Poster No. P6

RXC008, a GI-targeted ROCK Inhibitor, Suppresses Fibrosis and Tissue Injury in Models of Crohn's Disease

Kirsty Houslay[1], Peter Bunyard[1], Kay Eckersley[1], Adriana Gambardella[1], Amy Marshall[1], Chris Piekarski[1], Clifford Jones[1], Andrew Belfield[1], Diana Bishop[1], Helen McKeever[1], Helen Timmis[1], Jane Robertson[1], Richard Armer[1], Simon Bos[2], Debby Laukens[2]

[1]Redx Pharma PLC, UK, [2]University of Ghent, Belgium

INTRODUCTION

Fibrostenosis is an Unmet Clinical Need in Crohn's Disease

- Crohn's disease affects 1.7m people globally, of which 50% will develop strictures or complications leading to fibrostenosis.
- Caused by excessive build-up of collagen and extra-cellular matrix leading to narrowing of the GI tract and obstruction.
- There is no current pharmaceutical treatment.
- Treatment requires invasive surgery such as balloon dilation, stricture-plasty and resection.

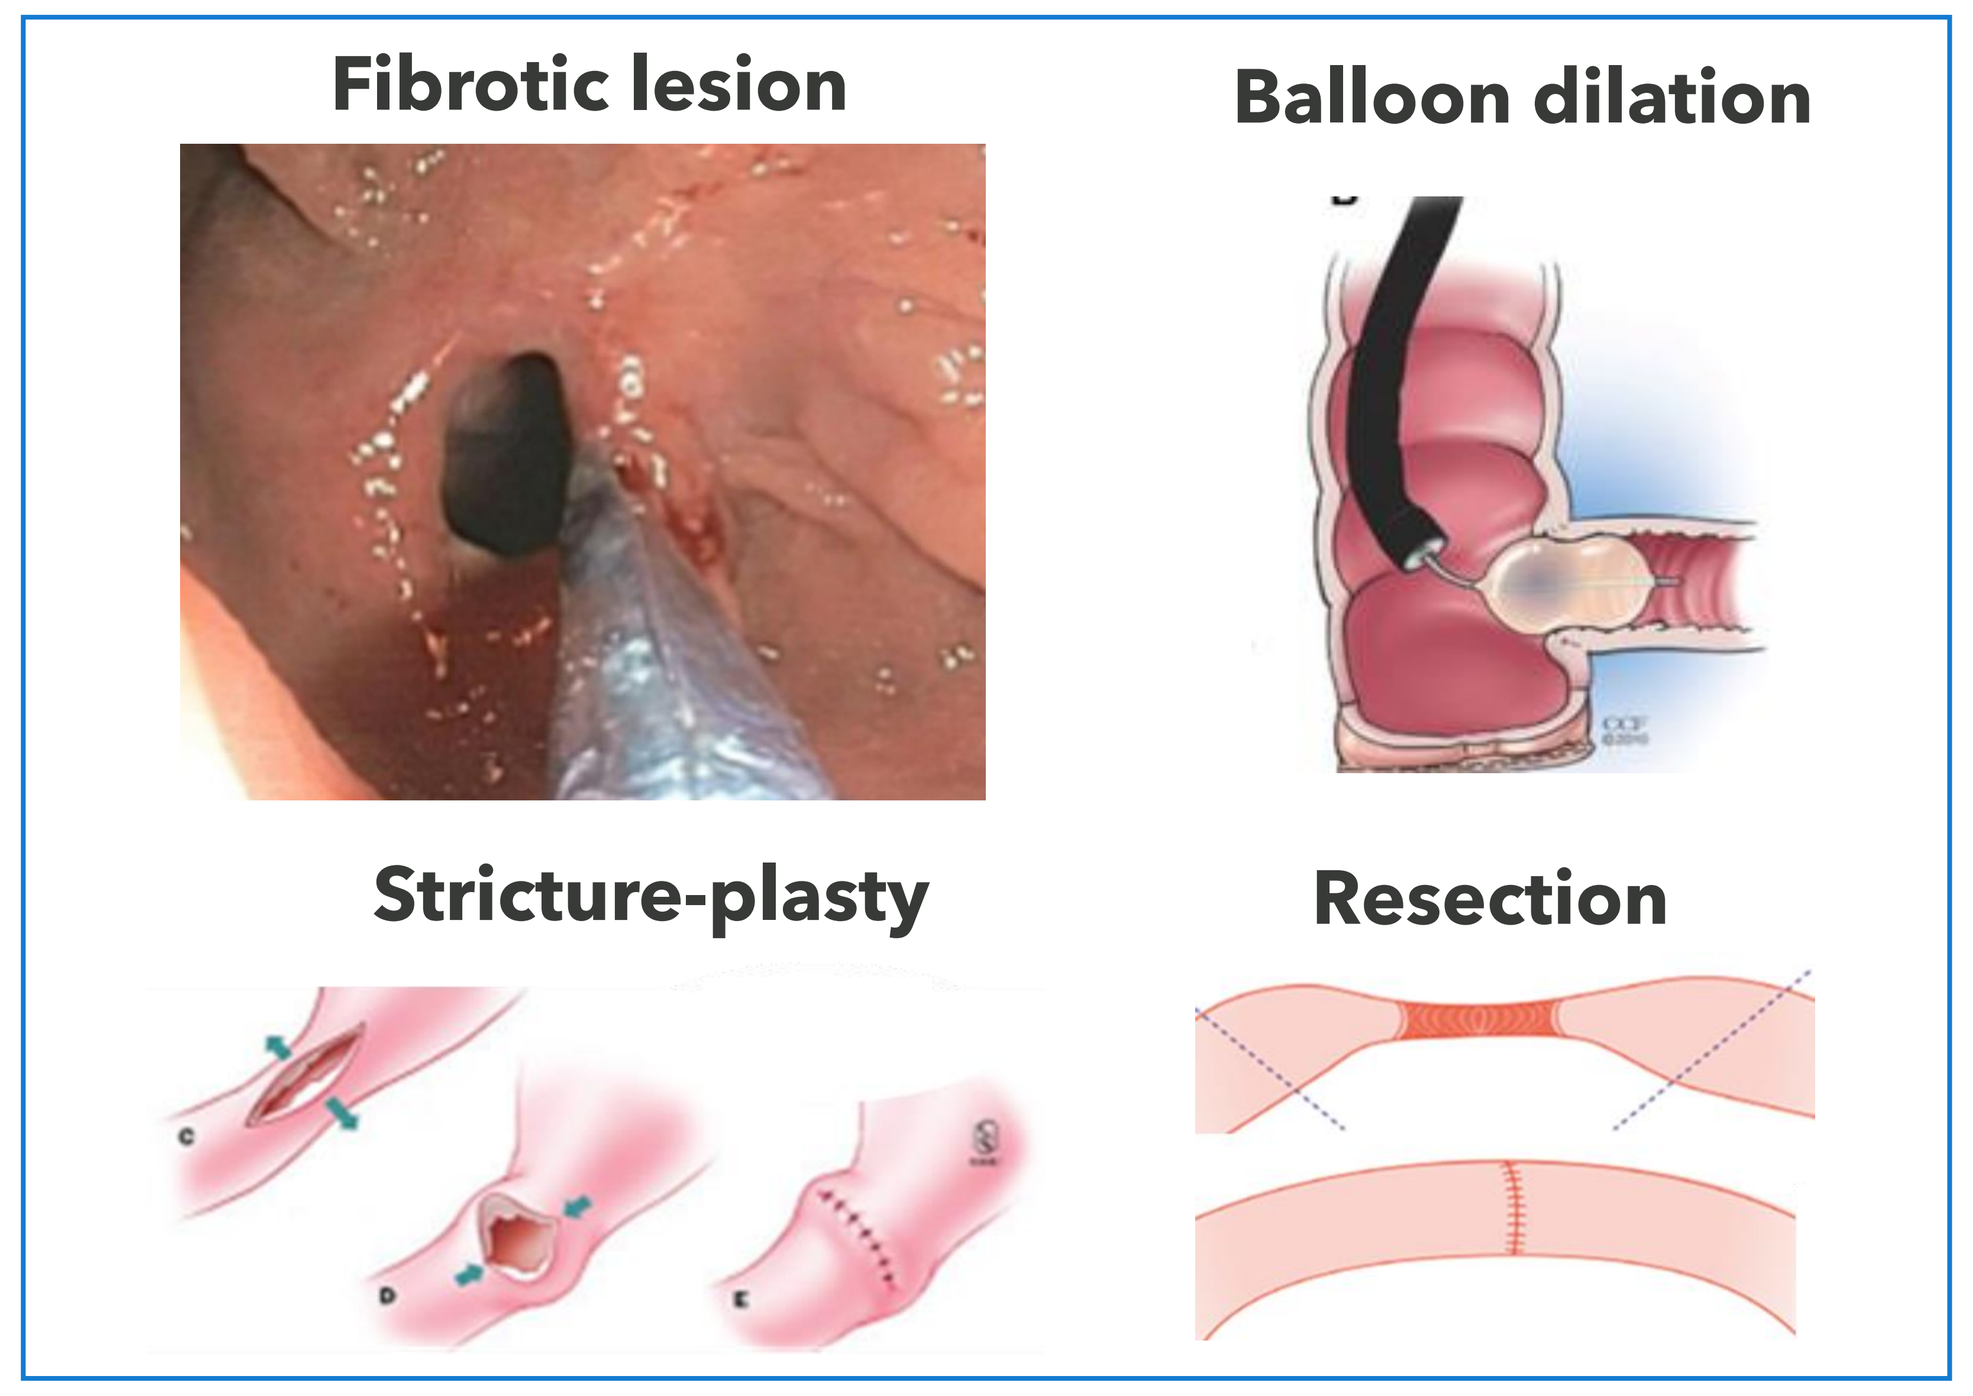

Fibrotic lesion
Balloon dilation
Stricture-plasty
Resection

Figure 1: Current Treatment strategies for Fibrostenosis

Rationale for Targeting ROCK in Crohn's Fibrostenosis

ROCK is a nodal point for pro-fibrotic signalling

ROCK signalling is involved in controlling several fibrotic responses such as fibroblast activation and migration, epithelial cell apoptosis, epithelial tight junction assembly and EMT (Epithelial to mesenchymal transition)

