Potential of RXC007, a highly selective Rho-Associated Coiled Kinase 2 (ROCK2) inhibitor, to tackle fibrotic lung disease

Nicolas E.S. Guisot, Adriana Gambardella, Peter R. Bunyard ; Redx Pharma Plc

Redx

Introduction

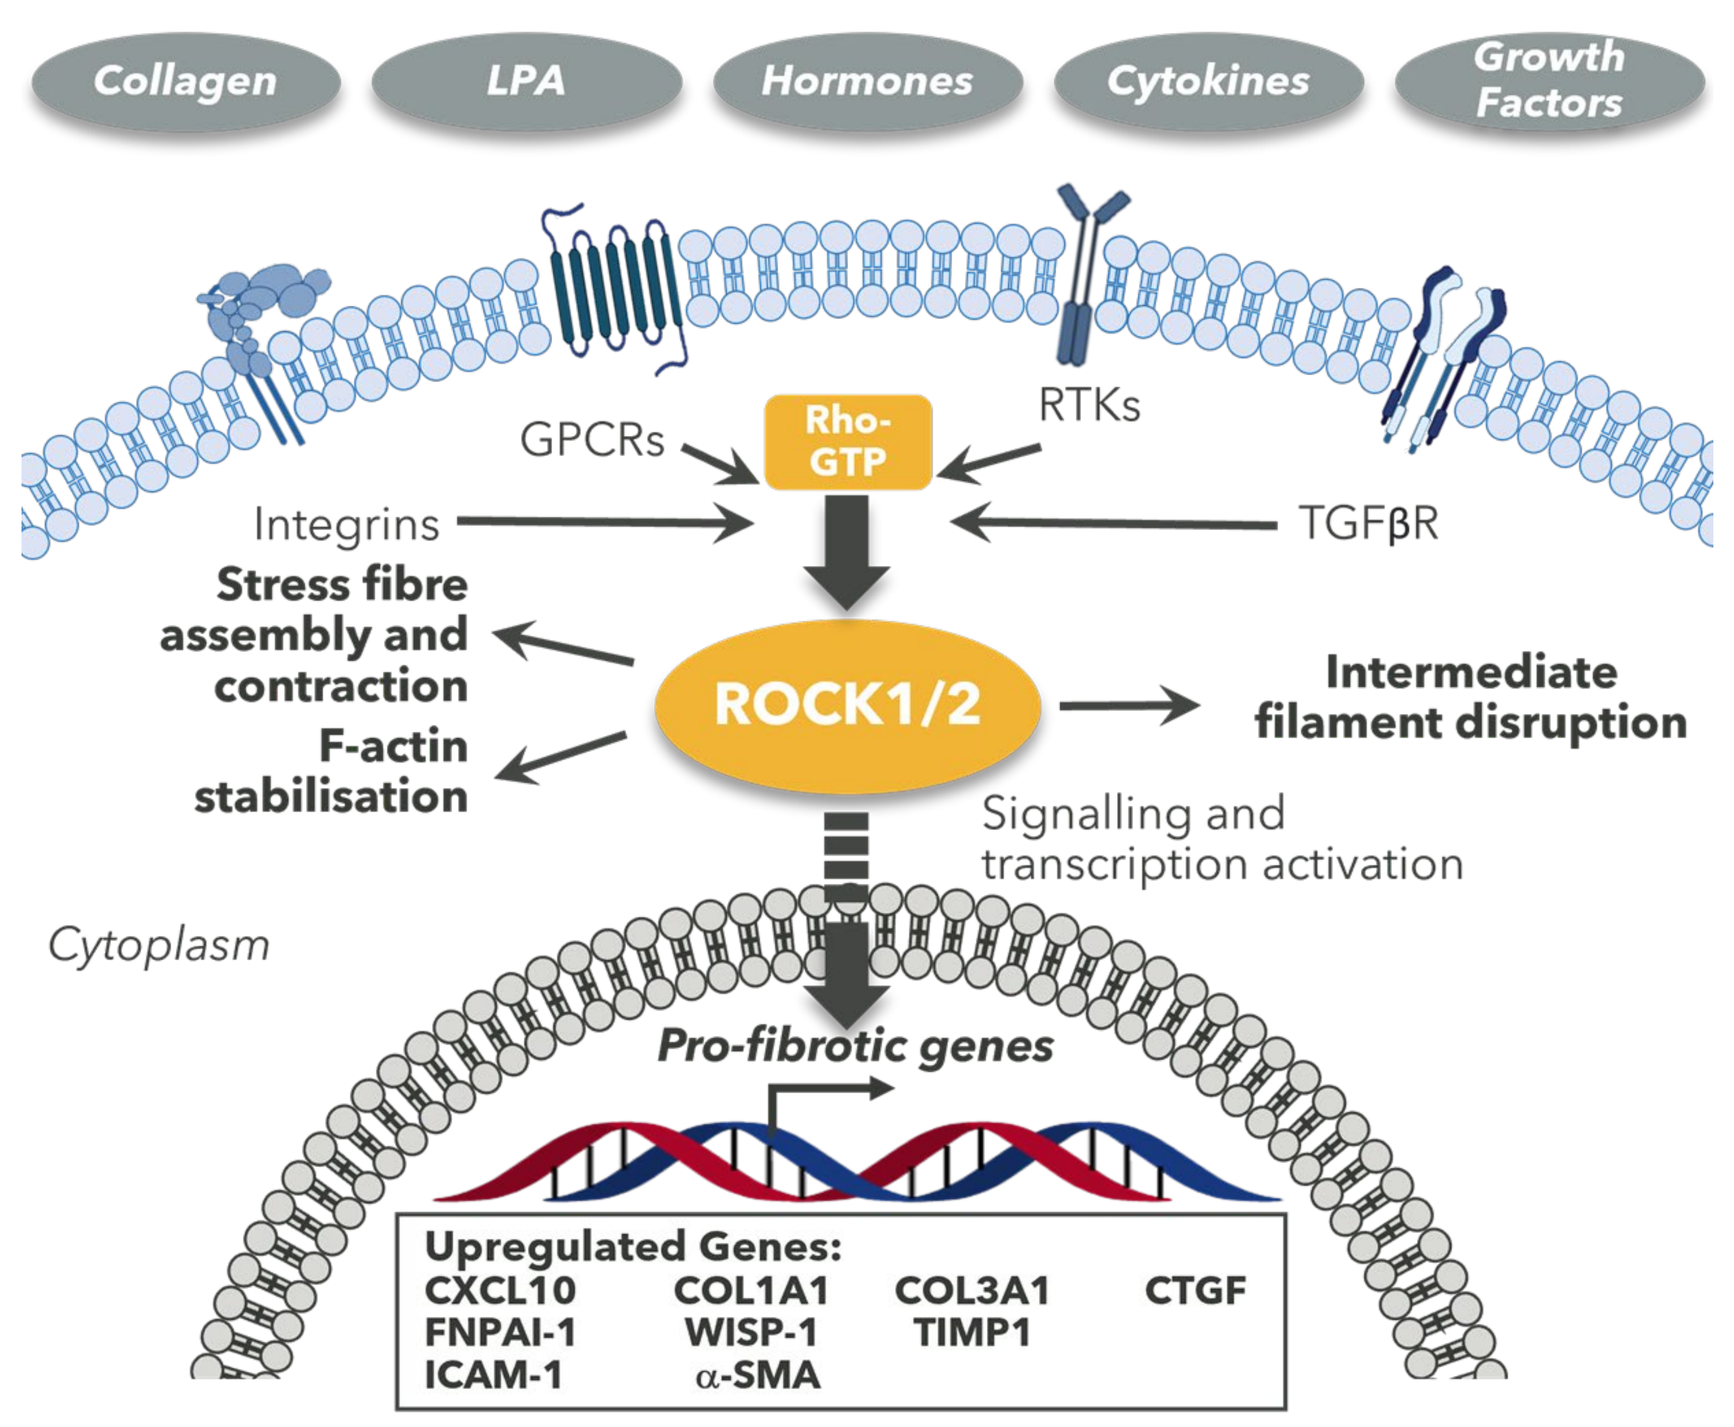

Collagen
LPA
Hormones
Cytokines
Growth Factors
GPCRs
Rho-GTP
RTKs
Integrins
TGFβR
Stress fibre assembly and contraction
F-actin stabilisation
ROCK1/2
Intermediate filament disruption
Signalling and transcription activation
Cytoplasm
Pro-fibrotic genes
Upregulated Genes:
CXCL10
FNPAI-1
ICAM-1
COL1A1
WISP-1
α-SMA
COL3A1
TIMP1
CTGF

