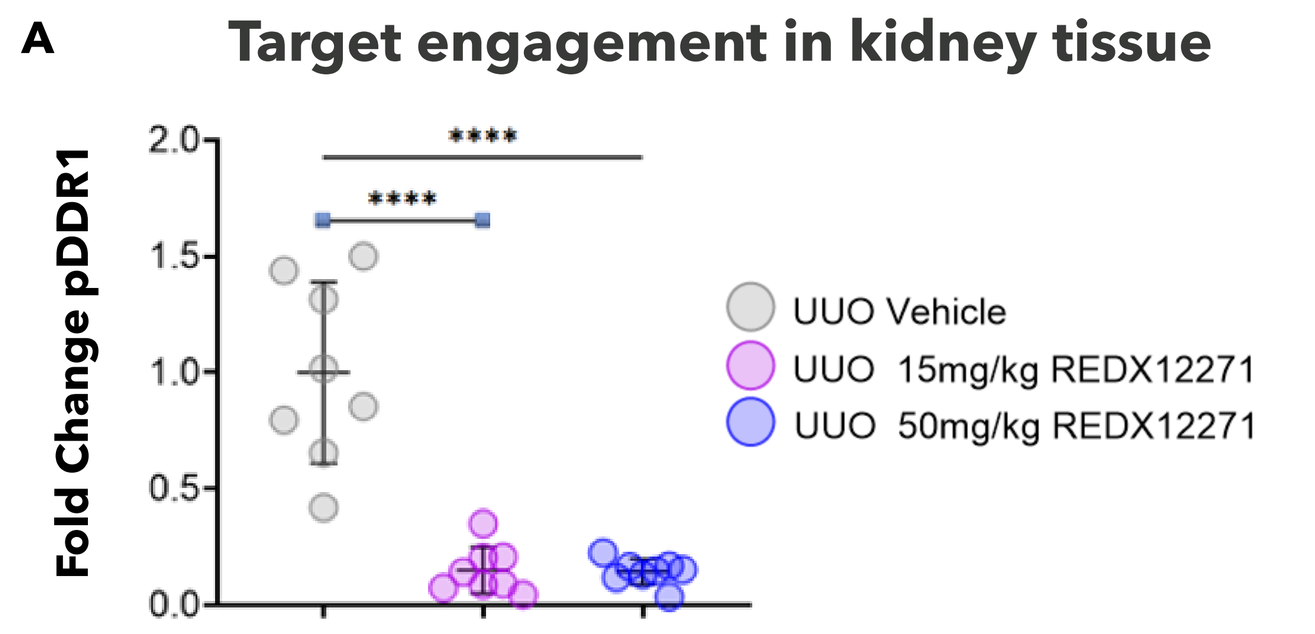
A
Target engagement in kidney tissue
Fold Change pDDR1
UUO Vehicle
UUO 15mg/kg REDX12271
UUO 50mg/kg REDX12271

- 8-Day prophylactic murine UUO kidney fibrosis model (Oral dosing from day -1 with surgery performed on day 1)
- Significantly reduced inflammation (F4/80), kidney injury (KIM-1) and fibrosis (Col1a1/α-SMA) as determined by immunohistochemistry
- Target engagement by REDX12271 was determined by immunoblotting for p-DDR1 (A)

REDX12271 Reduces Inflammation and Fibrosis in a Therapeutic Murine Unilateral Ureteral Obstruction Model