ROCK inhibitors can suppress fibrosis in: Animal models of lung, kidney, heart and liver fibrosis and human fibroblasts from diseased tissues

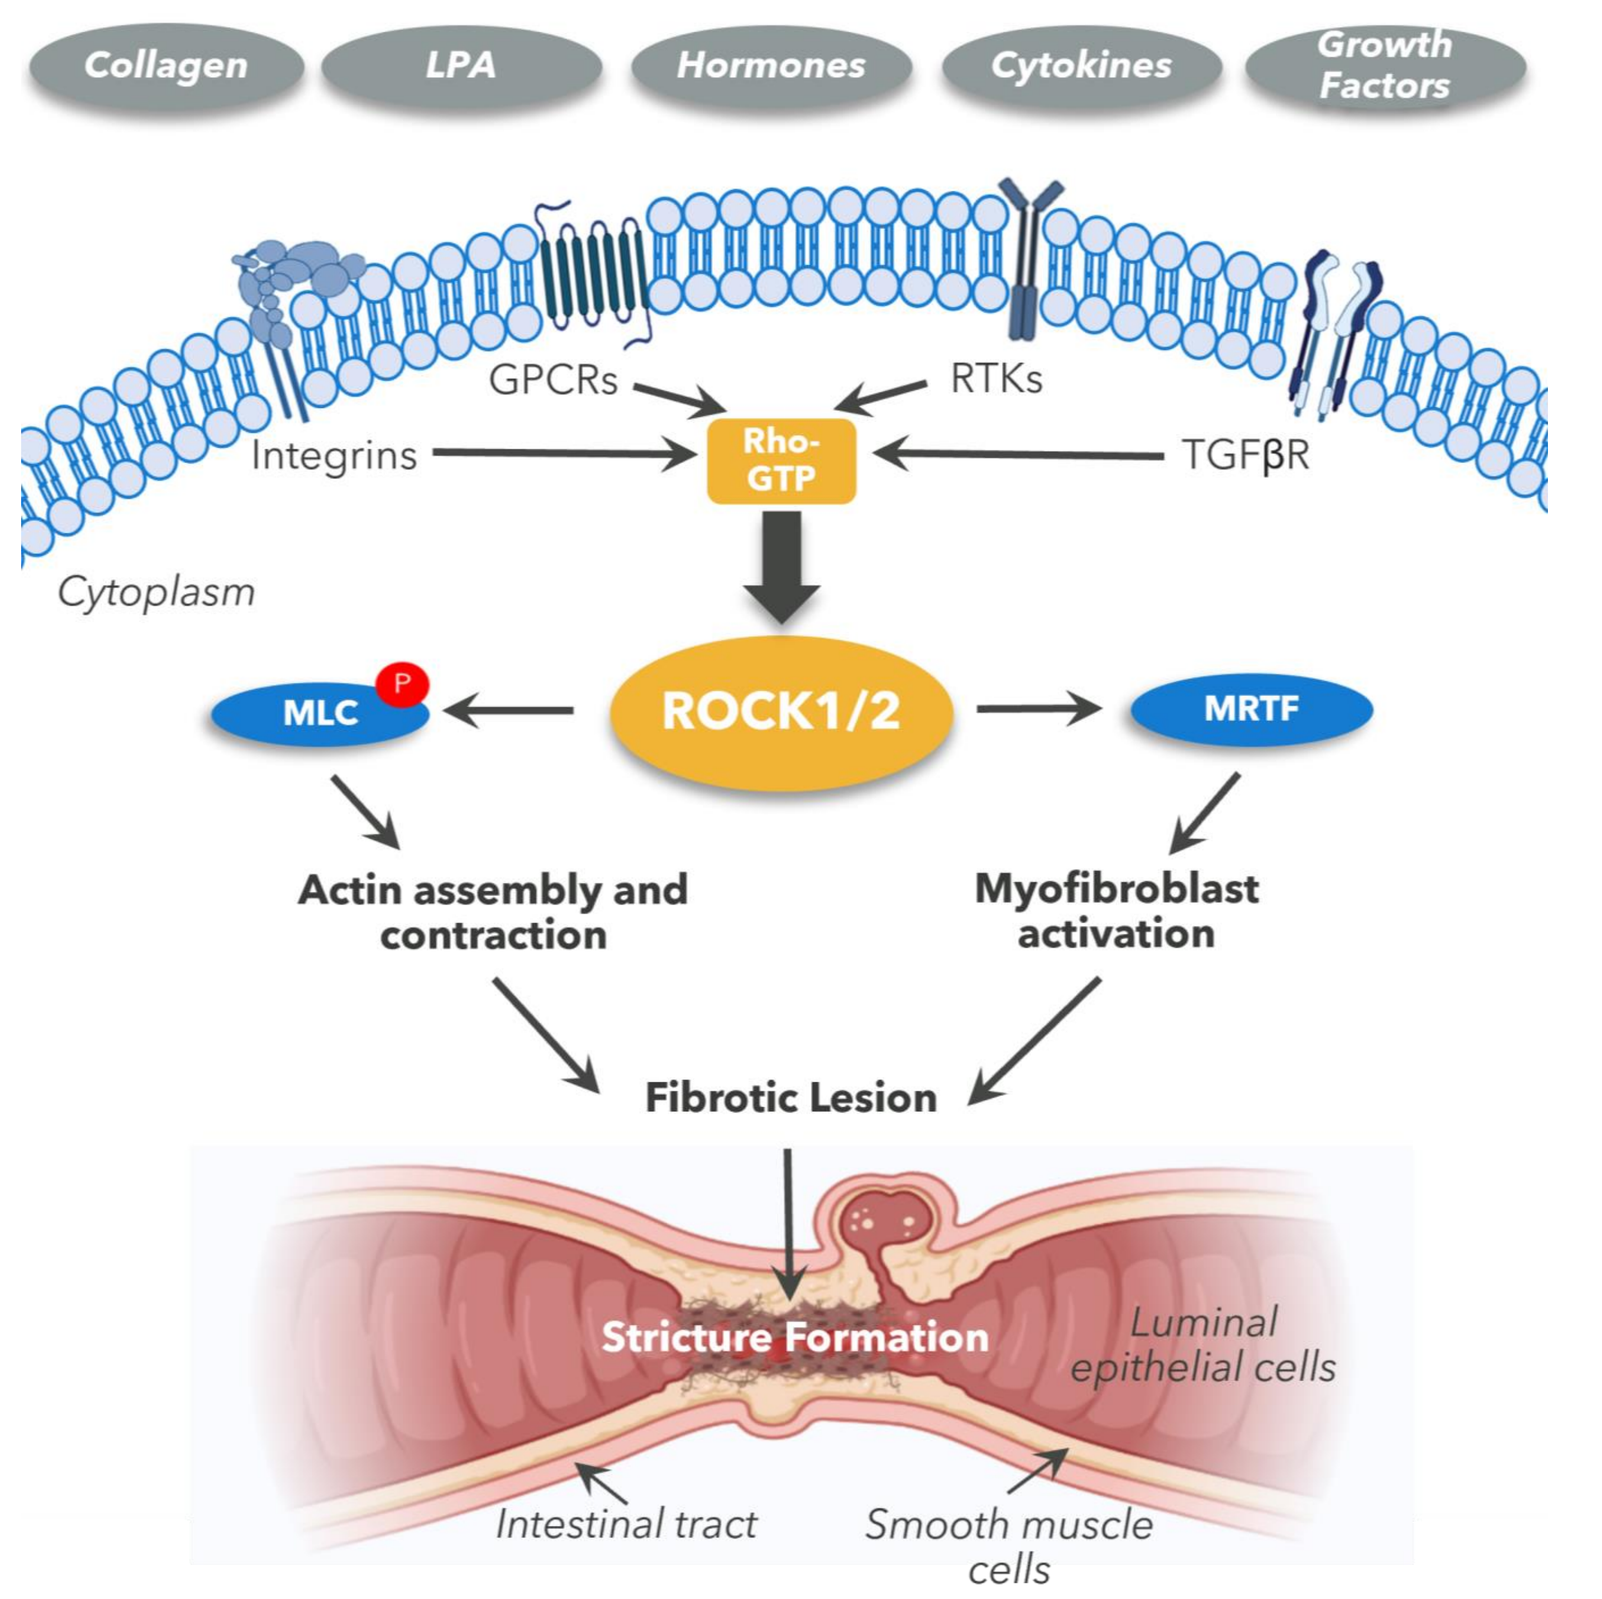

Collagen
LPA
Hormones
Cytokines
Growth Factors
GPCRs
RTKs
Integrins
Rho-GTP
TGFβR
Cytoplasm
MLC
ROCK1/2
MRTF
Actin assembly and contraction
Myofibroblast activation
Fibrotic Lesion
Stricture Formation
Luminal epithelial cells
Intestinal tract
Smooth muscle cells