- ROCK is a member of the AGC family of serine/threonine kinases that are involved in many aspects of cellular signalling
- RhoA/ROCK signalling closely involved in regulating cell morphology, growth, migration, and apoptosis
- Highly conserved in mammalian species, two isoforms of ROCK are known to exist: ROCK1 and ROCK2
- Both ROCK1 and ROCK2 are involved in numerous pro-fibrotic processes with pan-ROCK inhibitors able to strongly suppress TGFβ-stimulated myofibroblast activation, chemokine-driven fibroblast migration, and EMT in response to profibrotic mediators such as lysophosphatidic acid (LPA) and endothelin
- Systemic administration of pan-ROCK inhibitors induce vasodilation, hyperemia and hypotension, limiting their use as therapeutic agents but selective ROCK2 inhibition circumvents these side-effects

Rationale for ROCK2 Inhibition in Interstitial Lung Diseases and Idiopathic Pulmonary Fibrosis

ROCK activity in mouse & human lung tissue

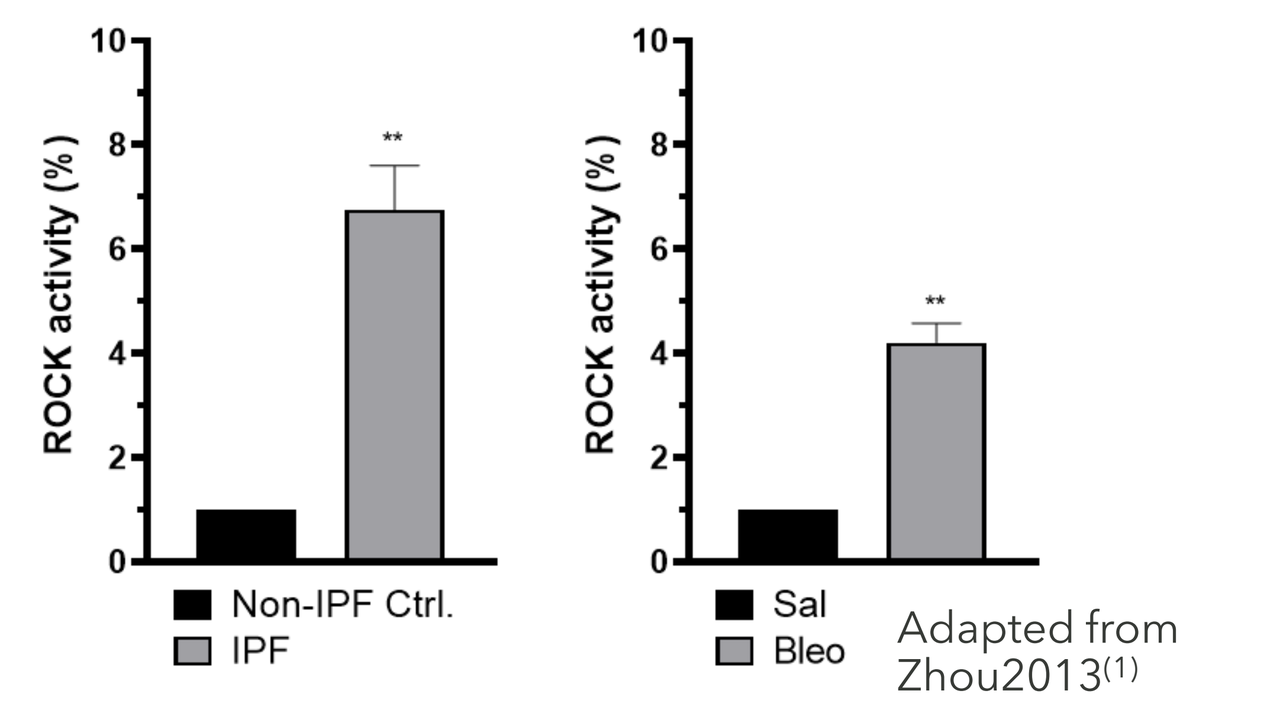

Human
Mouse
ROCK activity (%)
Non-IPF Ctrl.
IPF
Sal
Bleo
Adapted from Zhou2013(1)

Fibroblasts & myofibroblasts isolated from lungs of patients with IPF (n = 8), non-IPF control subjects (n = 6)
Fibroblasts & myofibroblasts isolated from lungs of bleomycin-treated mice (n = 8), and saline-treated mice (n = 8)
ROCK activity was measured by exogenous phosphorylation of a substrate peptide

- ROCK activity significantly increased in fibroblasts isolated from human IPF tissue and bleomycin-induced murine lung fibrosis

ROCK2 inhibition is sufficient to protect from bleomycin-induced lung fibrosis in mouse models

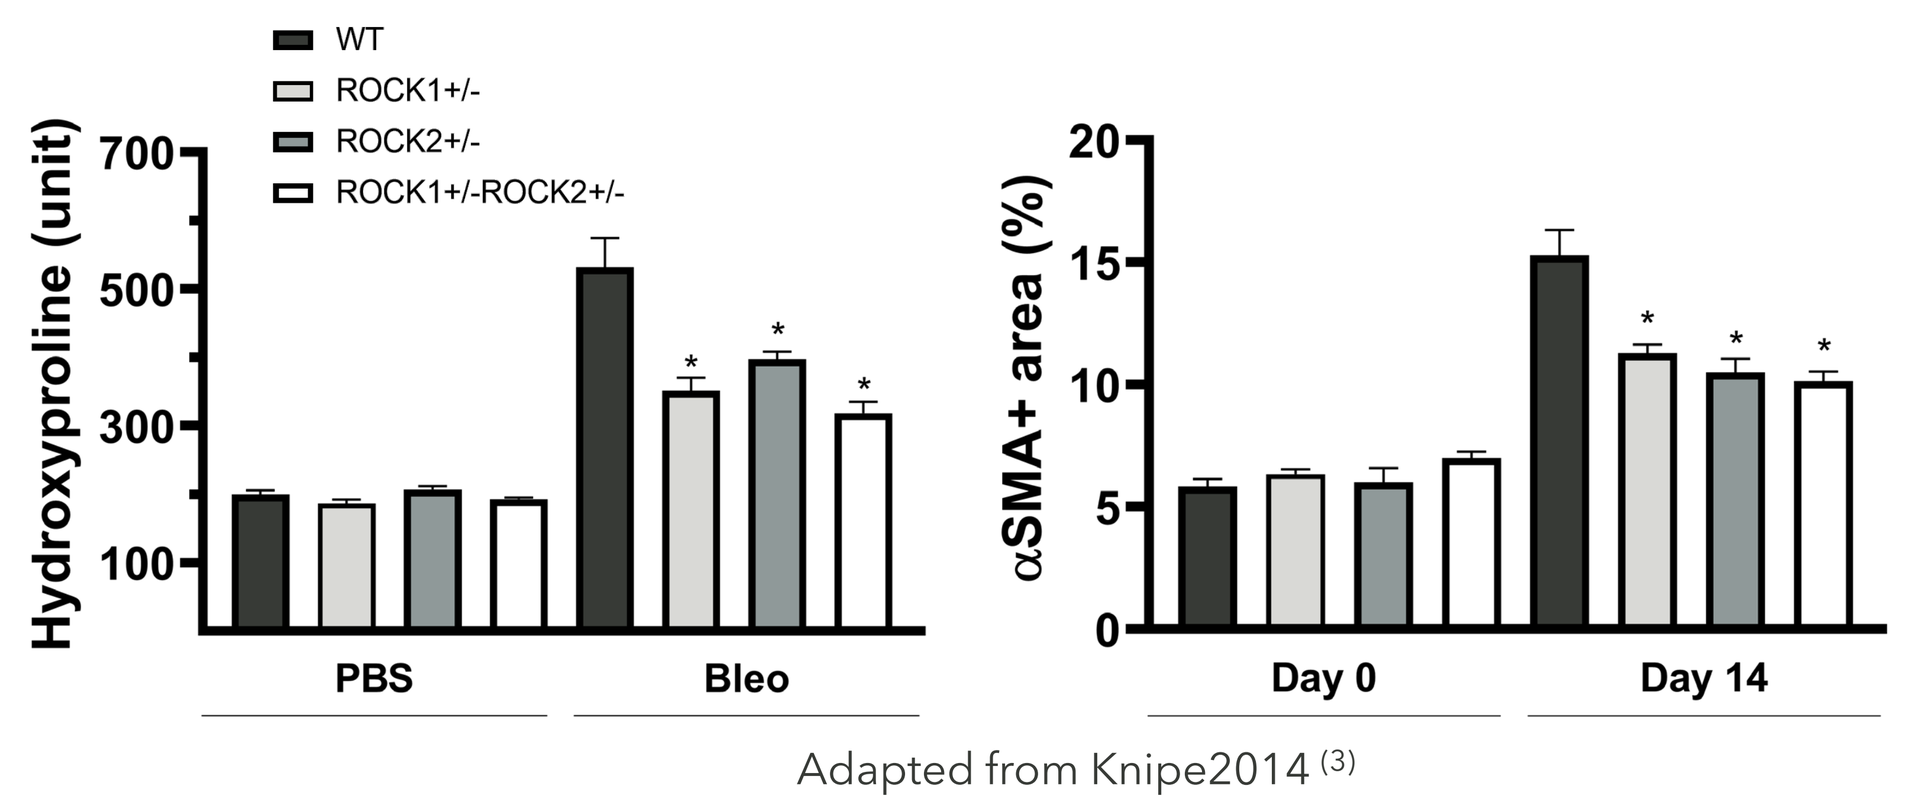

WT
ROCK1+/-
ROCK2+/-
ROCK1+/-ROCK2+/-
Hydroxyproline (unit)
αSMA+ area (%)
PBS
Bleo
Day 0
Day 14
Adapted from Knipe2014 (3)