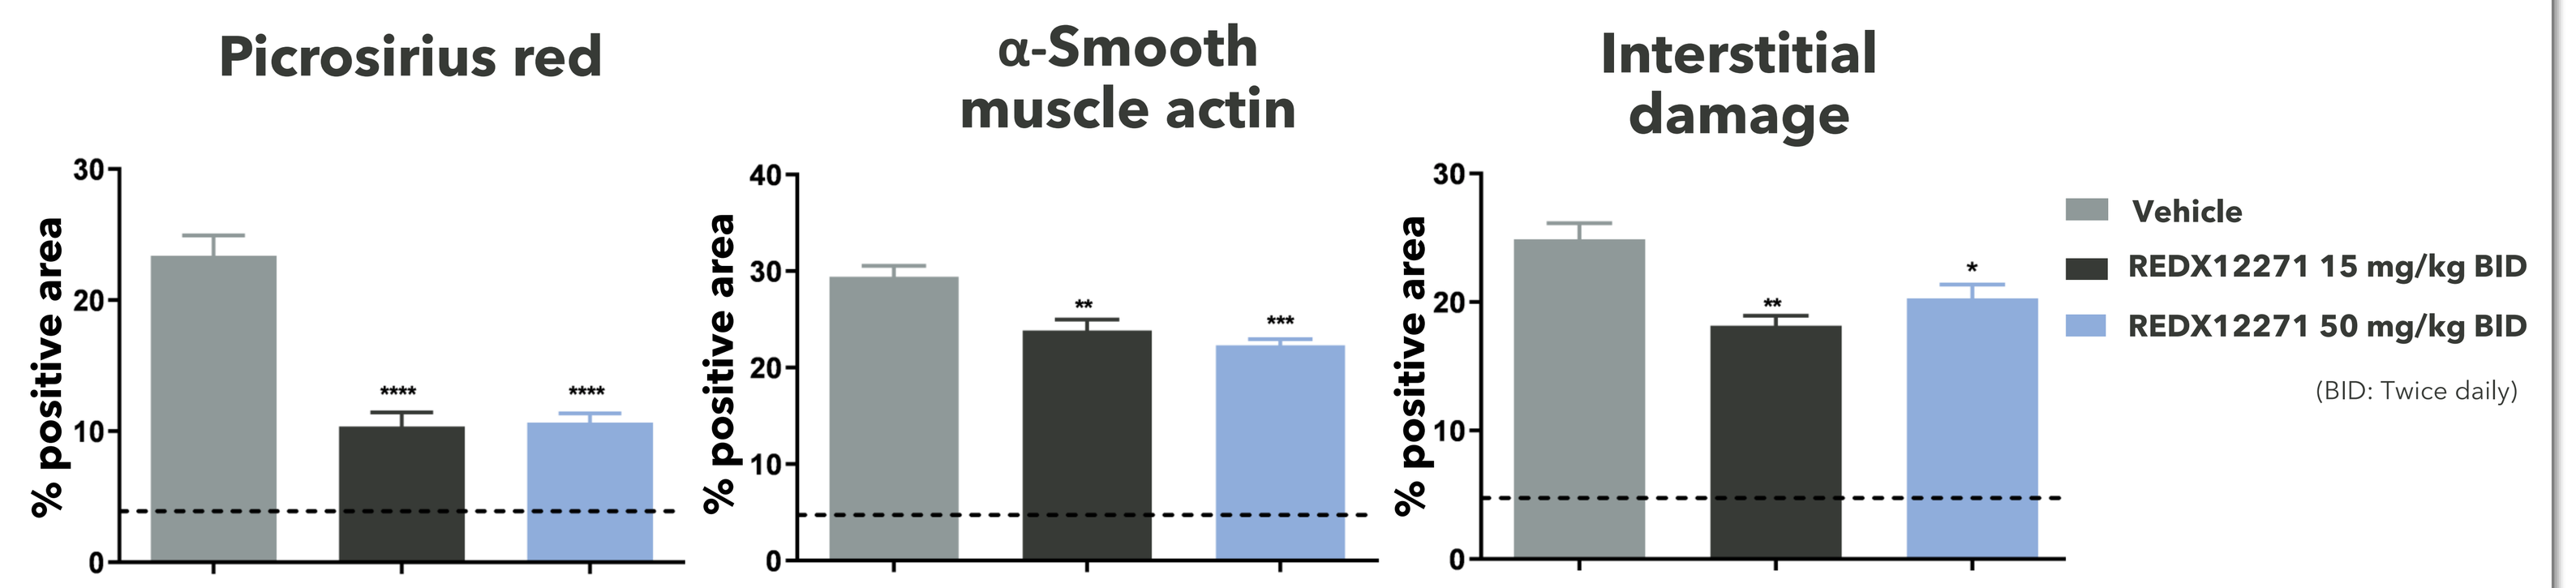
Picrosirius red
α-Smooth muscle actin
Interstitial damage
% positive area
Vehicle
REDX12271 15 mg/kg BID
REDX12271 50 mg/kg BID
(BID: Twice daily)