Figure 2: Schematic representation of ROCK pathway in Fibrostenosis

GI Targeted ROCK Inhibitors are Designed to Avoid Hypotensive/CV Effects Associated with Systemic ROCK Inhibition

Systemic ROCK Inhibitors | **GI Restricted ROCK Inhibitors**

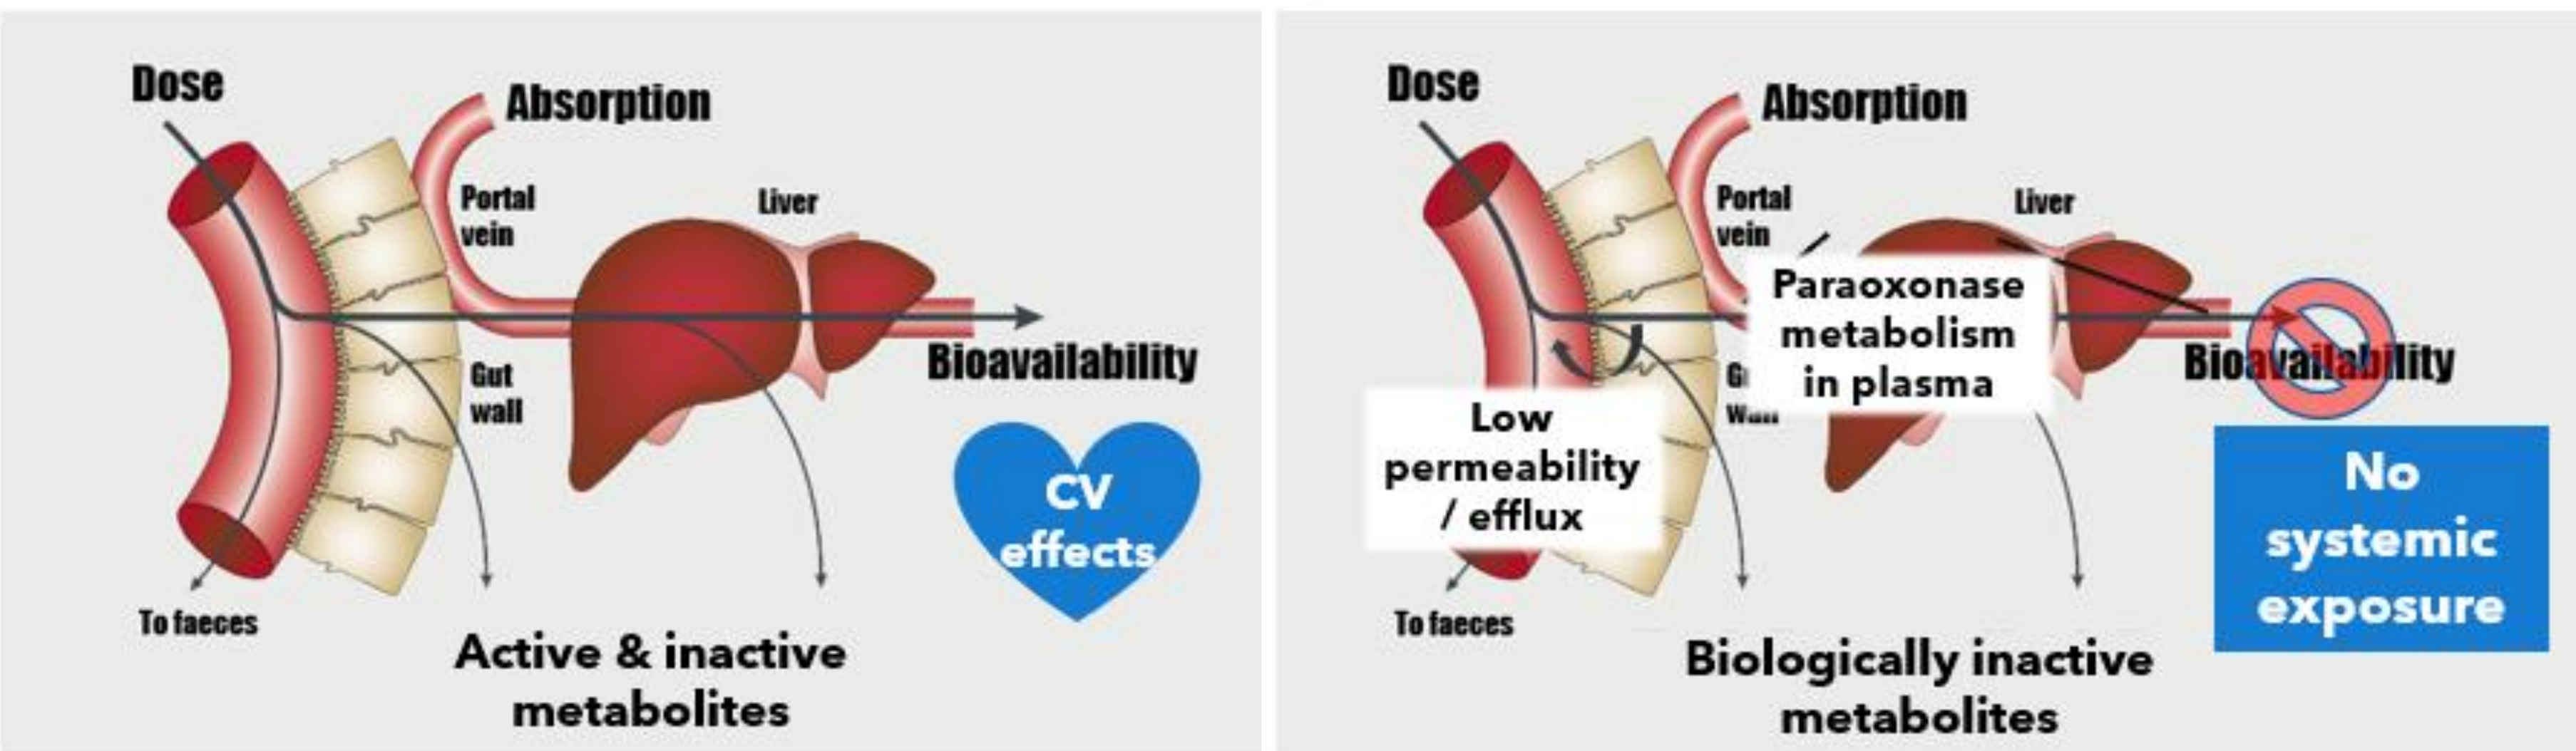

Dose
Absorption
Portal vein
Liver
Gut wall
Bioavailability
CV effects
To faeces
Active & inactive metabolites
Dose
Absorption
Portal vein
Liver
Paraoxonase metabolism in plasma
Low permeability / efflux
Bioavailability
No systemic exposure
To faeces
Biologically inactive metabolites

Figure 3: Schematic representation of Systemic ROCK inhibitors vs GI-targeted ROCK inhibitors