- Haploinsufficient ROCK1, ROCK2 and ROCK1/ROCK2 mice show reduced hydroxyproline expression in bleomycin induced lung fibrosis at day 14
- Similar reduction in α-SMA expression in fibroblasts is observed in the 3 haploinsufficient phenotypes

IHC analysis of ROCK2 expression in resected IPF lung tissue

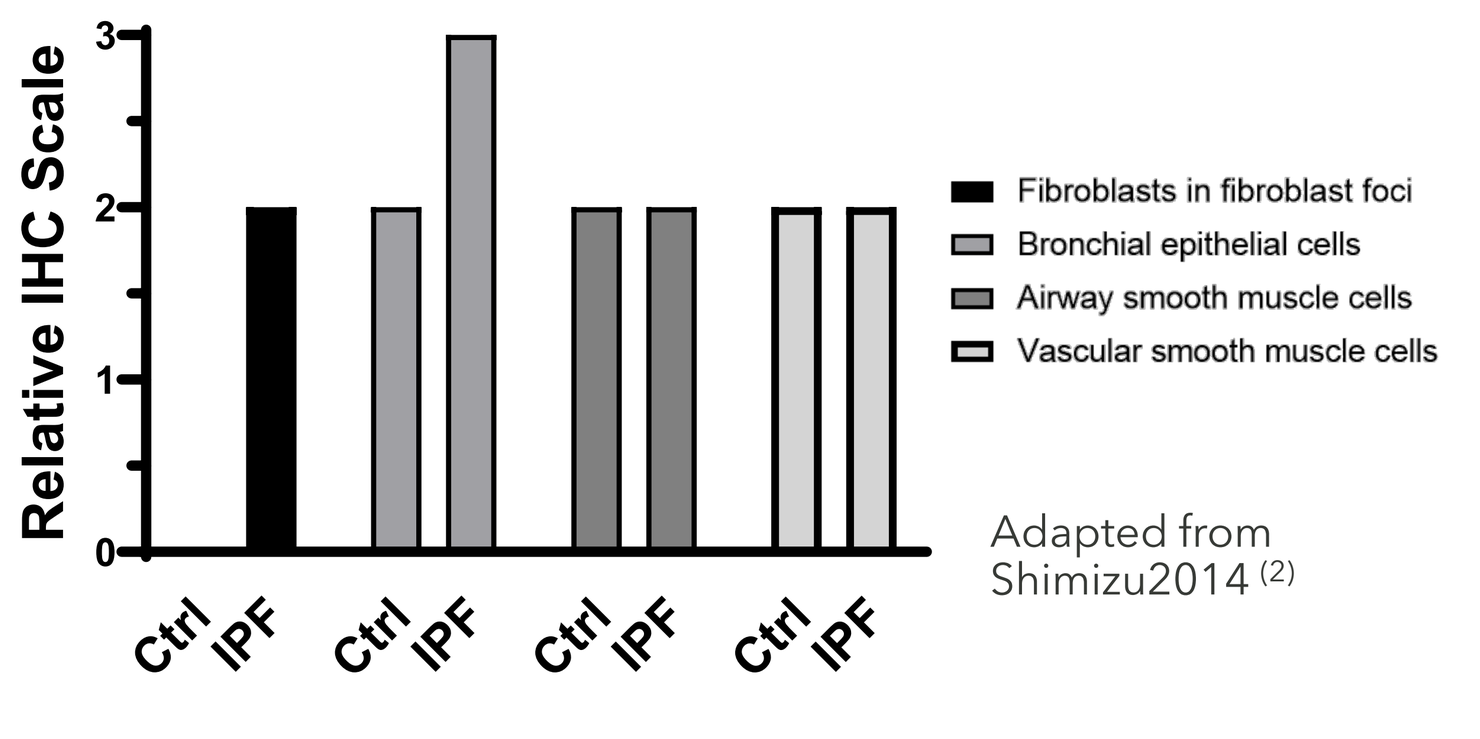

Relative IHC Scale
Fibroblasts in fibroblast foci
Bronchial epithelial cells
Airway smooth muscle cells
Vascular smooth muscle cells
Ctrl
IPF
Adapted from Shimizu2014 (2)

Intensity of immunostaining evaluated on a scale of 0 to 3
N=6 per group; NF: not found

- IPF fibrotic foci fibroblasts & bronchial epithelial cells show significant increases in ROCK2 expression
- ROCK2 expression is not increased in airway or vascular smooth muscle cells