Representative images
Kidney histology α-SMA

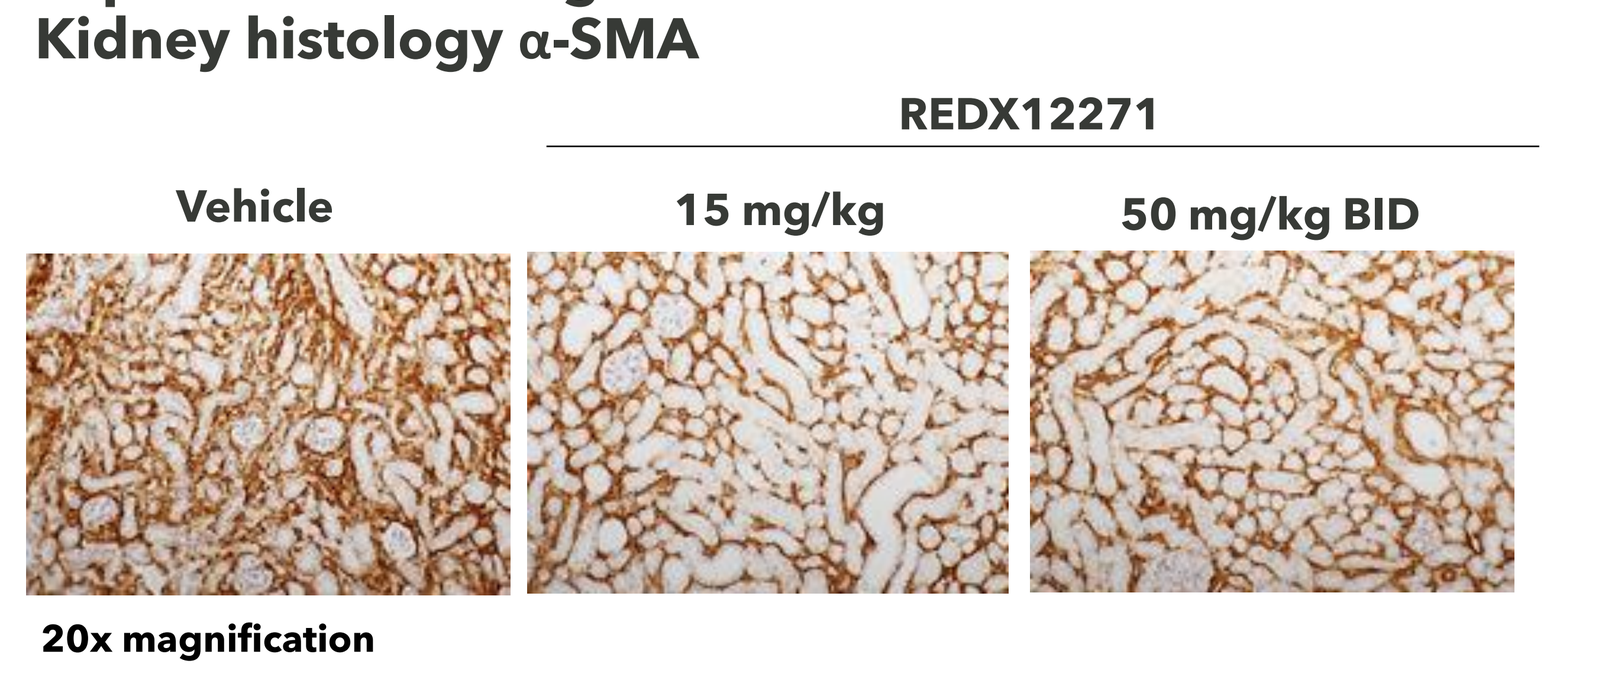
REDX12271
Vehicle
15 mg/kg
50 mg/kg BID

20x magnification

Representative images
Kidney histology Picrosirius red

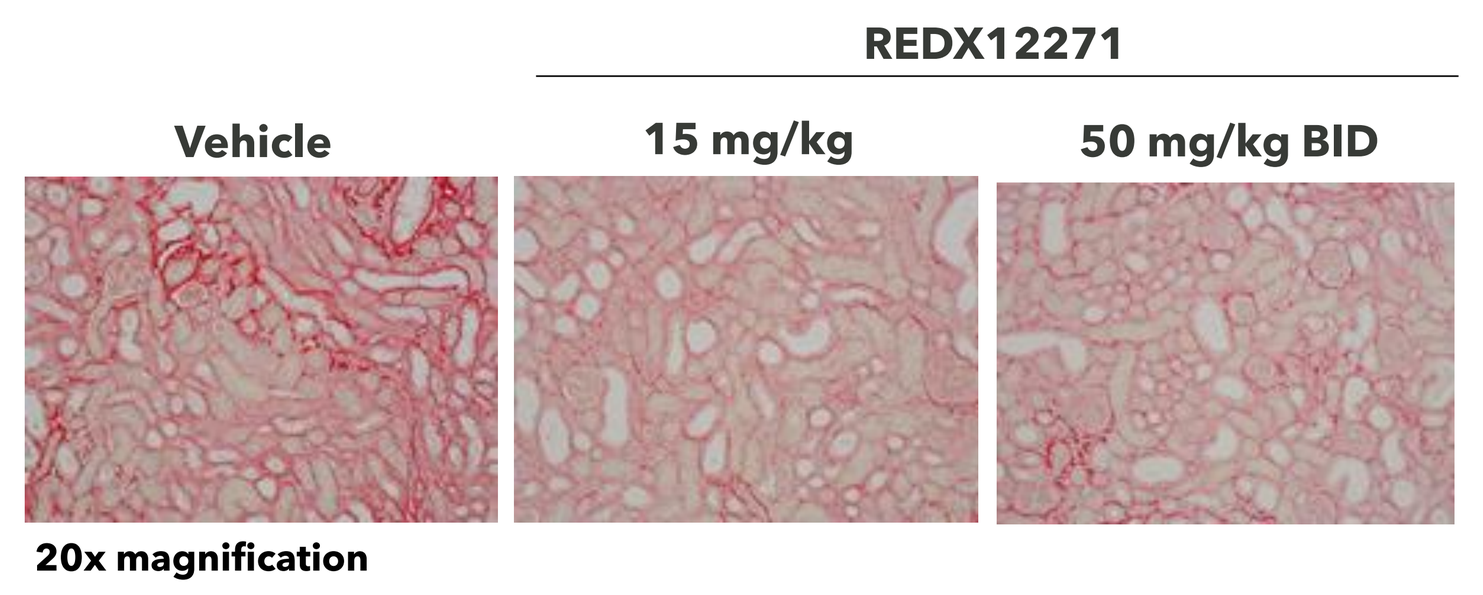
REDX12271
Vehicle
15 mg/kg
50 mg/kg BID

20x magnification

- 10-Day therapeutic murine UUO kidney fibrosis model (Oral dosing from day 5 post surgery)
- Significantly reduced fibrosis and collagen deposition (picrosirius red), myofibroblast transformation (α-SMA), inflammation (F4/80) and tubulointerstitial damage (H&E) as determined by immunohistochemistry
- No effect was observed with the ALK5 inhibitor in this model (data not shown)

SUMMARY

- These data show that selective inhibition of DDR1 with REDX12271 reduces inflammation and fibrosis in UUO models.
- To our knowledge this is the first example of selective inhibition of DDR1 with a small molecule giving rise to efficacy in mouse UUO models.
- Selective inhibition of DDR1 represents an attractive approach for further investigation towards the development of new treatments for CKD.

References: *Gross O et al. Loss of collagen-receptor DDR1 delays renal fibrosis in hereditary type IV collagen disease. Matrix Biology 2010; 29:346-356. doi:10.1016/j.matbio.2010.03.002*
Guerrot D et al. Discodin Domain Receptor 1 is a major mediator of inflammation and fibrosis in obstructive nephropathy. Am J Pathol. 2011;179:83-91. doi:10.1016/j.ajpath.2011.03.023
Kerroch M et al. Genetic inhibition of discoidin domain receptor 1 protects mice against crescentic glomerulonephritis. The FASEB Journal 2012; 26:4079-4091. doi:10.1096/fj.11-194902
Moll S et al. DDR1 role in fibrosis and its pharmacological targeting. BBA – Mol. Cell Res. 2019; 118474. doi:10.1016/j.bbamcr.2019.04.004
Chiusa et al. The extracellular matrix receptor Discoidin Domain Receptor 1 regulates collagen transcription by translocating to the nucleus. JASN 2019; 30:1605-1624. doi:10.1681/ASN.2018111160

ASN Kidney Week 2022
3-6 November | Orlando

k.eckersley@redxpharma.com
https://www.redxpharma.com/

QR Code placeholder