RESULTS

RXC008 is a potent inhibitor of ROCK1 and ROCK2 *in vitro*

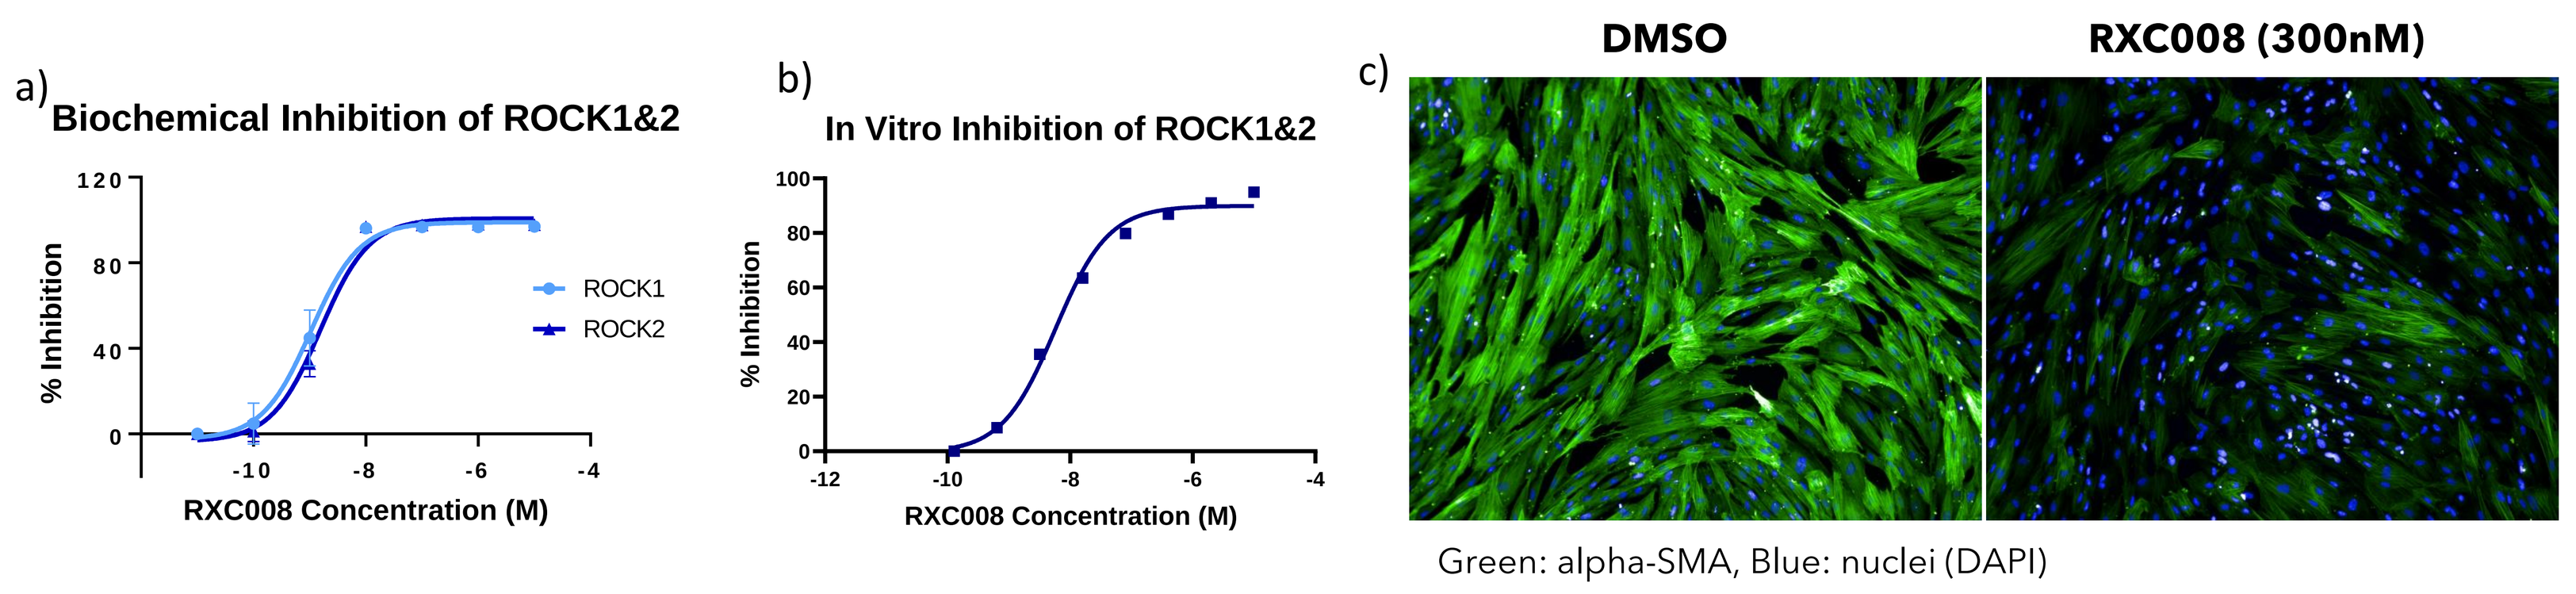

a) Biochemical Inhibition of ROCK1&2
% Inhibition
RXC008 Concentration (M)
ROCK1
ROCK2
b) In Vitro Inhibition of ROCK1&2
% Inhibition
RXC008 Concentration (M)
c) DMSO
RXC008 (300nM)
Green: alpha-SMA, Blue: nuclei (DAPI)

Figure 4: RXC008 inhibits ROCK1 and ROCK2. a) Both ROCK1 and ROCK2 are inhibited with similar nanomolar potency in ADP-Glo based ROCK kinase activity assays, b) RXC008 inhibits ROCK1&2 mediated phospho-MYPT1 in MCF7 cells, c) RXC008 inhibits alpha-SMA production in TGFβ stimulated Human Intestinal Fibroblasts.