Single cell IPF cell atlas data

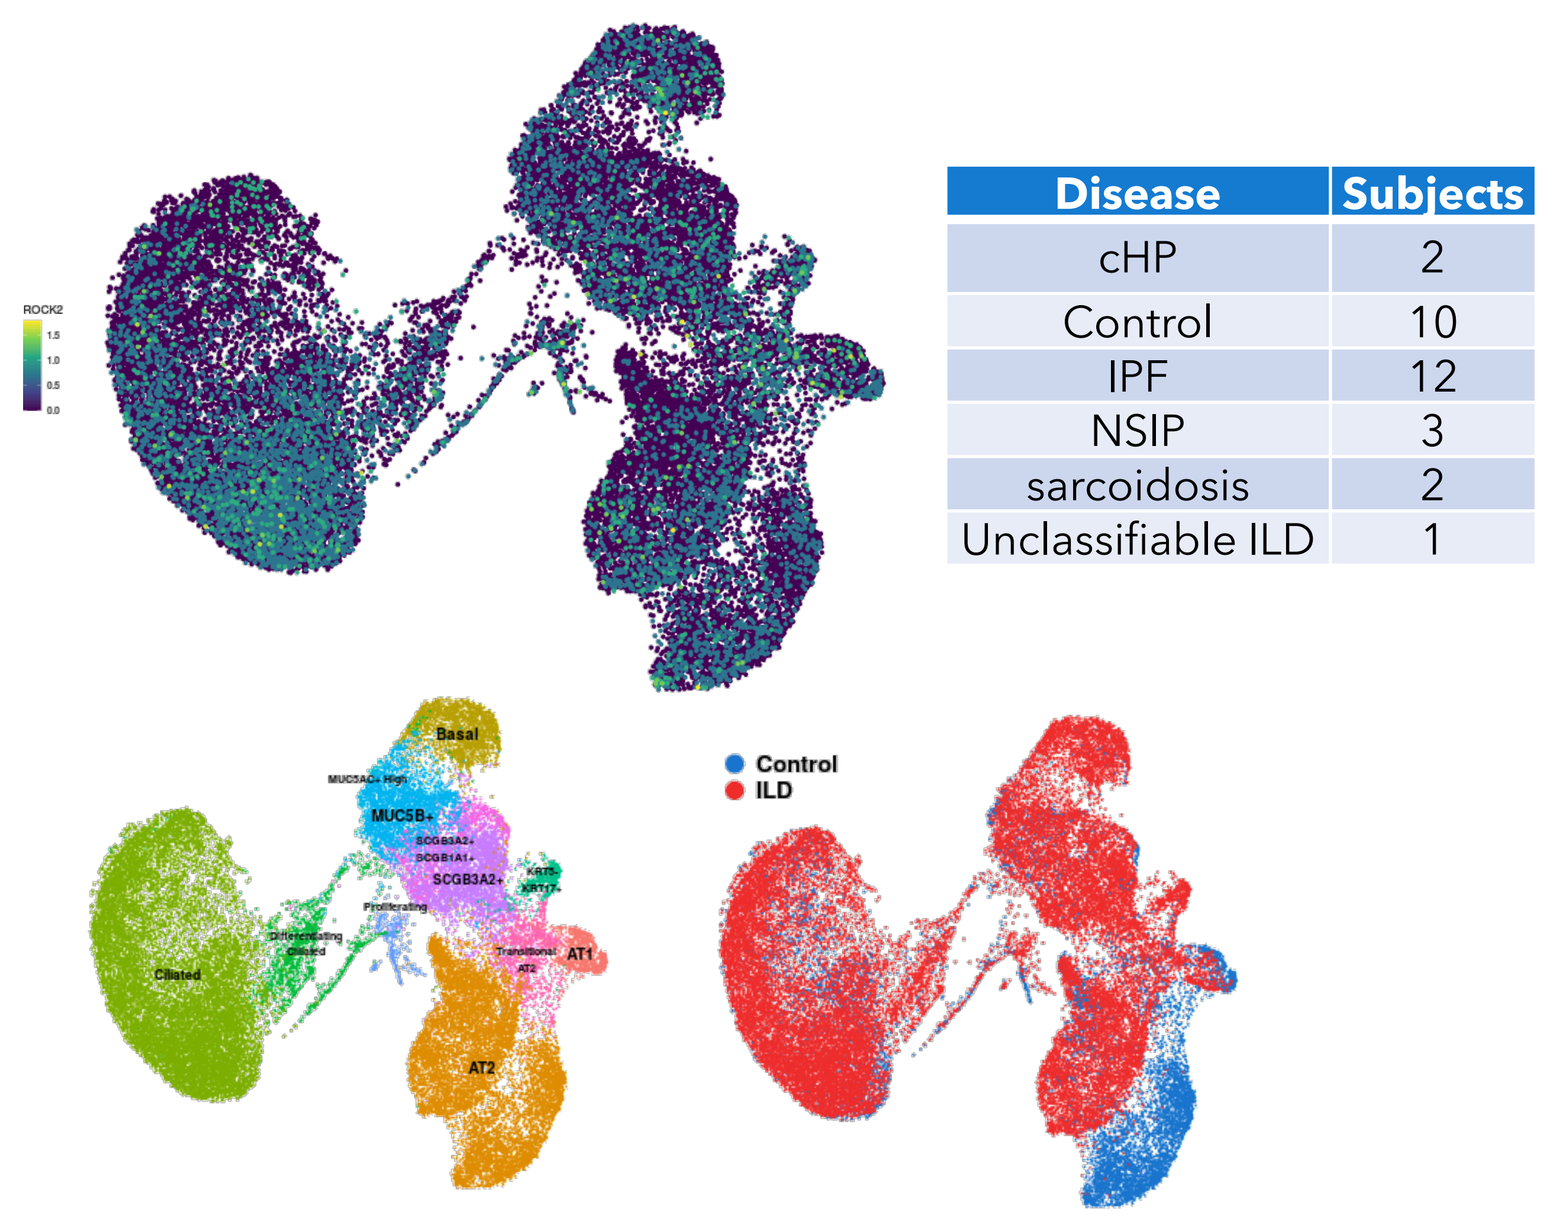

Control
ILD

Disease	Subjects
cHP	2
Control	10
IPF	12
NSIP	3
sarcoidosis	2
Unclassifiable ILD	1

- Increased ROCK2 expression is observed in ciliated bronchial epithelial cells

RESULTS

RXC007 Reduces Fibrosis and Collagen Deposition in a Therapeutic Murine Bleomycin-induced Lung Fibrosis Model

Study Design of the Therapeutic Murine Bleomycin-induced Lung Fibrosis Model (14-day treatment)

A total of 65 male C57BL/6 mice from Taconic mice were included in this study. The animals were divided into 6 groups. Animals in groups 2, 3, 4, 5, and 6, were administered 1.5 U/kg amounts of clinical bleomycin via oropharyngeal route. RXC007 and pirfenidone were dosed therapeutically from Day 7 to Day 21 via oral gavage.

RXC007 Reduces Fibrosis and Collagen Deposition in a Therapeutic Murine Bleomycin-induced Lung Fibrosis Model

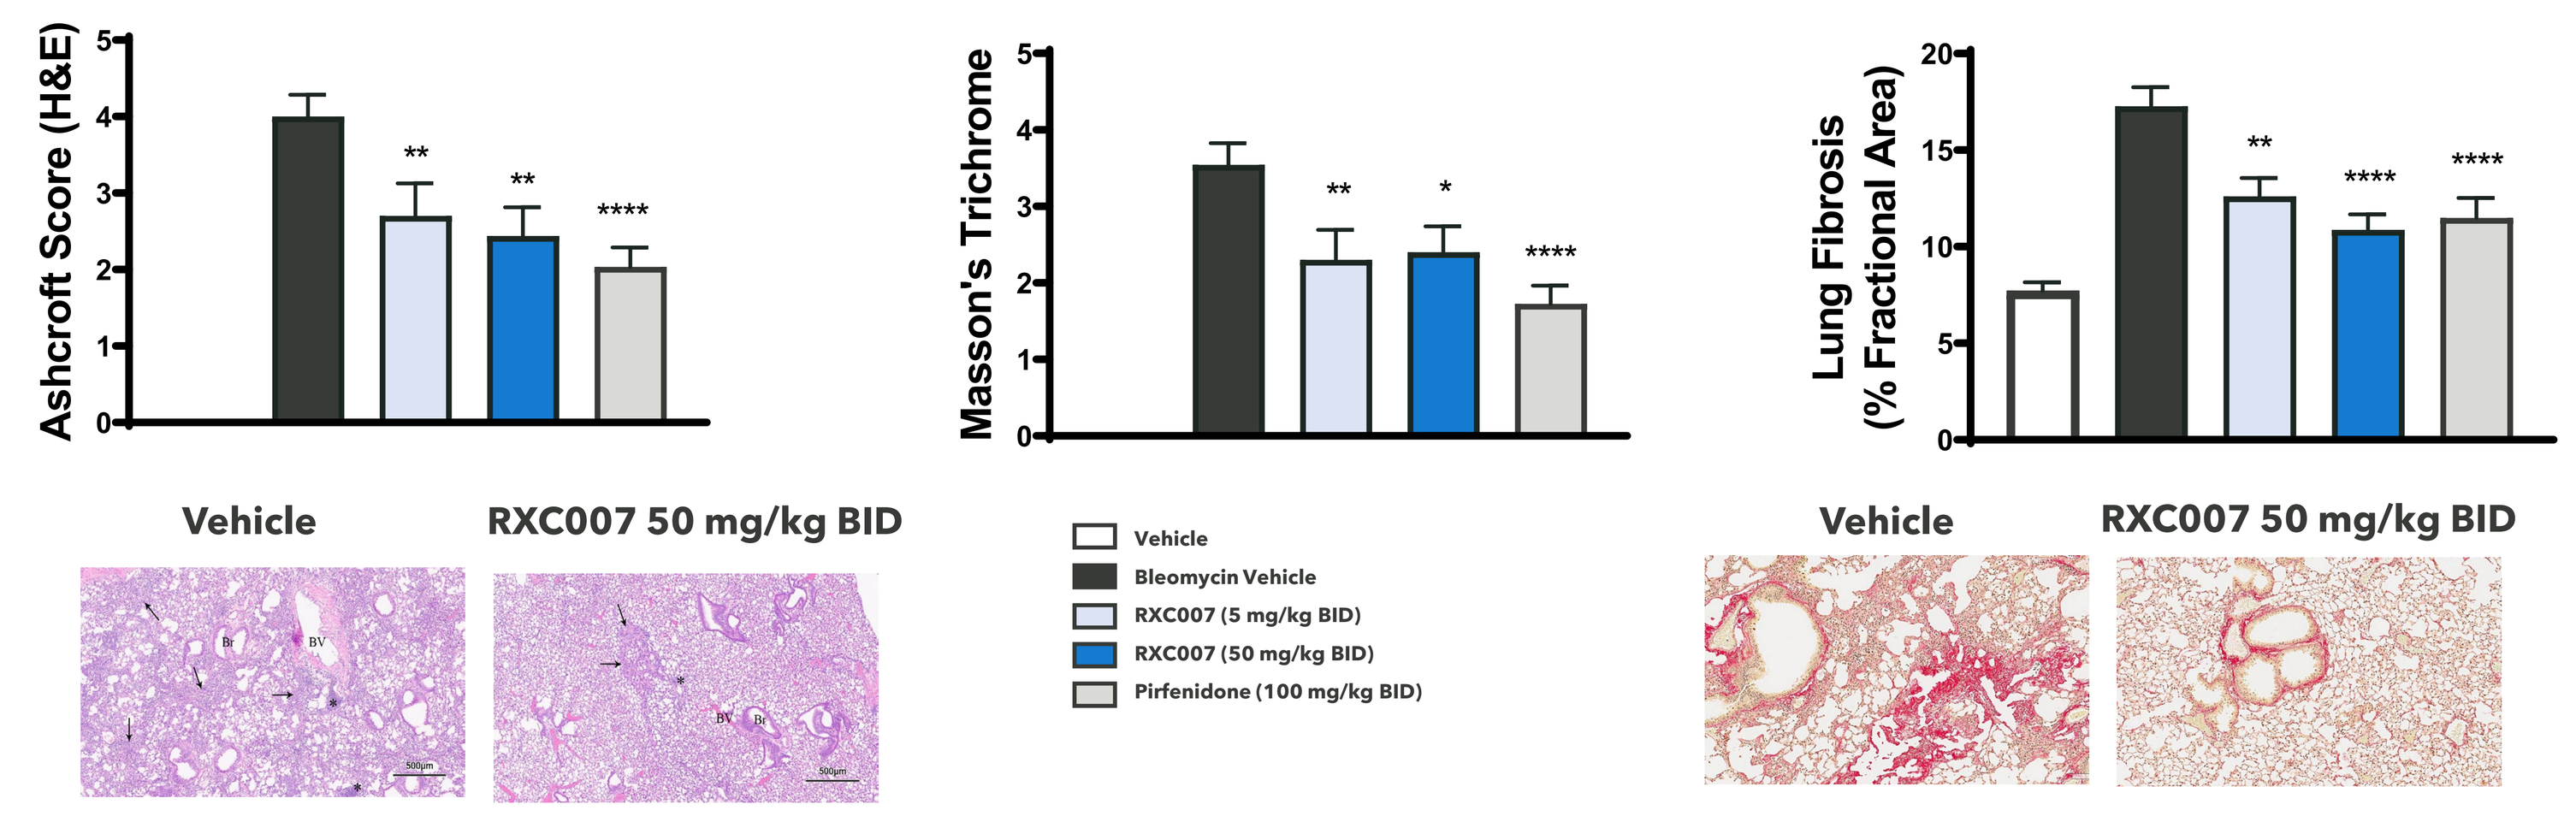

Ashcroft score
Mason's trichrome
Picrosirius red
Ashcroft Score (H&E)
Masson's Trichrome
Lung Fibrosis (% Fractional Area)
Vehicle
RXC007 50 mg/kg BID
Vehicle
Bleomycin Vehicle
RXC007 (5 mg/kg BID)
RXC007 (50 mg/kg BID)
Pirfenidone (100 mg/kg BID)

RXC007 Suppresses the Expression of a Number of Drivers Strongly Associated with Fibrosis and in IPF