RXC008 Reduces Fibrosis in an Adoptive Transfer Model

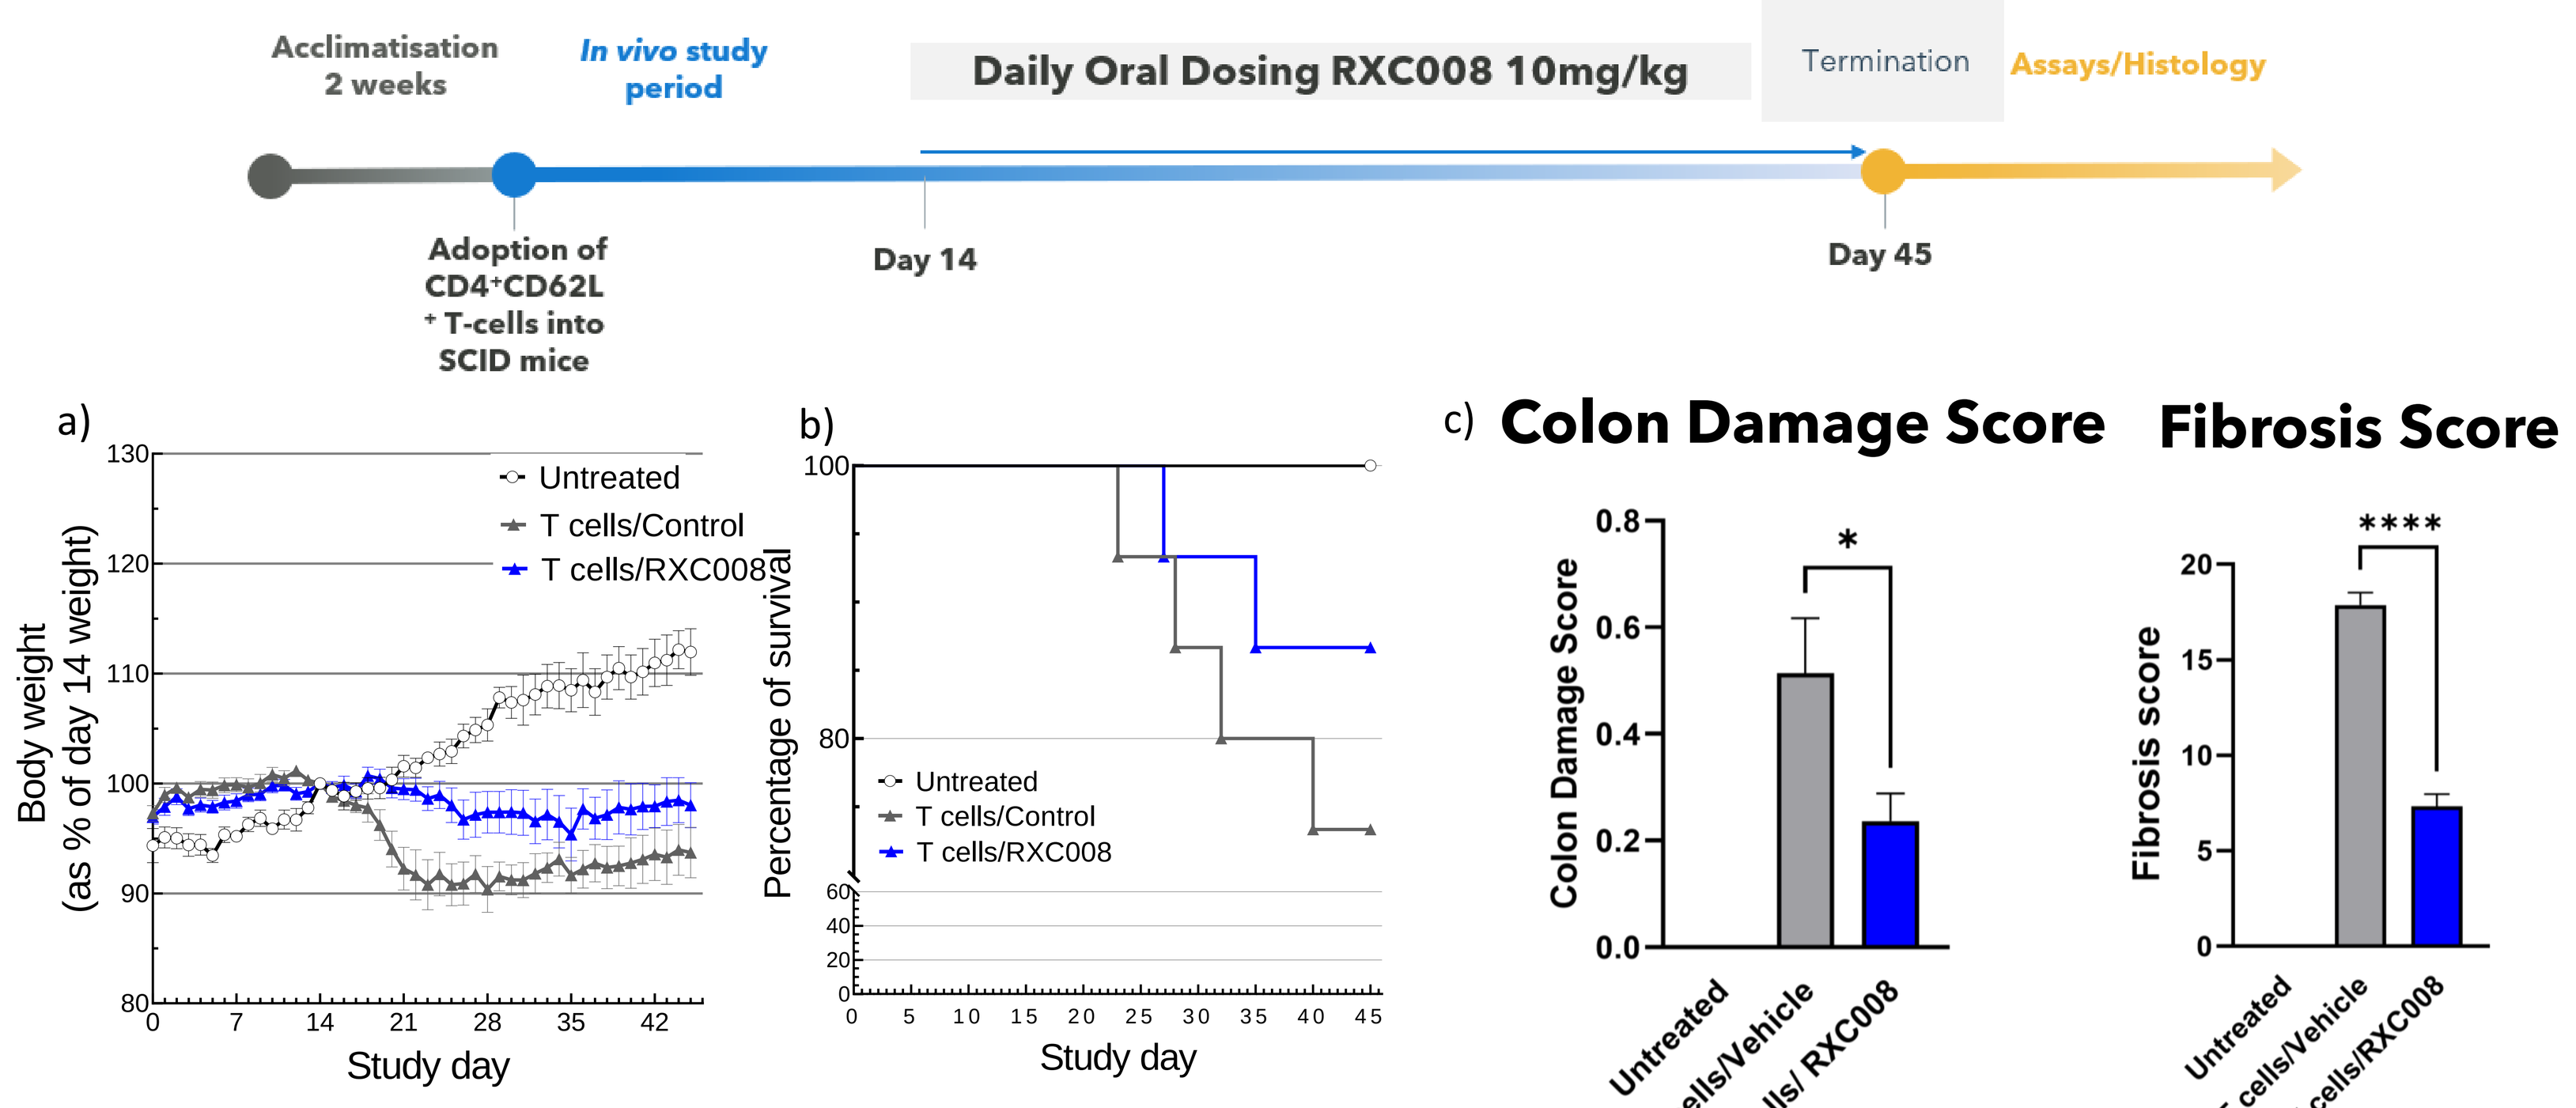

Acclimatisation 2 weeks
In vivo study period
Daily Oral Dosing RXC008 10mg/kg
Termination
Assays/Histology
Adoption of CD4+CD62L+ T-cells into SCID mice
Day 14
Day 45
a)
Body weight (as % of day 14 weight)
Study day
Untreated
T cells/Control
T cells/RXC008
b)
Percentage of survival
Study day
Untreated
T cells/Control
T cells/RXC008
c) Colon Damage Score
Fibrosis Score
Colon Damage Score
Fibrosis score
Untreated
T cells/Vehicle
T cells/ RXC008
T cells/RXC008