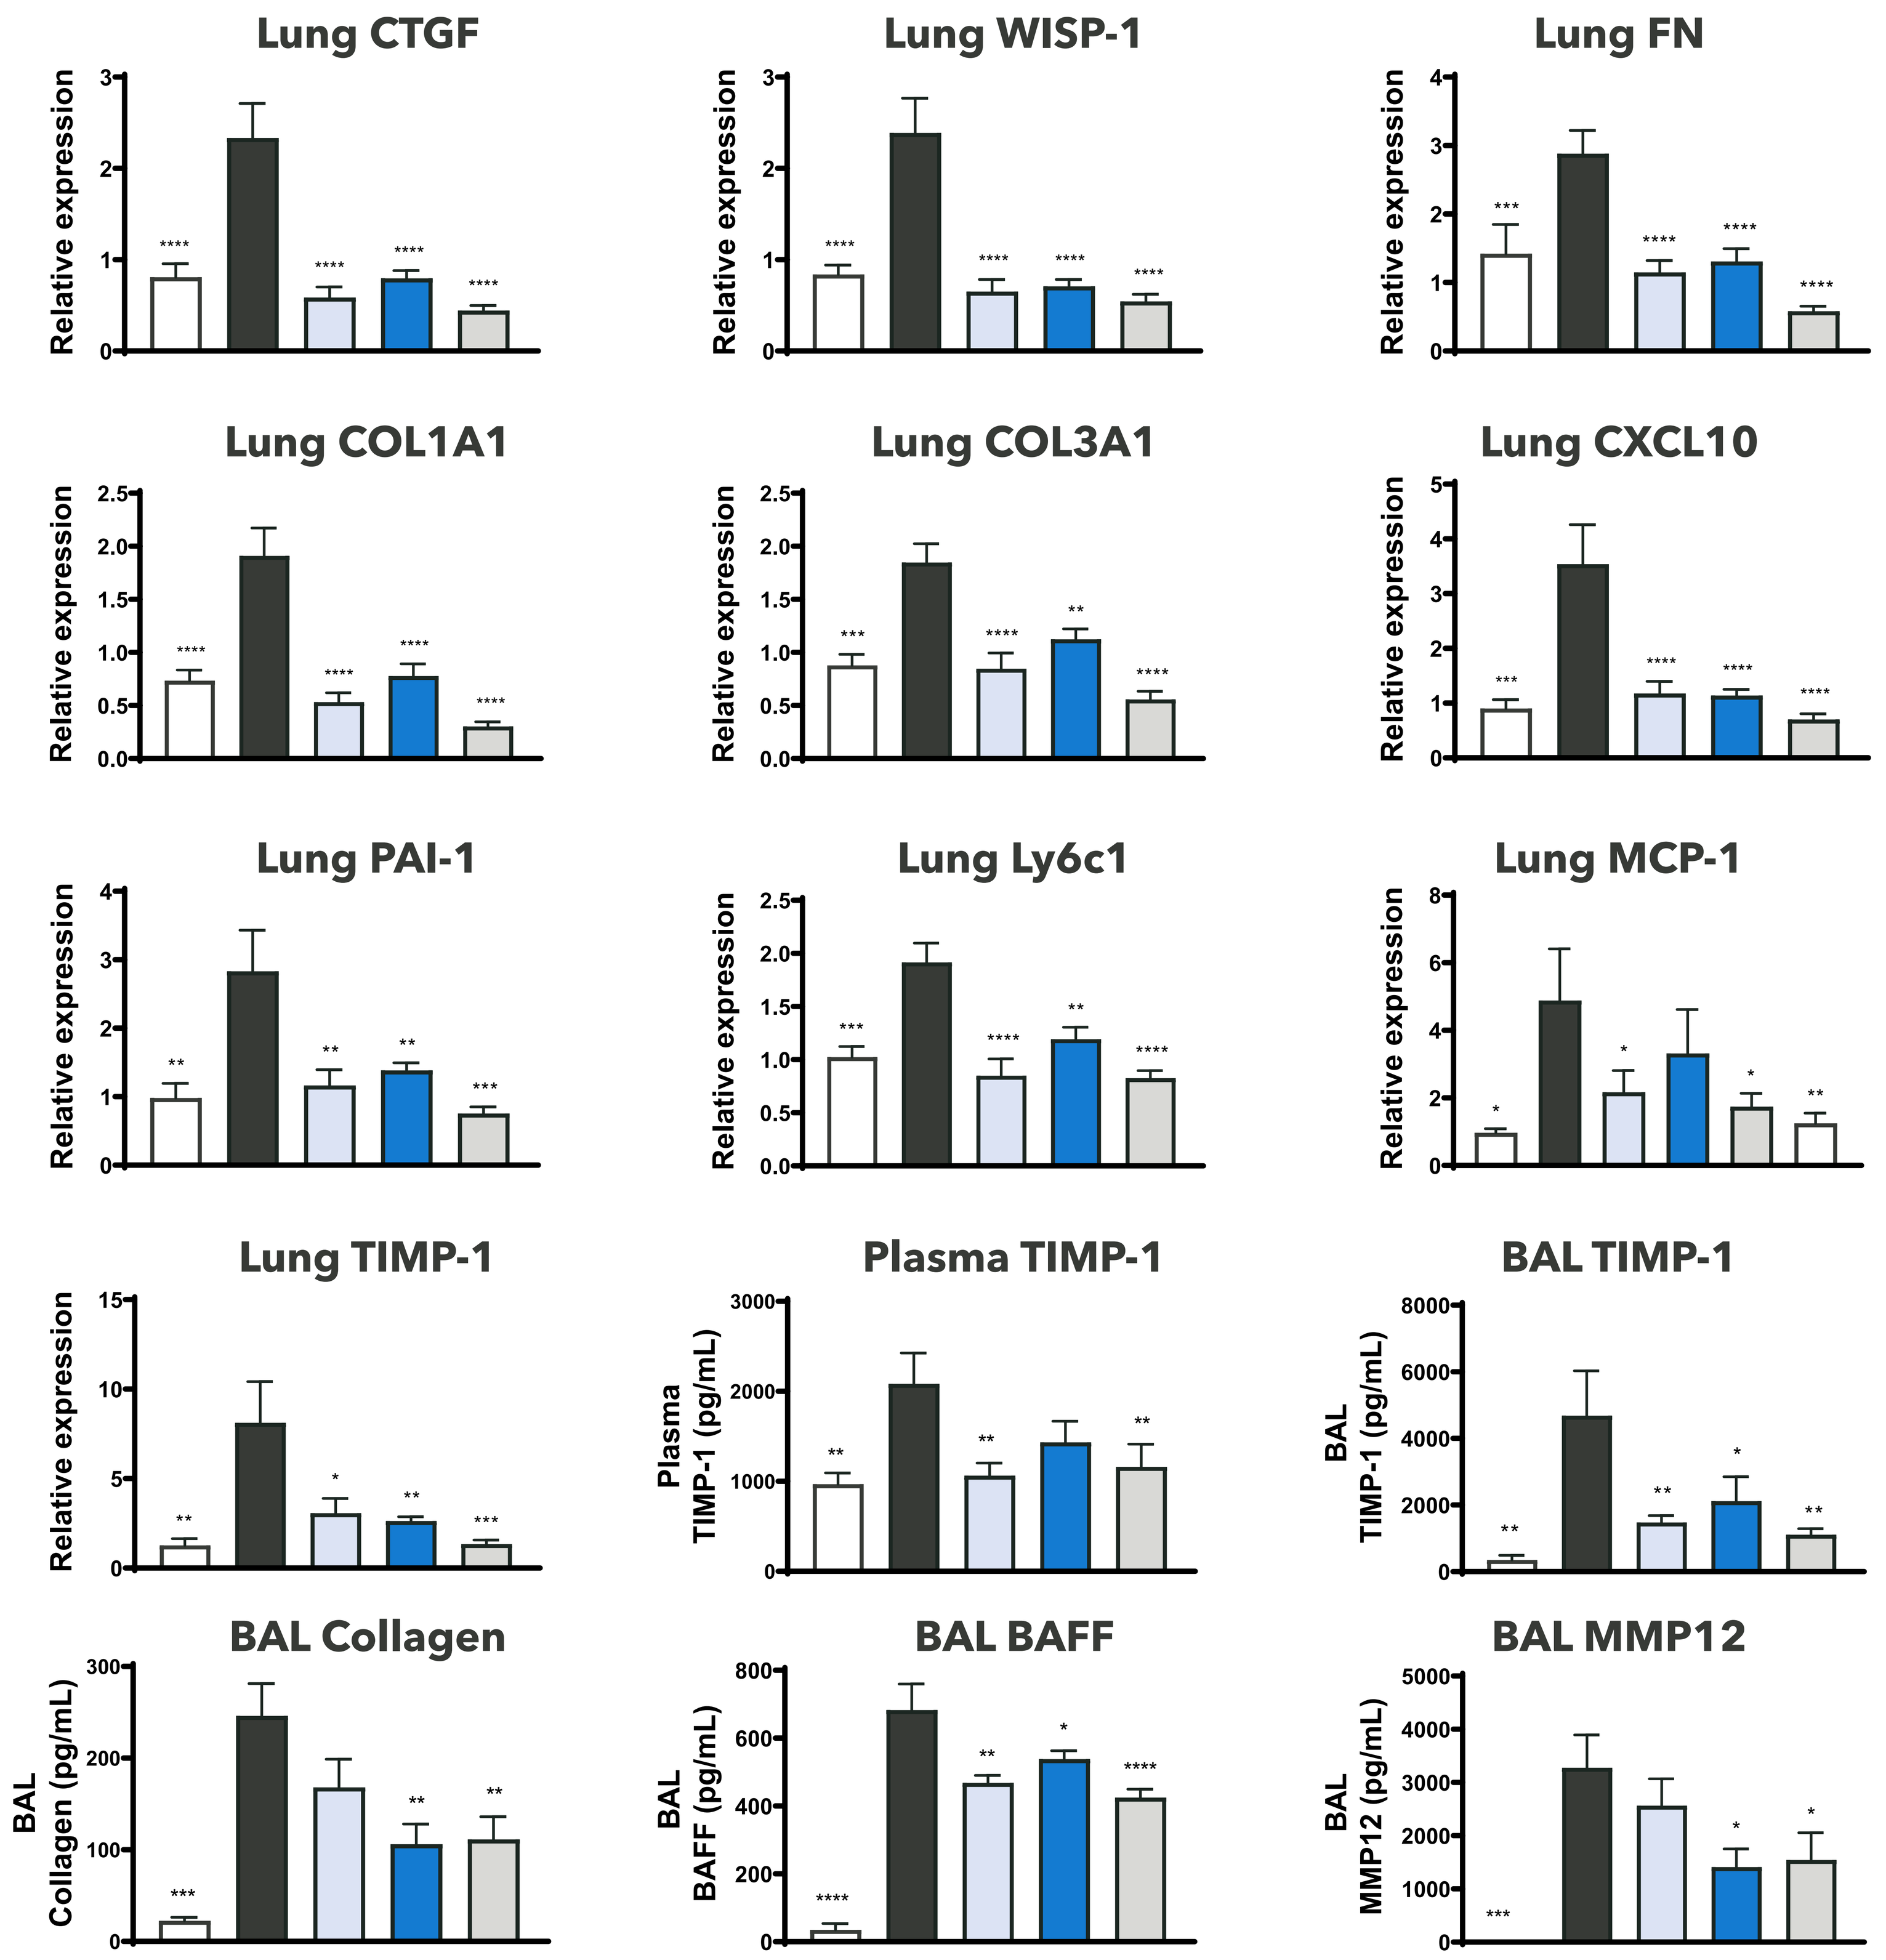

Lung CTGF
Lung WISP-1
Lung FN
Lung COL1A1
Lung COL3A1
Lung CXCL10
Lung PAI-1
Lung Ly6c1
Lung MCP-1
Lung TIMP-1
Plasma TIMP-1
BAL TIMP-1
BAL Collagen
BAL BAFF
BAL MMP12
Relative expression
Plasma TIMP-1 (pg/mL)
BAL TIMP-1 (pg/mL)
BAL Collagen (pg/mL)
BAL BAFF (pg/mL)
BAL MMP12 (pg/mL)

Significant effect of treatment determined by ANOVA and Fishers LSD post-test. *p<0.05, **p<0.01, ***p <0.001, ****p<0.0001. No animals were excluded from the analysis. 20 mg/kg dose tested not shown for clarity
Note: Different vehicle used for pirfenidone. Only used as positive control.

Pleiotropic Effects of RXC007 in a Murine Sclerodermatous chronic Graft versus Host Model

Study Design of the B10.D2→Balb/c (H-2(d)) minor histocompatibility antigen-mismatched model (28-day treatment)

Recipient mice (BALB/c (H-2d)) of an age of 8 weeks received total body irradiation with 700 cGy. Six hours after irradiation, all BALB/c (H-2d) recipients received bone marrow from either BALB/c (H-2d) in a syngeneic or B10.D2 (H-2d) in an allogeneic transplantation manner. For transplantation, 5 x 10^6 splenocytes and 2 x 10^6 bone marrow cells from donor mice were resuspended in 0.2 mL of PBS and injected via tail veins. To reflect the clinical situation with treatment only upon clinical signs of cGvHD, administration of RXC007 and nintedanib started 21 days after bone marrow transplantation and thus several days after the first clinically detectable manifestations of cGvHD in allogeneically transplanted mice. RXC007 and nintedanib administered by oral gavage BID for 28 days.

Significant reduction of collagen content and collagen deposition in the skin and lungs

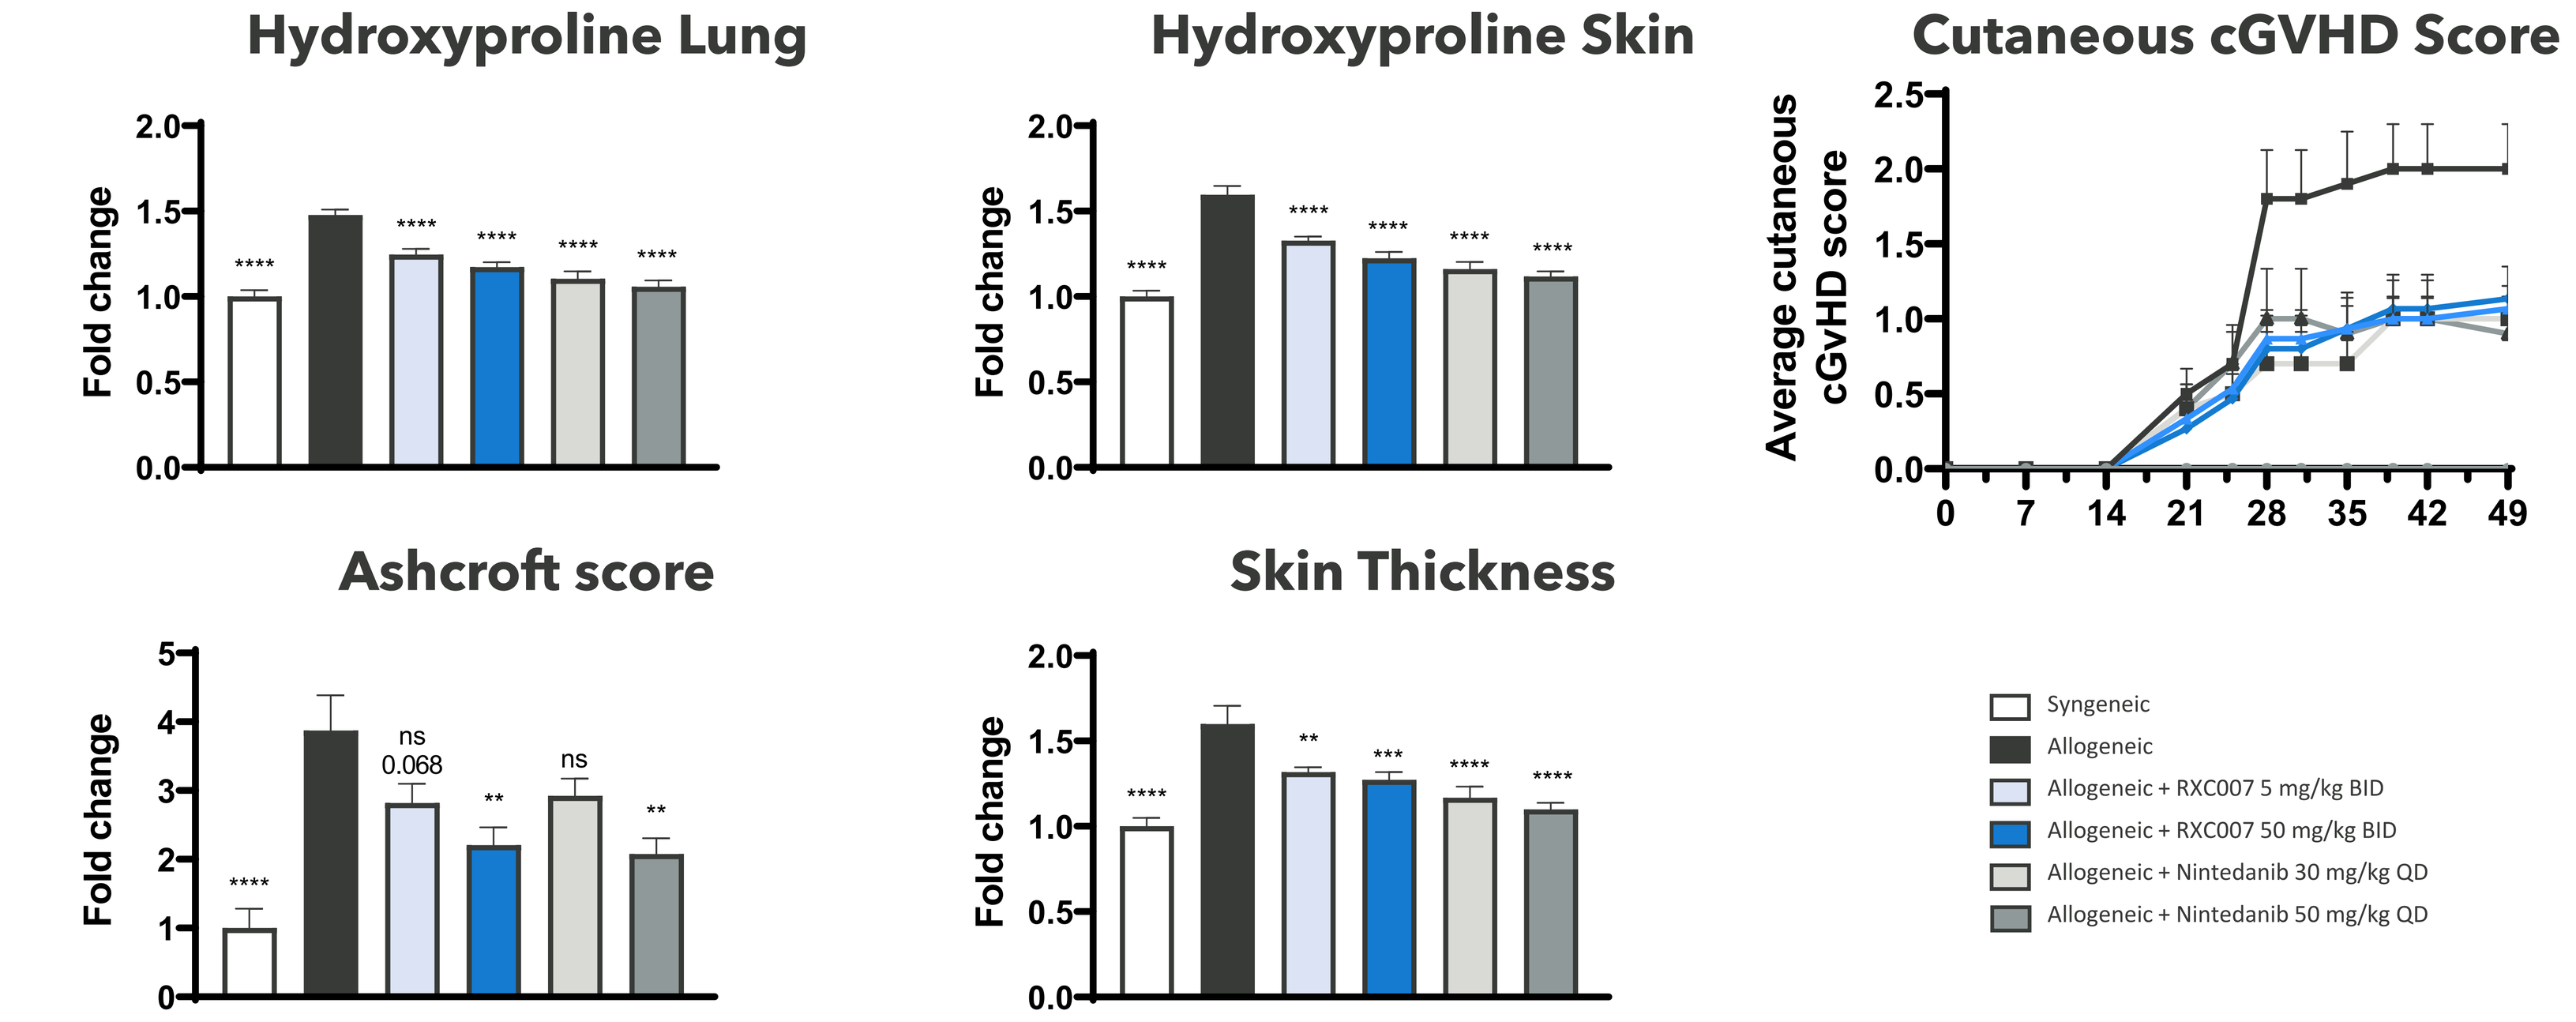

Hydroxyproline Lung
Hydroxyproline Skin
Cutaneous cGVHD Score
Ashcroft score
Skin Thickness
Fold change
Average cutaneous cGVHD score
Syngeneic
Allogeneic
Allogeneic + RXC007 5 mg/kg BID
Allogeneic + RXC007 50 mg/kg BID
Allogeneic + Nintedanib 30 mg/kg QD
Allogeneic + Nintedanib 50 mg/kg QD