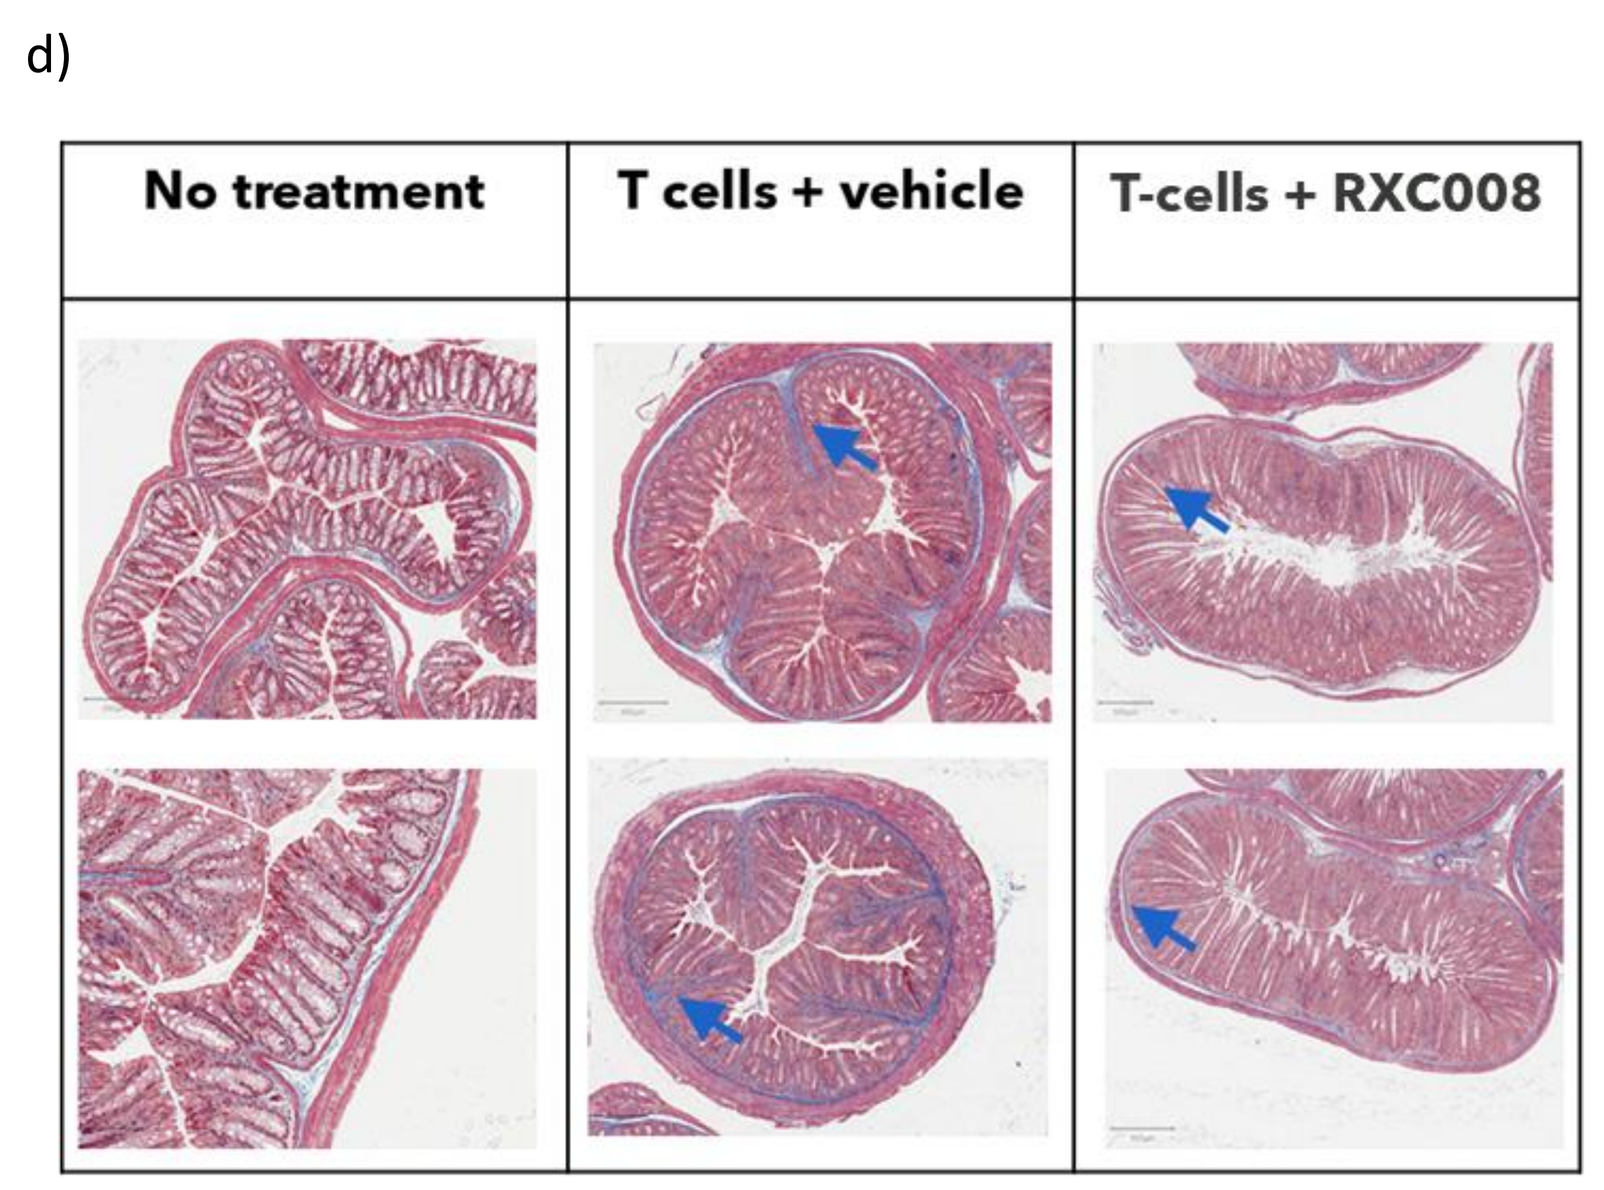

d)
No treatment
T cells + vehicle
T-cells + RXC008

Figure 5: RXC008 (10mg/kg) reduces Fibrosis in an Adoptive Transfer Model of Colitis. a) RXC008 treated mice had less body weight loss than vehicle control treated animals, b) RXC008 treated mice had higher percentage survival than vehicle control treated animals, c) Colon and Fibrosis Scores were significantly lower in RXC008 treated mice compared to vehicle control treated animals, d) Representative Trichrome staining showing reduced Fibrosis is RXC008 treated mice.