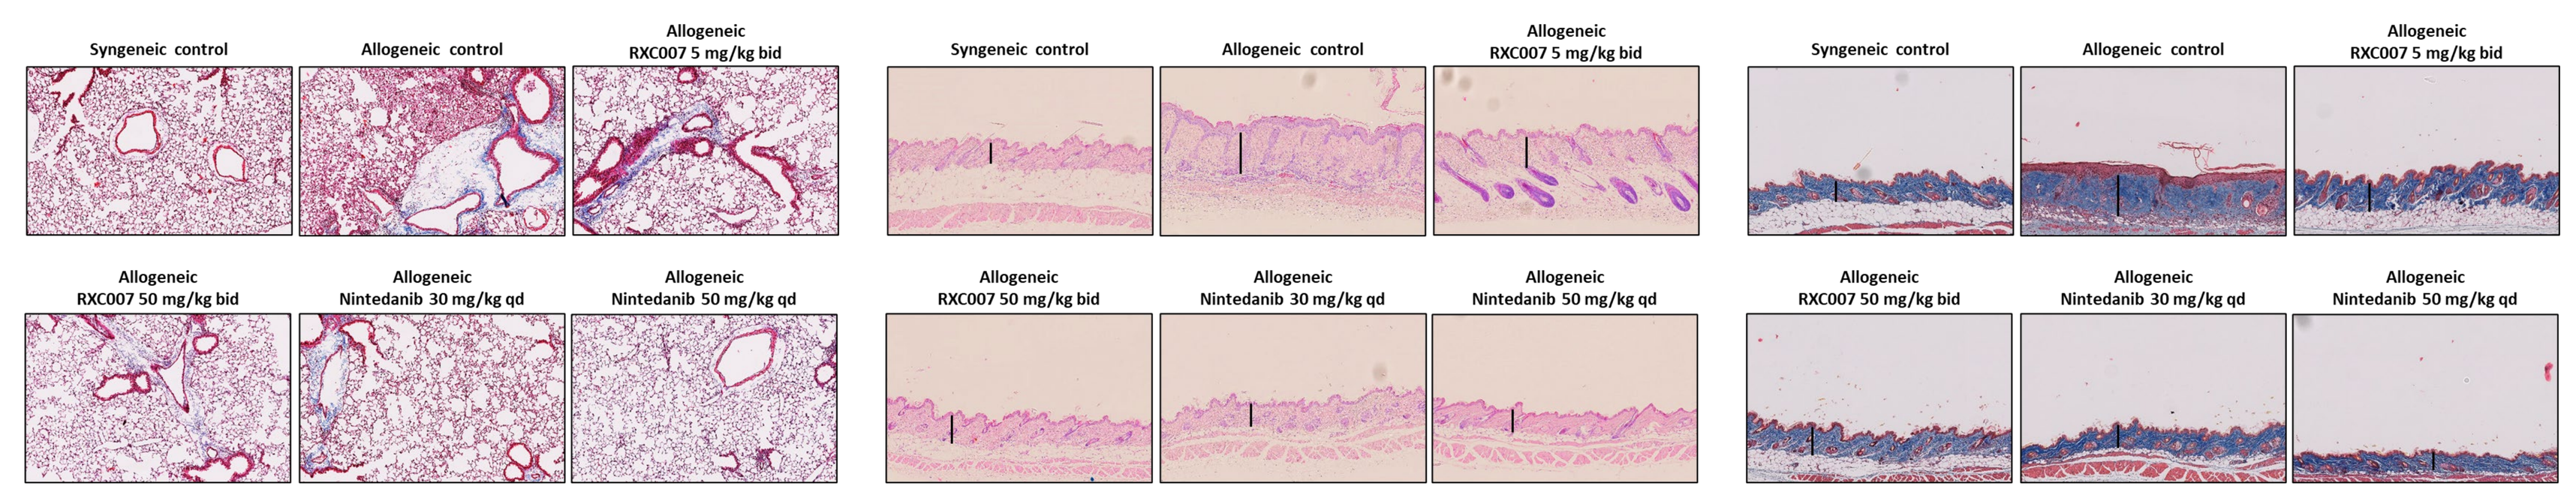

Syngeneic control
Allogeneic control
Allogeneic RXC007 5 mg/kg bid
Allogeneic RXC007 50 mg/kg bid
Allogeneic Nintedanib 30 mg/kg qd
Allogeneic Nintedanib 50 mg/kg qd

RXC007 reduces cutaneous PDGFRa+pJNK+ fibroblasts

JNK signalling is known to be strongly upregulated in activated fibroblasts and associated with pro-inflammatory and pro-fibrotic gene expression.

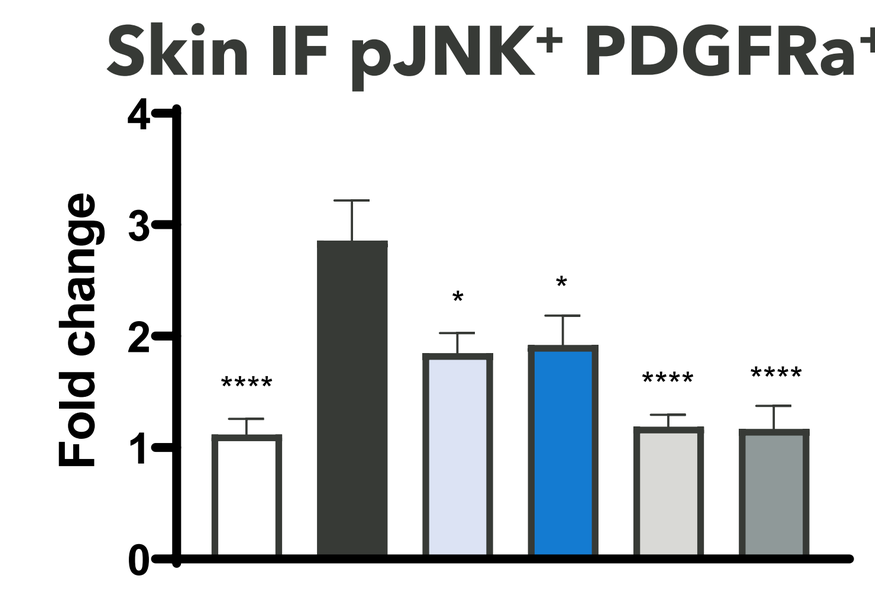

Skin IF pJNK+ PDGFRa+
Fold change

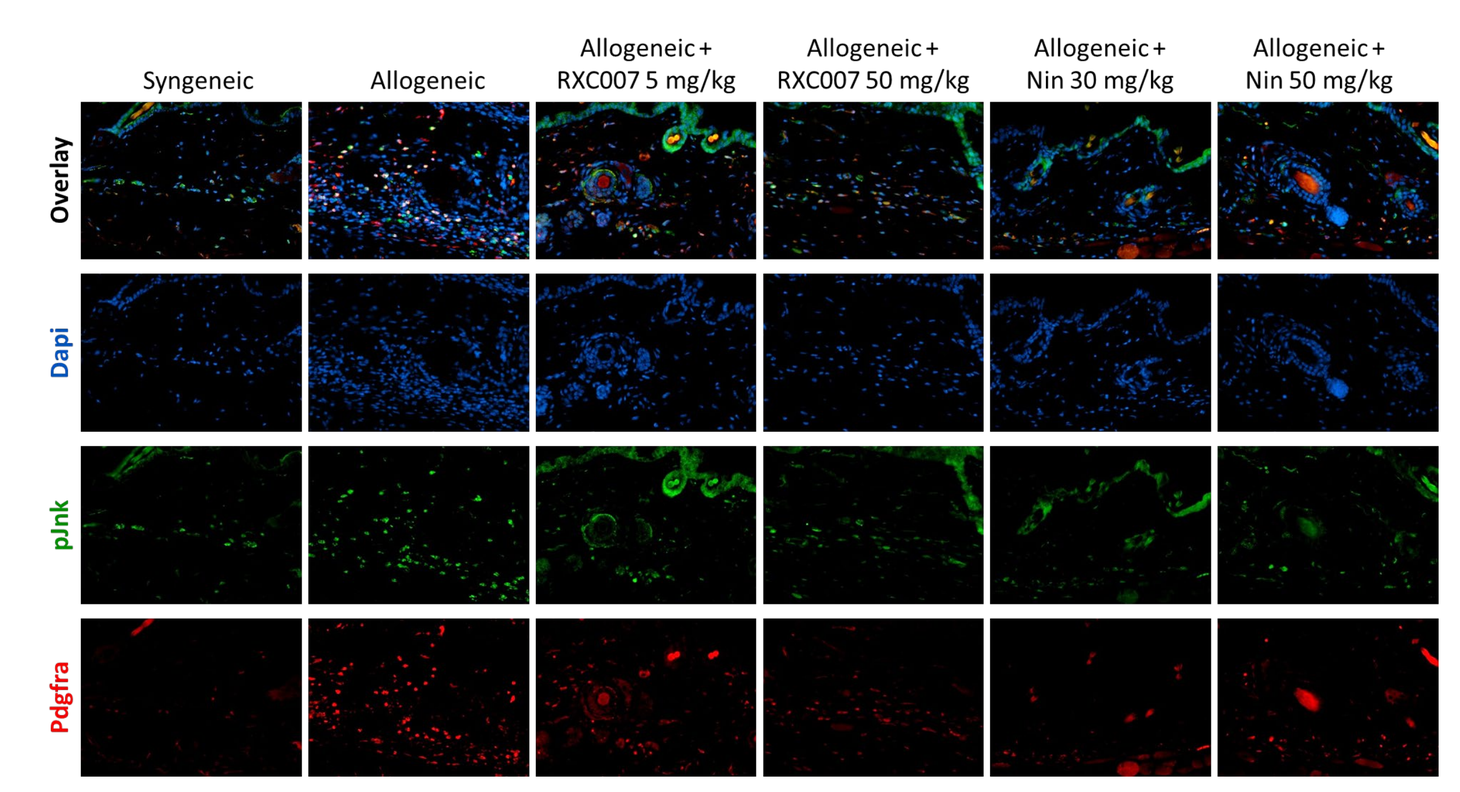

Syngeneic
Allogeneic
Allogeneic + RXC007 5 mg/kg
Allogeneic + RXC007 50 mg/kg
Allogeneic + Nin 30 mg/kg
Allogeneic + Nin 50 mg/kg
Overlay
Dapi
pJnk
Pdgfra

Summary

- RXC007 is a highly potent, selective and orally-active ROCK2 inhibitor currently in Phase 2a in IPF
- ROCK2 is a validated, compelling target at a key junction in cell signalling pathways central to fibrosis
- Robust preclinical efficacy data across disease models supports clinical development plan in lung fibrosis – IPF and CF-ILD

References

1. **Zhou et al**; *J Clin Invest. 2013 Mar;123(3):1096-108.*
2. **Shimizu et al**; Int J Immunopathol Pharmacol. 2014 Jan-Mar;27(1):37-44.
3. **Knipe et al**; *Am J Respir Cell Mol Biol. 2018 Apr; 58(4): 471–48.*

6th Annual AFDD Antifibrotic Drug Development

Redx Pharma, Block 33S, Mereside, Alderley Park, Cheshire, SK10 4TG, UK . e:n.guisot@redxpharma.com, www.redxpharma.com