RXC008 Reduces Fibrosis in a Chronic DSS Model

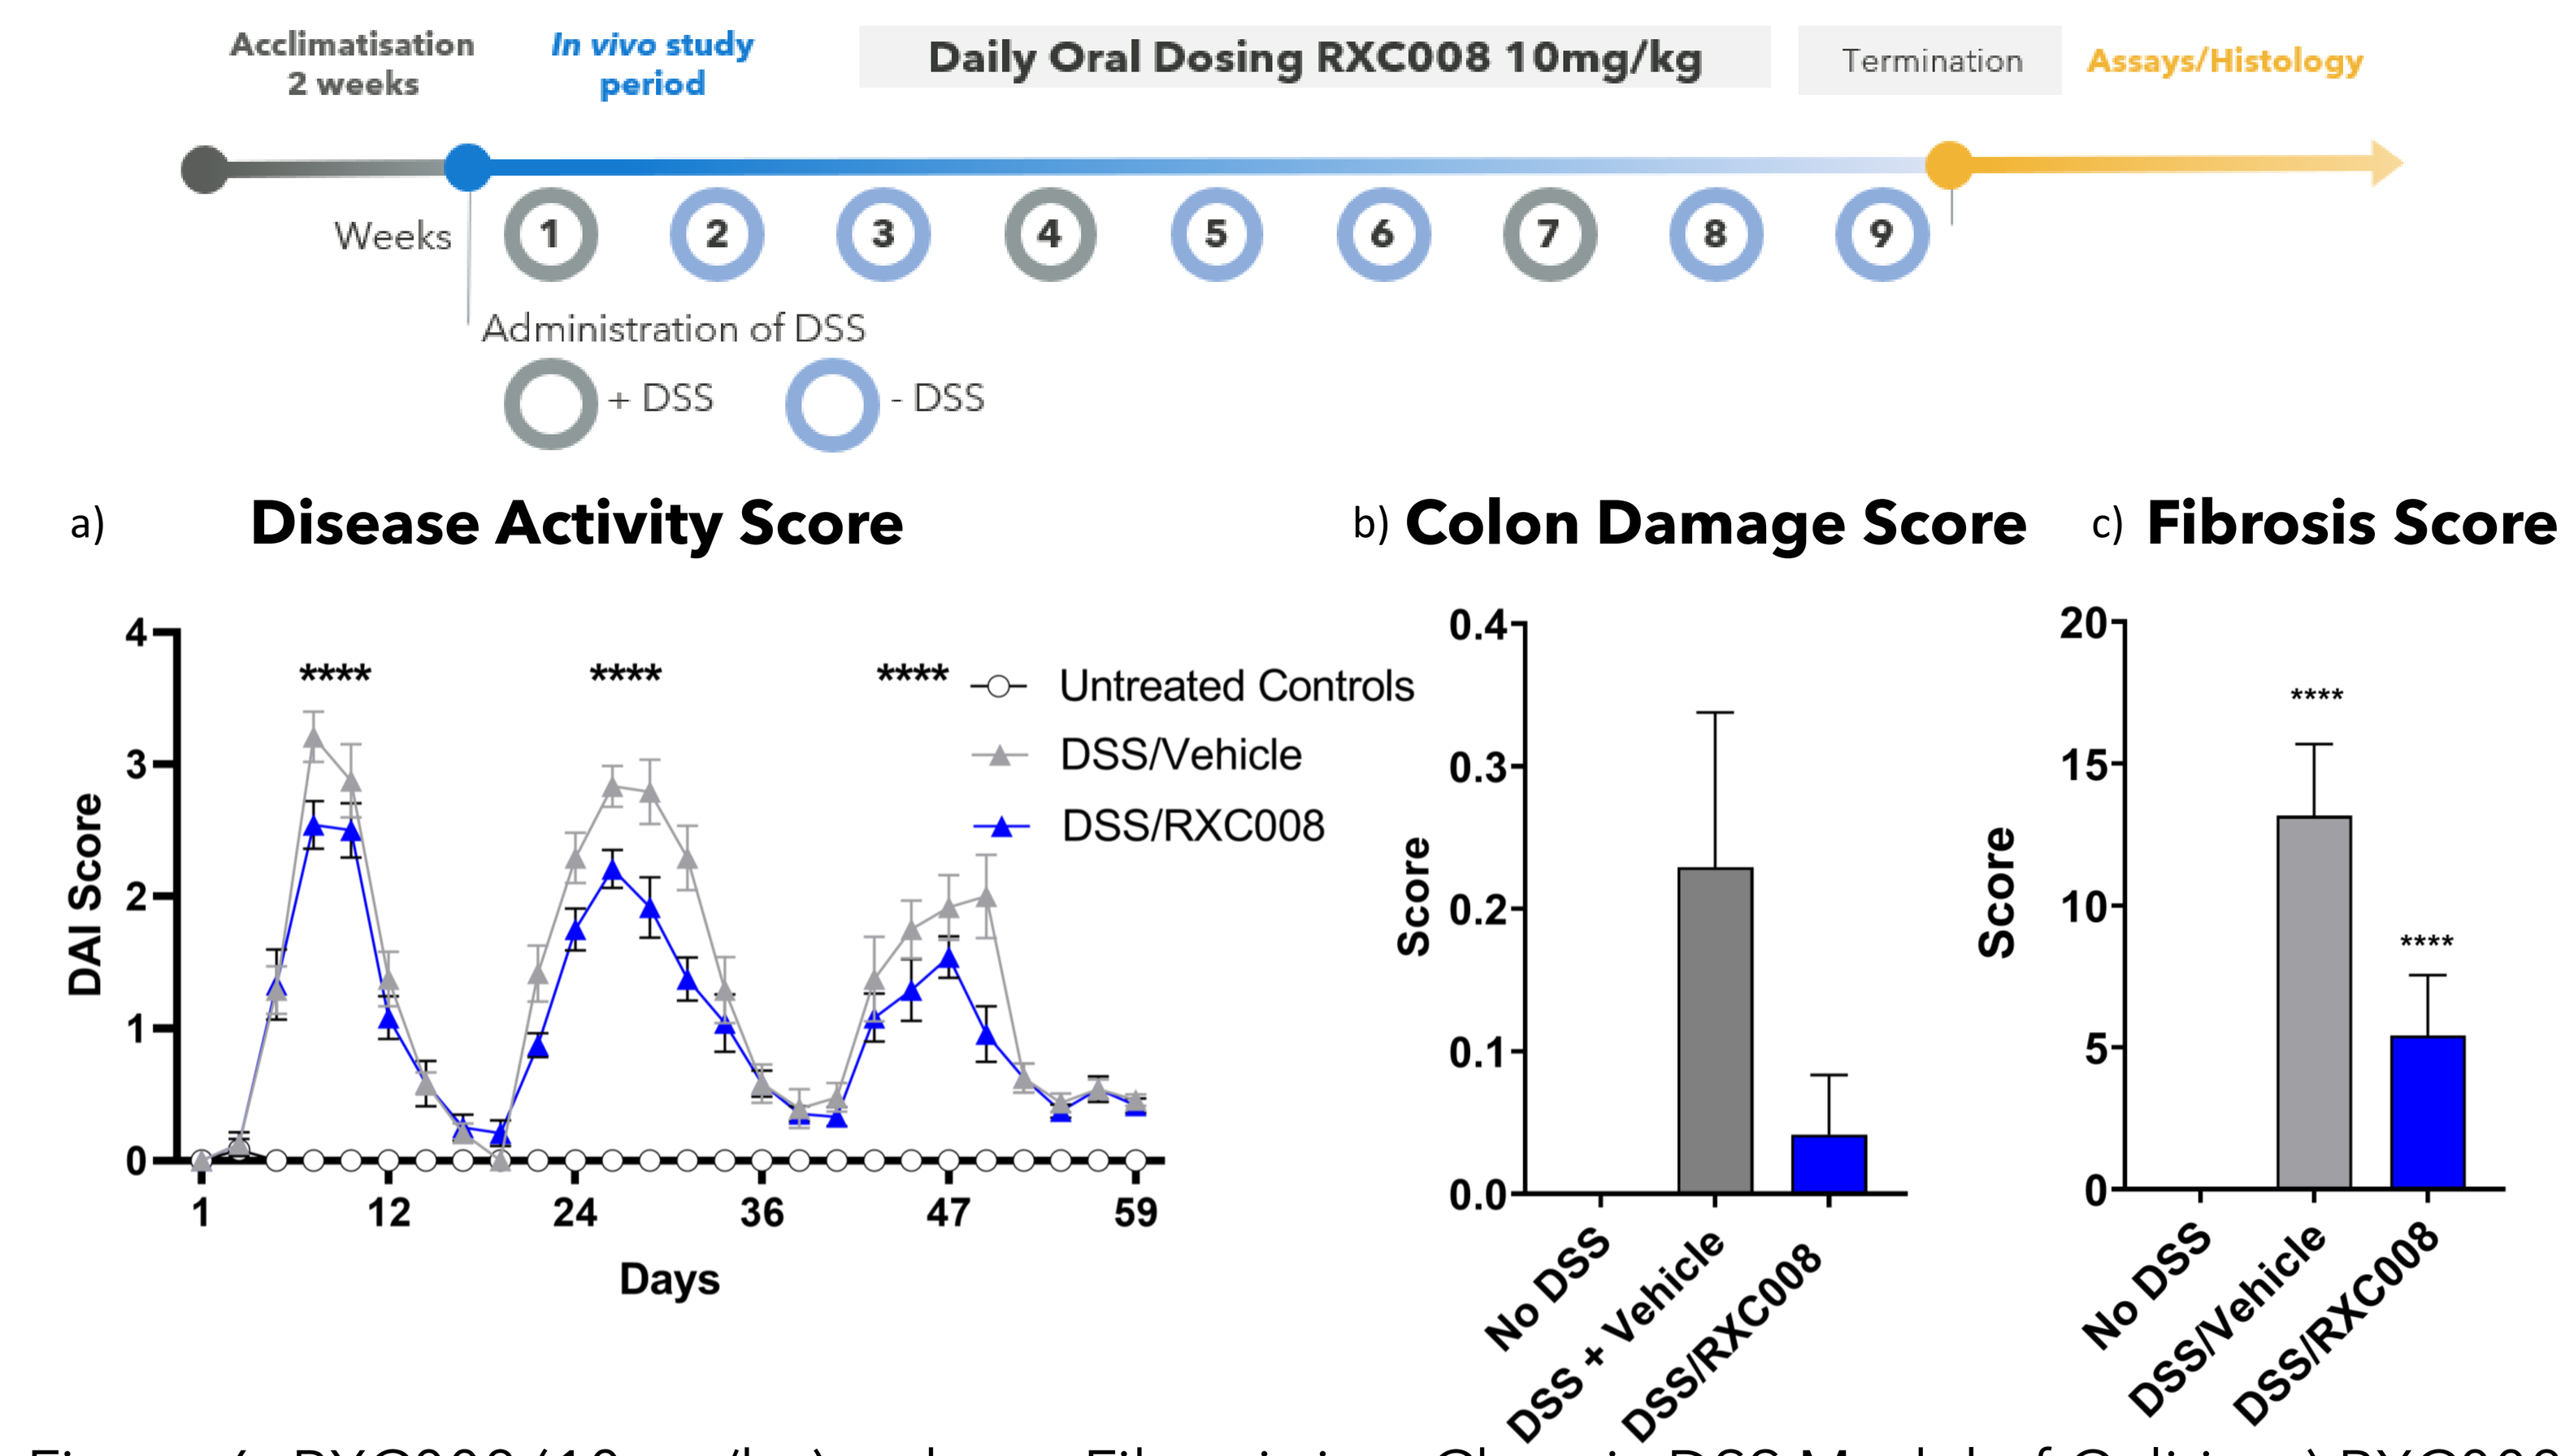

Acclimatisation 2 weeks
In vivo study period
Daily Oral Dosing RXC008 10mg/kg
Termination
Assays/Histology
Weeks
Administration of DSS
+ DSS
- DSS
a) Disease Activity Score
DAI Score
Days
Untreated Controls
DSS/Vehicle
DSS/RXC008
b) Colon Damage Score
Score
No DSS
DSS + Vehicle
DSS/RXC008
c) Fibrosis Score
Score
No DSS
DSS/Vehicle
DSS/RXC008

Figure 6: RXC008 (10mg/kg) reduces Fibrosis in a Chronic DSS Model of Colitis. a) RXC008 treated mice had lower disease activity score than vehicle control treated animals, b) RXC008 treated mice had slightly lower colon damage than vehicle control treated animals, c) RXC008 treated mice had a lower fibrosis score than vehicle control treated animals.

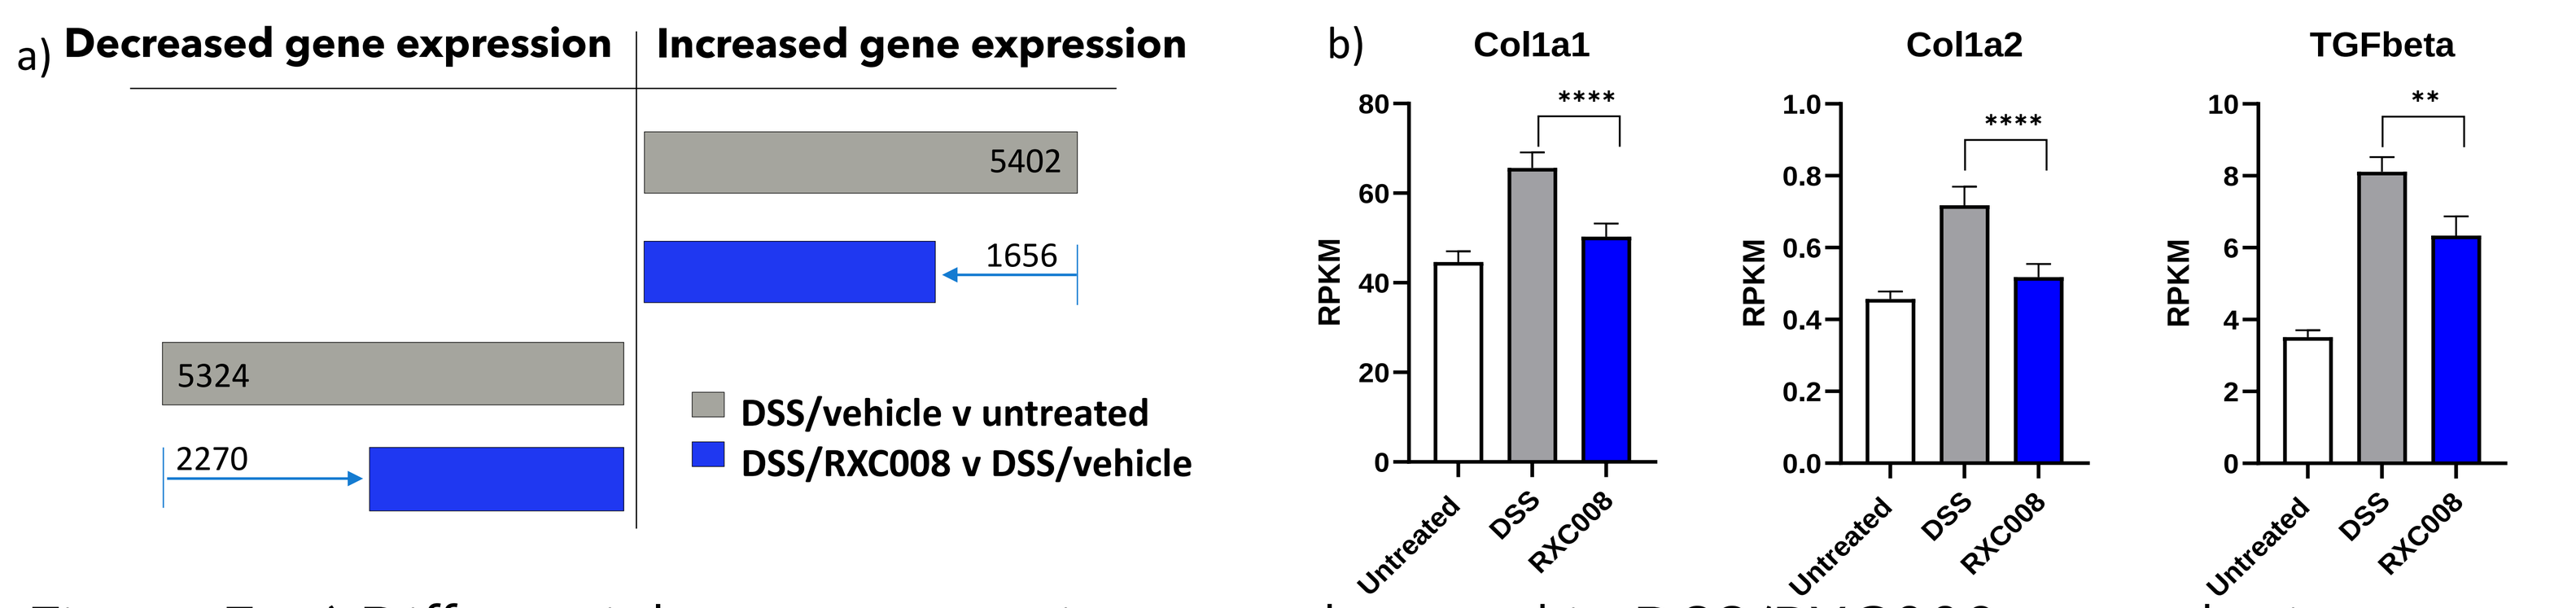

a) Decreased gene expression
Increased gene expression
5402
1656
5324
2270
DSS/vehicle v untreated
DSS/RXC008 v DSS/vehicle
b) Col1a1
Col1a2
TGFbeta
RPKM
Untreated
DSS
RXC008

Figure 7: a) Differential gene expression was observed in DSS/RXC008 treated mice compared to DSS/vehicle control, b) examples of fibrosis related genes regulated by RXC008.

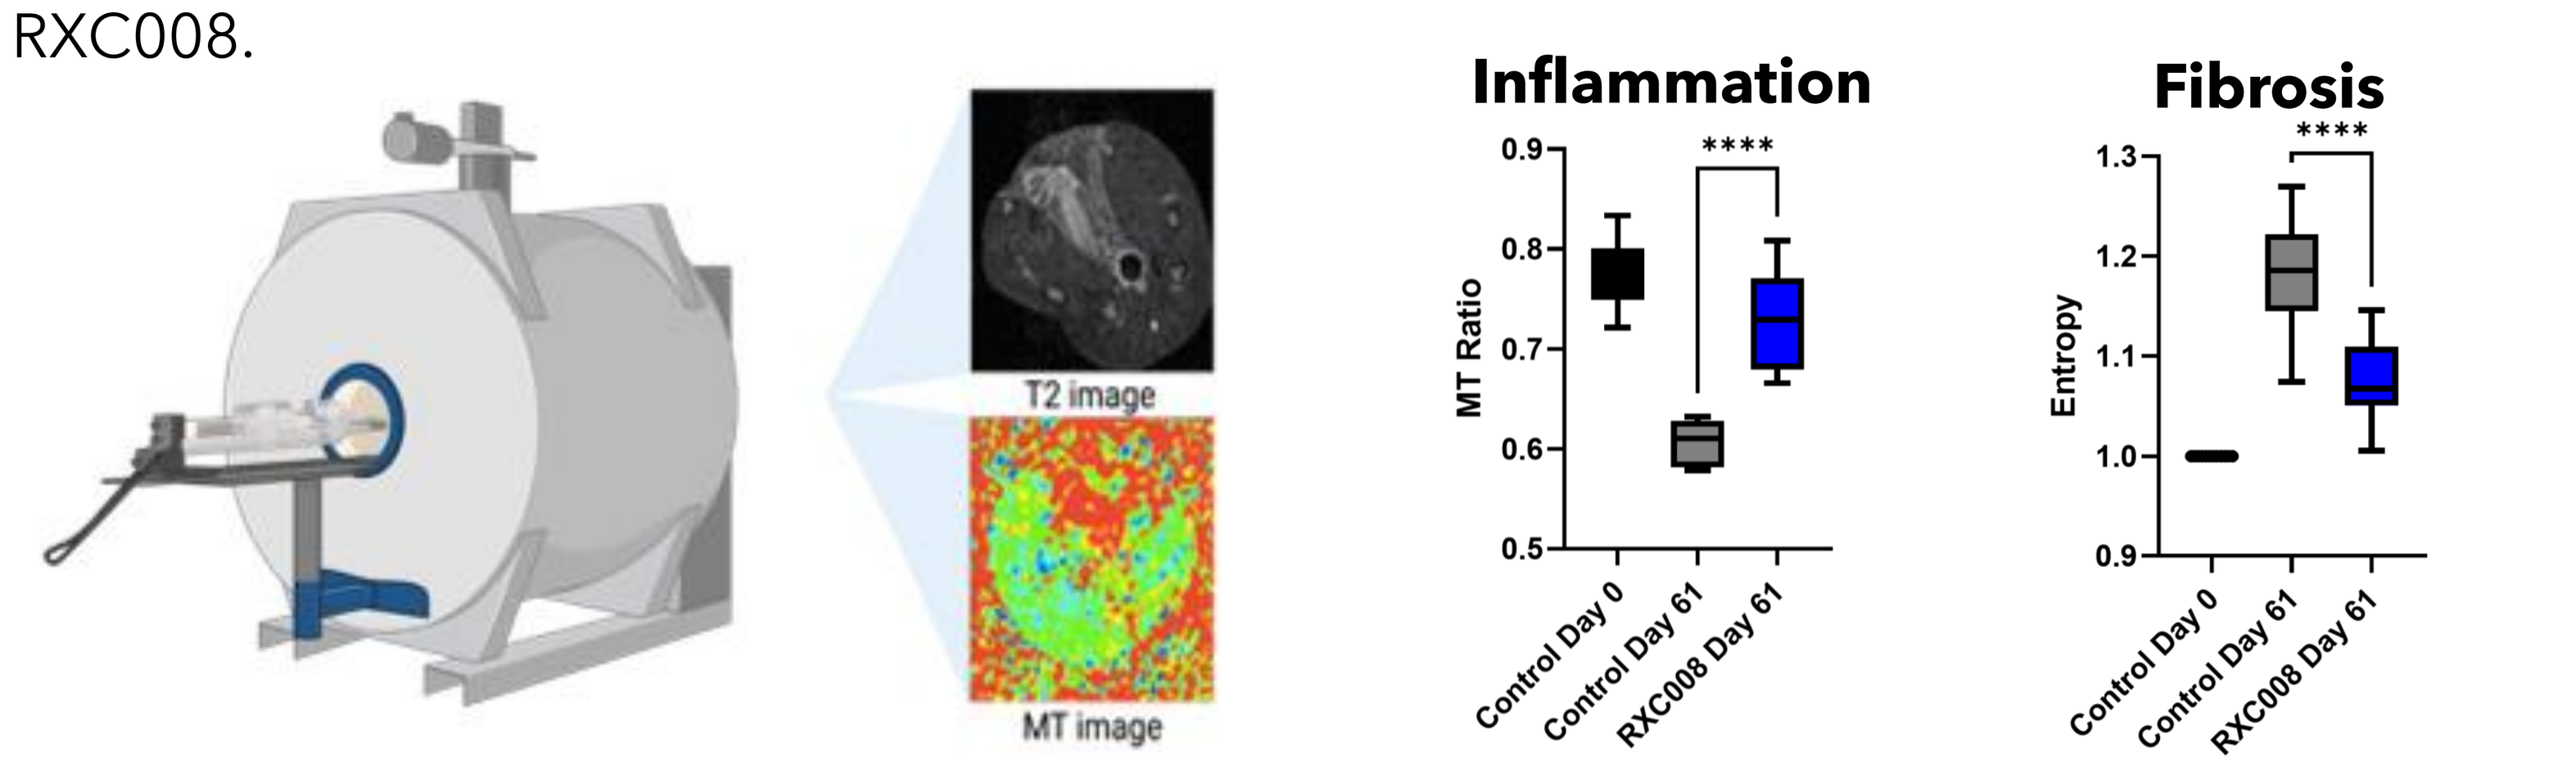

T2 image
MT image
Inflammation
MT Ratio
Fibrosis
Entropy
Control Day 0
Control Day 61
RXC008 Day 61

Figure 8: MRI Imaging Shows Improved Tissue Injury and Fibrosis

SUMMARY

- RXC008 is a potent, selective, GI restricted inhibitor of ROCK1 and ROCK2.
- RXC008 can suppress fibrosis and attenuate tissue injury in animal models of GI fibrosis and has the potential to be developed as a novel therapy to inhibit fibrotic stricture formation in Crohn's disease.
- IND enabling activities are progressing with the aim to commence Phase 1 clinical trial by the end of 2023.

REFERENCES

Chan et al: Crohn's disease associated strictures. J Gastroenterol Hepatol. 2018 May; 33(5):998-1008.
Noma, K. et al: Physiological role of ROCKs in the cardiovascular system. Am J Physiol Cell Physiol. 2006 Mar;290(3):C661-8.
Holvoet, T. et al: Treatment of Intestinal Fibrosis in Experimental Inflammatory Bowel Disease by the Pleiotrophic Actions of a Local Rho Kinase Inhibitor. Gastroenterology. 2017 Oct;153(4):1054-1067.
Biancheri, P. et al: The role of transforming growth factor (TGF)-β in modulation the immune response and fibrogenesis in the gut. Cytokine Growth Factor Rev. 2014 Feb;25(1):45-55.

IBD NORDIC CONFERENCE 2022
23-25 November | Malmo

https://www.redxpharma.com